Prevalence of Complications Among Adult and Pediatric Medicaid-Insured Patients With Sickle Cell Disease in the United States

Tanisha Smith, NP[1]; Andy Nguyen, PhD[2]; Crystal Henderson, PharmD[2]; Anne Beaubrun, PhD[2]; Melissa Lingohr-Smith, PhD[3]; Jay Lin, PhD, MBA[3]; Andrew Campbell, MD[4]

[1]Our Lady of the Lake Regional Medical Center, Baton Rouge, LA, USA; [2]Global Blood Therapeutics, South San Francisco, CA, USA; [3]Novosys Health, Green Brook, NJ, USA; [4]Children's National Medical Center, Washington, DC, USA

INTRODUCTION

- Sickle cell disease (SCD) is the most common inherited blood disorder in the United States, affecting approximately 100,000 Americans.[1,2]
- In the US, SCD is usually identified at birth through newborn screening. It is a lifelong, progressive disorder characterized by hemolytic anemia and vaso-occlusion, which can cause several acute and chronic complications, substantial morbidity, and early mortality.[3,4]
- Although previous studies have examined the complications associated with SCD, information on the real-world clinical burden of SCD in the US remains limited.

OBJECTIVE

- To examine the prevalence of SCD-related complications among adult and pediatric patients with SCD in the US.

METHODS

Study Population

- Adult (aged ≥18 years) and pediatric (aged <18 years) patients with ≥3 inpatient or outpatient SCD-related diagnoses (primary or secondary) on different days[5] between July 1, 2016, and December 31, 2019, were selected from the MarketScan Medicaid database.
 - The earliest SCD-related diagnosis date was defined as the index date.
- Patients were required to have continuous enrollment in a health plan for 6 months before the index date (baseline period) and ≥12 months after the index date (follow-up period) to be included in this study.
- Patients who received a hematopoietic stem cell transplant or bone marrow transplant before the index date were excluded from the study population.

Study Outcomes

- Patient demographic characteristics were analyzed for each patient during the baseline period and index date.
- The prevalence of SCD-related complications among adult and pediatric patients with SCD was evaluated during the follow-up period.
 - SCD-related complications were defined by clinical experts and identified by the International Classification of Diseases, Tenth Revision (ICD-10) codes within the MarketScan claims dataset.

RESULTS

- A total of 10,367 patients with SCD were included in the study; 54.2% were adult patients with a median follow-up of 47 months, and 45.8% were pediatric patients with a median follow-up of 50 months (**Table**).
- During the follow-up period, 94.7% of all patients with SCD experienced SCD-related complications; prevalence of any complication was slightly higher in adult (97.0%) than in pediatric (92.0%) patients (**Figure**).
- The 5 most prevalent complications in adult patients with SCD included vaso-occlusive crisis (VOC, 80.6%), anemia (75.9%), acute kidney injury (35.7%), acute chest syndrome (32.5%), and avascular necrosis (26.8%) (**Figure**).
- The 5 most prevalent complications in pediatric patients with SCD included VOC (79.7%), anemia (43.8%), acute chest syndrome (36.7%), infections (26.1%), which were predominately influenza, and sleep-disordered breathing (23.2%) (**Figure**).

Table. Demographics of Patients With SCD

	All (N=10,367)	Adult (N=5622)	Pediatric (N=4745)
Age in years, mean (SD)	23.0 (16.6)	35.1 (13.2)	8.8 (4.8)
Age group, n (%)			
≤3 years	850 (8.2)	0 (0.0)	850 (17.9)
4-11 years	2357 (22.7)	0 (0.0)	2357 (49.7)
12-17 years	1538 (14.8)	0 (0.0)	1538 (32.4)
18-30 years	2607 (25.2)	2607 (46.4)	0 (0.0)
31-44 years	1719 (16.6)	1719 (30.6)	0 (0.0)
45-54 years	715 (6.9)	715 (12.7)	0 (0.0)
55-64 years	414 (4.0)	414 (7.4)	0 (0.0)
≥65 years	167 (1.6)	167 (3.0)	0 (0.0)
Sex, n (%)			
Female	5669 (54.7)	3387 (60.3)	2282 (48.1)
Male	4698 (45.3)	2235 (39.8)	2463 (51.9)
Race/ethnicity, n (%)			
Black	8242 (79.5)	4745 (84.4)	3497 (73.7)
White	184 (1.8)	94 (1.7)	90 (1.9)
Hispanic	96 (0.9)	35 (0.6)	61 (1.3)
Other	69 (0.7)	32 (0.6)	37 (0.8)
Follow-up duration, months			
Mean (SD)	39.4 (15.2)	38.8 (15.3)	40.2 (15.0)
Median	49	47	50

SCD, sickle cell disease.

