A Multicenter Chart Review Study on Real-World Experience of Patients With Sickle Cell Disease Treated With Voxelotor

Ken Bridges, MD[1]; Nicole Allie, PharmD[2]; Kelly Kueh[3]; Dale Chandler, PhD[2]; Clare Tong[1]; Andy Nguyen, PhD[1]; Anne Beaubrun, PhD[1]

[1]Global Blood Therapeutics, South San Francisco, CA, USA; [2]Ipsos UK Ltd, London, UK; [3]Ipsos Malaysia, Kuala Lumpur, Malaysia

INTRODUCTION

- Sickle cell disease (SCD) is an inherited blood disorder in which sickle hemoglobin (HbS) polymerization, the root cause of the disease, leads to a range of symptoms and complications, including pain, fatigue, and irreversible end-organ damage, that result in a substantial humanistic burden.[1,2]
- Characteristic manifestations of SCD include chronic hemolytic anemia, painful episodic vaso-occlusive crises (VOCs), widespread organ damage, and other complications, such as leg ulcers, all of which are linked to deficits in both health outcomes and quality of life (QOL).[1,3]
- Voxelotor is a first-in-class HbS polymerization inhibitor approved in the United States for patients with SCD aged ≥4 years and in the European Union, Great Britain, the United Arab Emirates, Oman, and Kuwait for patients aged ≥12 years.[4-6]
- Real-world evidence broadens our existing knowledge of the effectiveness and safety of voxelotor in treating patients with SCD and provides new insights that can benefit the clinical decision-making process for healthcare providers (HCPs).[7]

OBJECTIVE

- To examine real-world outcomes among patients receiving voxelotor.

METHODS

- In this multicenter online medical chart review, records of patients with SCD aged ≥12 years were collected between November 2021 and March 2022 from US physicians with 20 to 40 years of specialty practice, with ≥7 patients with SCD in their direct care who were seen in the last 6 months.
- Mean hemoglobin (Hb) level was evaluated at baseline and various timepoints in follow-up per standard of care, and the average numbers of VOCs and transfusions in the 6 months before and after starting voxelotor were compared.
- Changes in patients' QOL were based on physicians' assessments.
 - Physicians were asked to report the number of work or school days missed, blood transfusions, and symptoms, such as changes in pain intensity and fatigue.
 - Visual analog, numeric rating, and faces pain scales were the most used tools to assess patients' pain levels.

RESULTS

Patients

- Of 1138 patients with SCD aged ≥12 years, 185 were receiving voxelotor and had a mean age of 31 years (**Table**).
- Overall, most patients were male (58% of all patients, 58% of voxelotor-treated patients), and the majority were homozygous for SCD (HbSS; 61% of all patients, 57% of voxelotor-treated patients) (**Table**).
- Mean Hb at treatment initiation was comparable between patients with HbSS and hemoglobin SC disease (HbSC) genotypes (**Table**).

Table. Baseline Characteristics

	All patients (N=1138)	Voxelotor-treated patients (n=185)
Age, mean, years (SD)	28 (13.79)	31 (15.55)
Male, n (%)	660 (58%)	108 (58%)
Race, n (%)		
African American	791 (70%)	107 (58%)
White	169 (15%)	36 (19%)
Asian	30 (3%)	7 (4%)
Mixed race	46 (4%)	11 (6%)
Native American/Alaskan Native	19 (2%)	6 (3%)
Ethnicity, n (%)		
Hispanic	78 (7%)	18 (10%)
Non-Hispanic	1060 (93%)	167 (90%)
HbSS genotype, n (%)	689 (61%)	105 (57%)
Hb, mean (SD), g/dL	8.90 (2.31)	8.91 (2.52)
HbSC genotype, n (%)	287 (25%)	42 (23%)
Hb, mean (SD), g/dL	9.28 (2.49)	9.08 (2.50)

Hb, hemoglobin; HbSC, hemoglobin SC disease; HbSS, homozygous for SCD.

Increased Hb With Voxelotor

- An increase in Hb of 0.9 g/dL was observed as early as 2 weeks following voxelotor treatment initiation, with the greatest mean increase of 1.5 g/dL noted at 7 to 9 months post-treatment initiation (**Figure 1**).
- Among patients who received voxelotor for ≥6 months (n=77), mean Hb increased by 1.18 g/dL from treatment initiation.