Figure. Prevalence of SCD-Related Complications[a] in the Follow-up Period Among Adult and Pediatric Patients With SCD

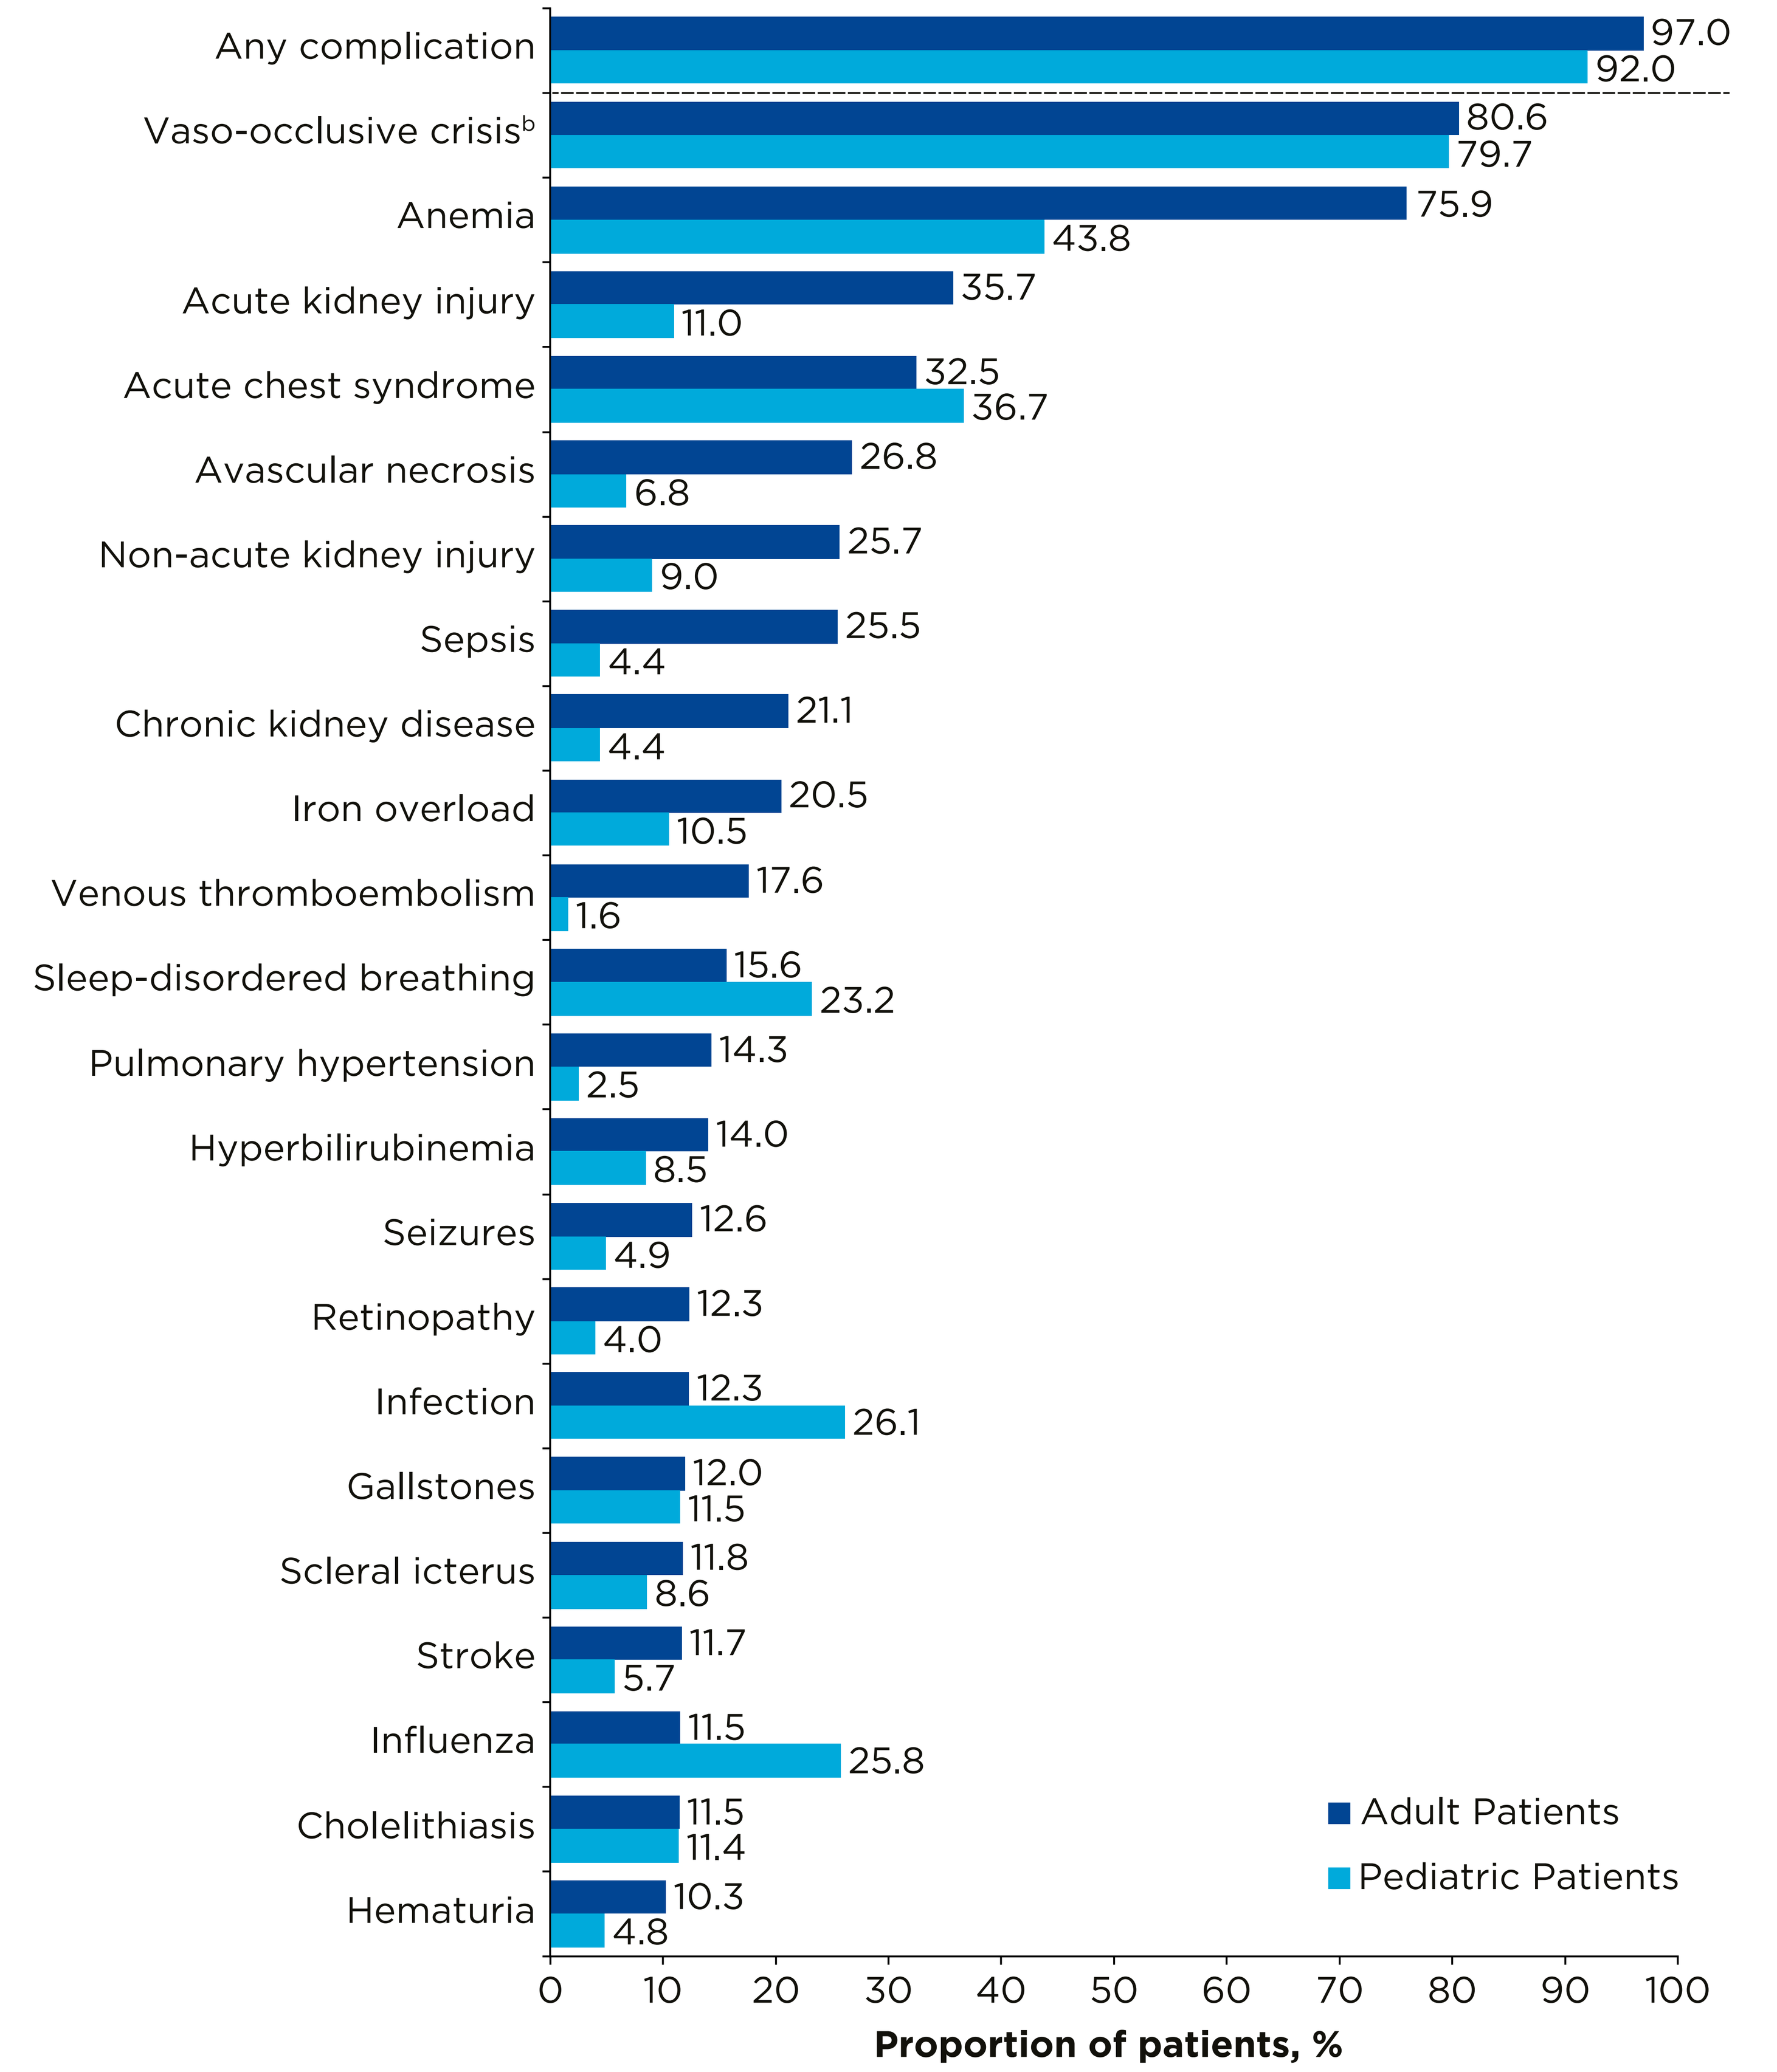
Any complication 97.0 92.0
Vaso-occlusive crisis[b] 80.6 79.7
Anemia 75.9 43.8
Acute kidney injury 35.7 11.0
Acute chest syndrome 32.5 36.7
Avascular necrosis 26.8 6.8
Non-acute kidney injury 25.7 9.0
Sepsis 25.5 4.4
Chronic kidney disease 21.1 4.4
Iron overload 20.5 10.5
Venous thromboembolism 17.6 1.6
Sleep-disordered breathing 15.6 23.2
Pulmonary hypertension 14.3 2.5
Hyperbilirubinemia 14.0 8.5
Seizures 12.6 4.9
Retinopathy 12.3 4.0
Infection 12.3 26.1
Gallstones 12.0 11.5
Scleral icterus 11.8 8.6
Stroke 11.7 5.7
Influenza 11.5 25.8
Cholelithiasis 11.5 11.4
Hematuria 10.3 4.8
Adult Patients
Pediatric Patients
0 10 20 30 40 50 60 70 80 90 100
Proportion of patients, %

[a]Includes complications occurring in >10% of adult patients with SCD in the overall follow-up period. [b]A diagnosis of vaso-occlusive crisis is indicated with the same ICD-10 code used to indicate a diagnosis of dactylitis; therefore, the reported percentage may also include occurrences of dactylitis.

ICD-10, International Classification of Diseases, Tenth Revision; SCD, sickle cell disease.

LIMITATIONS

- This study is a claims-based database analysis and may be subject to the general limitations associated with administrative claims data.
 - As with most claims databases, the MarketScan Medicaid database comprises claims submitted by healthcare providers for reimbursement and may contain coding errors, coding for rule-out, and undercoding or overcoding.
 - SCD-related complications were identified by ICD-10 codes on administrative claims, and diagnoses were not confirmed using physician notes or other methods.
 - The study population represents patients with SCD with US Medicaid coverage; thus, although the study findings are generalizable to similar patients across the US, they may not be generalizable to patients with SCD with different or no insurance coverage.

CONCLUSIONS

- Most patients with SCD included in the study experienced disease-related complications, and prevalence rates for different complications were generally higher in adult than in pediatric patients.
- While VOCs are the most common complication in patients with SCD, a multitude of complications, such as anemia, acute chest syndrome, acute kidney injury, and avascular necrosis, are also highly prevalent.
- These study findings indicate a need for early interventions to potentially reduce the risk of chronic and progressive complications and improve long-term outcomes of Medicaid-insured patients with SCD in the US.

ACKNOWLEDGMENTS

- We thank all the patients with sickle cell disease, families, caregivers, and investigators contributing to these studies.

DISCLOSURES

- **Tanisha Smith:** Employee: Novartis Pharmaceuticals; research participation: Global Blood Therapeutics. **Andy Nguyen:** Employee, equity ownership: Global Blood Therapeutics. **Crystal Henderson:** Employee, equity ownership: Global Blood Therapeutics. **Anne Beaubrun:** Employee, equity ownership: Global Blood Therapeutics. **Melissa Lingohr-Smith:** Employee: Novosys Health; research funding: Global Blood Therapeutics. **Jay Lin:** Employee: Novosys Health; research funding: Global Blood Therapeutics. **Andrew Campbell:** Consultant: Global Blood Therapeutics, Cyclerion Therapeutics; research funding: Global Blood Therapeutics, Novartis, Cyclerion Therapeutics.

REFERENCES

1. Centers for Disease Control and Prevention. Data and statistics on sickle cell disease. Accessed September 1, 2022. https://www.cdc.gov/ncbddd/sicklecell/data.html
2. Food and Drug Administration. Sickle cell disease. Accessed September 6, 2022. https://www.fda.gov/consumers/minority-health-and-health-equity-resources/sickle-cell-disease
3. Abboud MR. Standard management of sickle cell disease complications. *Hematol Oncol Stem Cell Ther*. 2020;13(2):85-90.
4. Kato GJ, Piel FB, Reid CD, et al. Sickle cell disease. *Nat Rev Dis Primers*. 2018;4:18010.
5. Snyder AB, Zhou M, Theodore R, et al. Improving an administrative case definition for longitudinal surveillance of sickle cell disease. *Public Health Rep*. 2019;134(3):274-281.

Presented at AMCP Nexus; October 11-14, 2022; National Harbor, MD, USA.

© 2022 Global Blood Therapeutics.