Figure 1. Mean Increase From Baseline in Hb Levels With Voxelotor

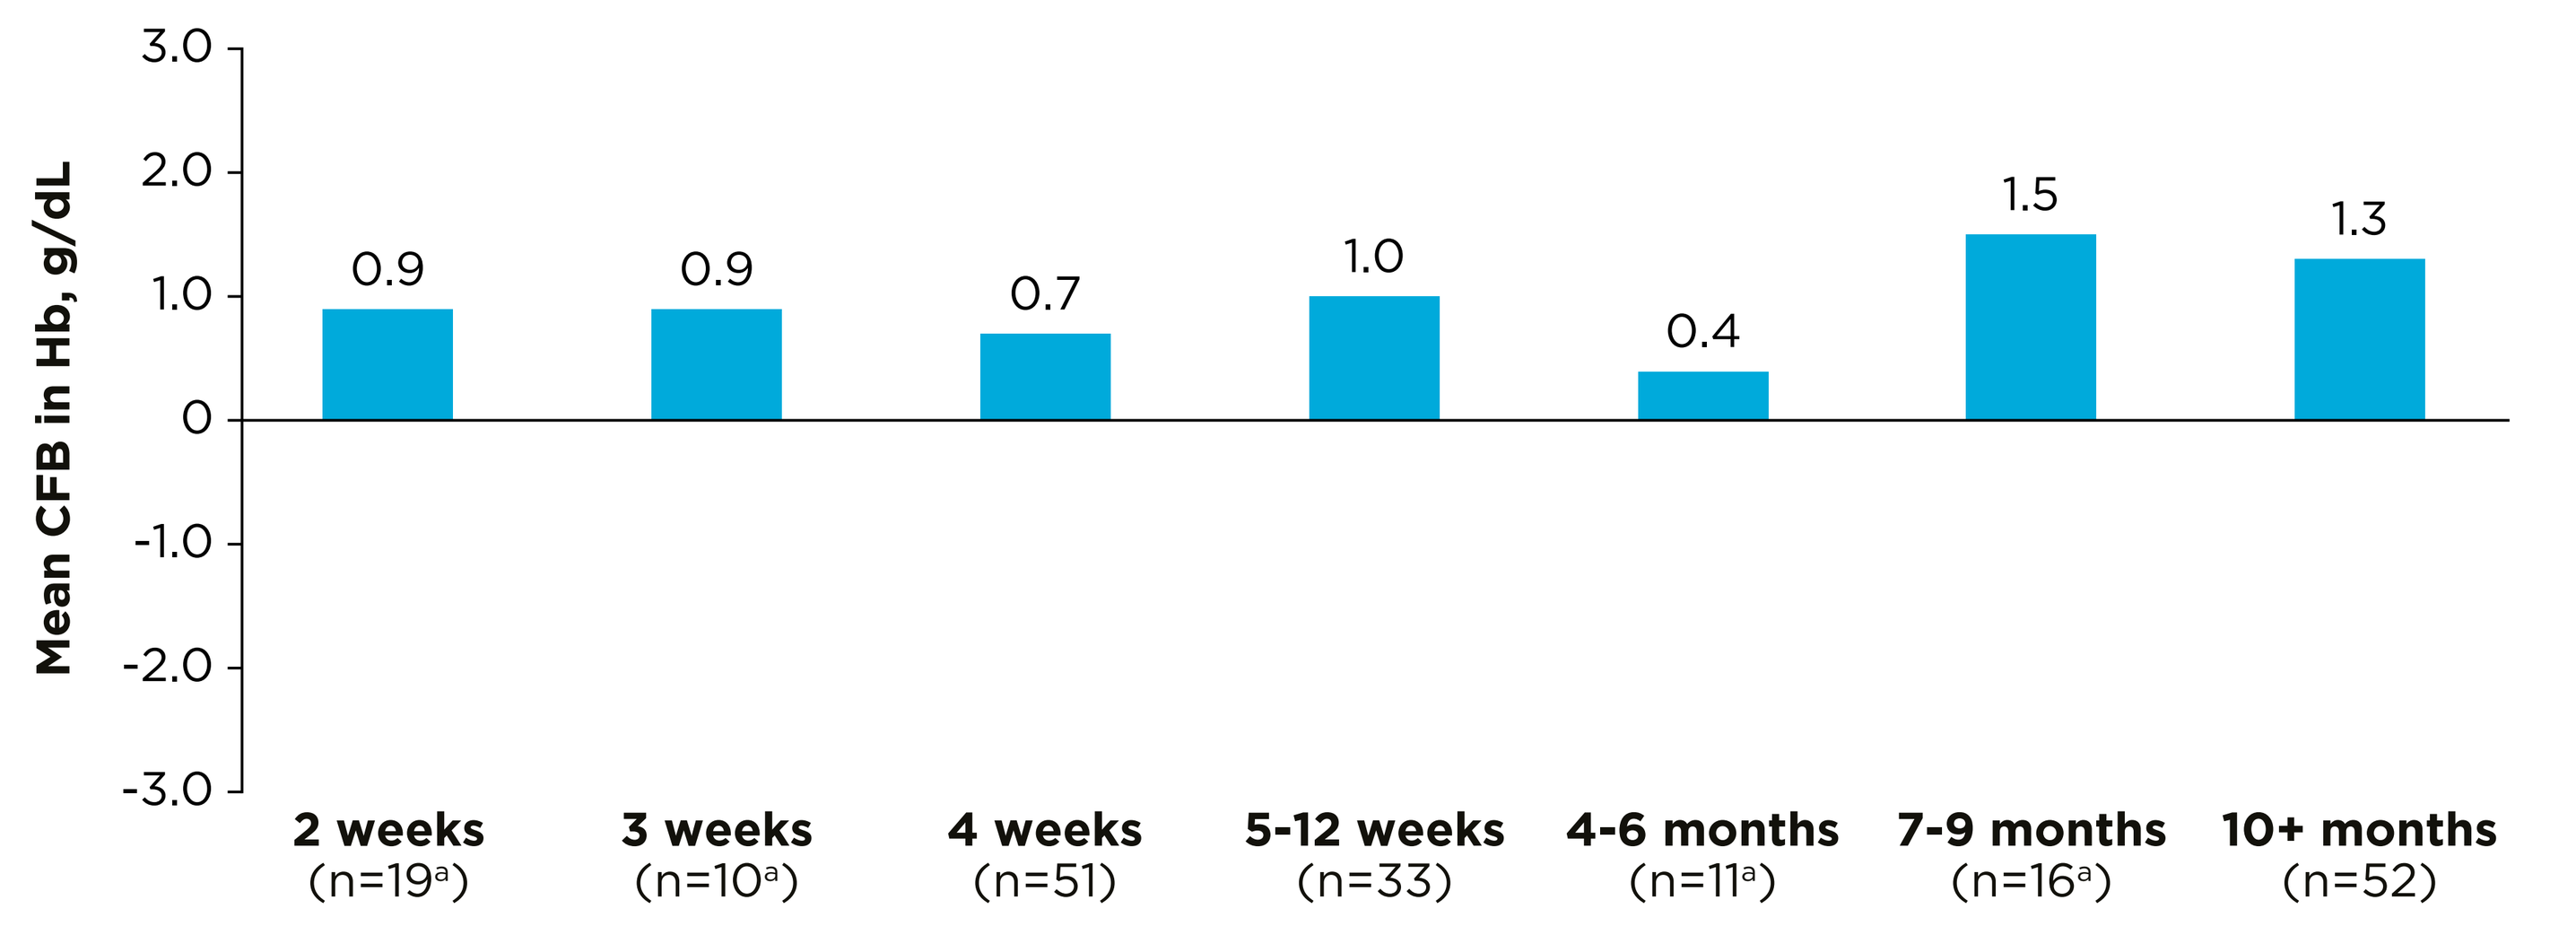
Mean CFB in Hb, g/dL
3.0
2.0
1.0
0
-1.0
-2.0
-3.0
0.9
0.9
0.7
1.0
0.4
1.5
1.3
2 weeks (n=19a)
3 weeks (n=10a)
4 weeks (n=51)
5-12 weeks (n=33)
4-6 months (n=11a)
7-9 months (n=16a)
10+ months (n=52)

[a]Low base.
CFB, change from baseline; Hb, hemoglobin.

Decreased VOCs and Transfusions With Voxelotor

- Overall, patients experienced a decrease in the number of VOCs and the number of blood transfusions following voxelotor treatment initiation (**Figure 2**).
- Among patients treated with voxelotor for ≥6 months:
 - The mean number of VOCs decreased by 56%, from 1.86 pre-voxelotor to 0.81 post-voxelotor (**Figure 2A**).
 - The mean number of transfusions decreased by 71%, from 1.37 pre-voxelotor to 0.40 post-voxelotor (**Figure 2B**).
- Among patients treated with voxelotor for <6 months:
 - The mean number of VOCs decreased by 38%, from 2.26 pre-voxelotor to 1.41 post-voxelotor (**Figure 2C**).
 - The mean number of transfusions decreased by 33%, from 2.26 pre-voxelotor to 1.51 post-voxelotor (**Figure 2D**).

Figure 2. Fewer VOCs and Blood Transfusions With Voxelotor Treatment

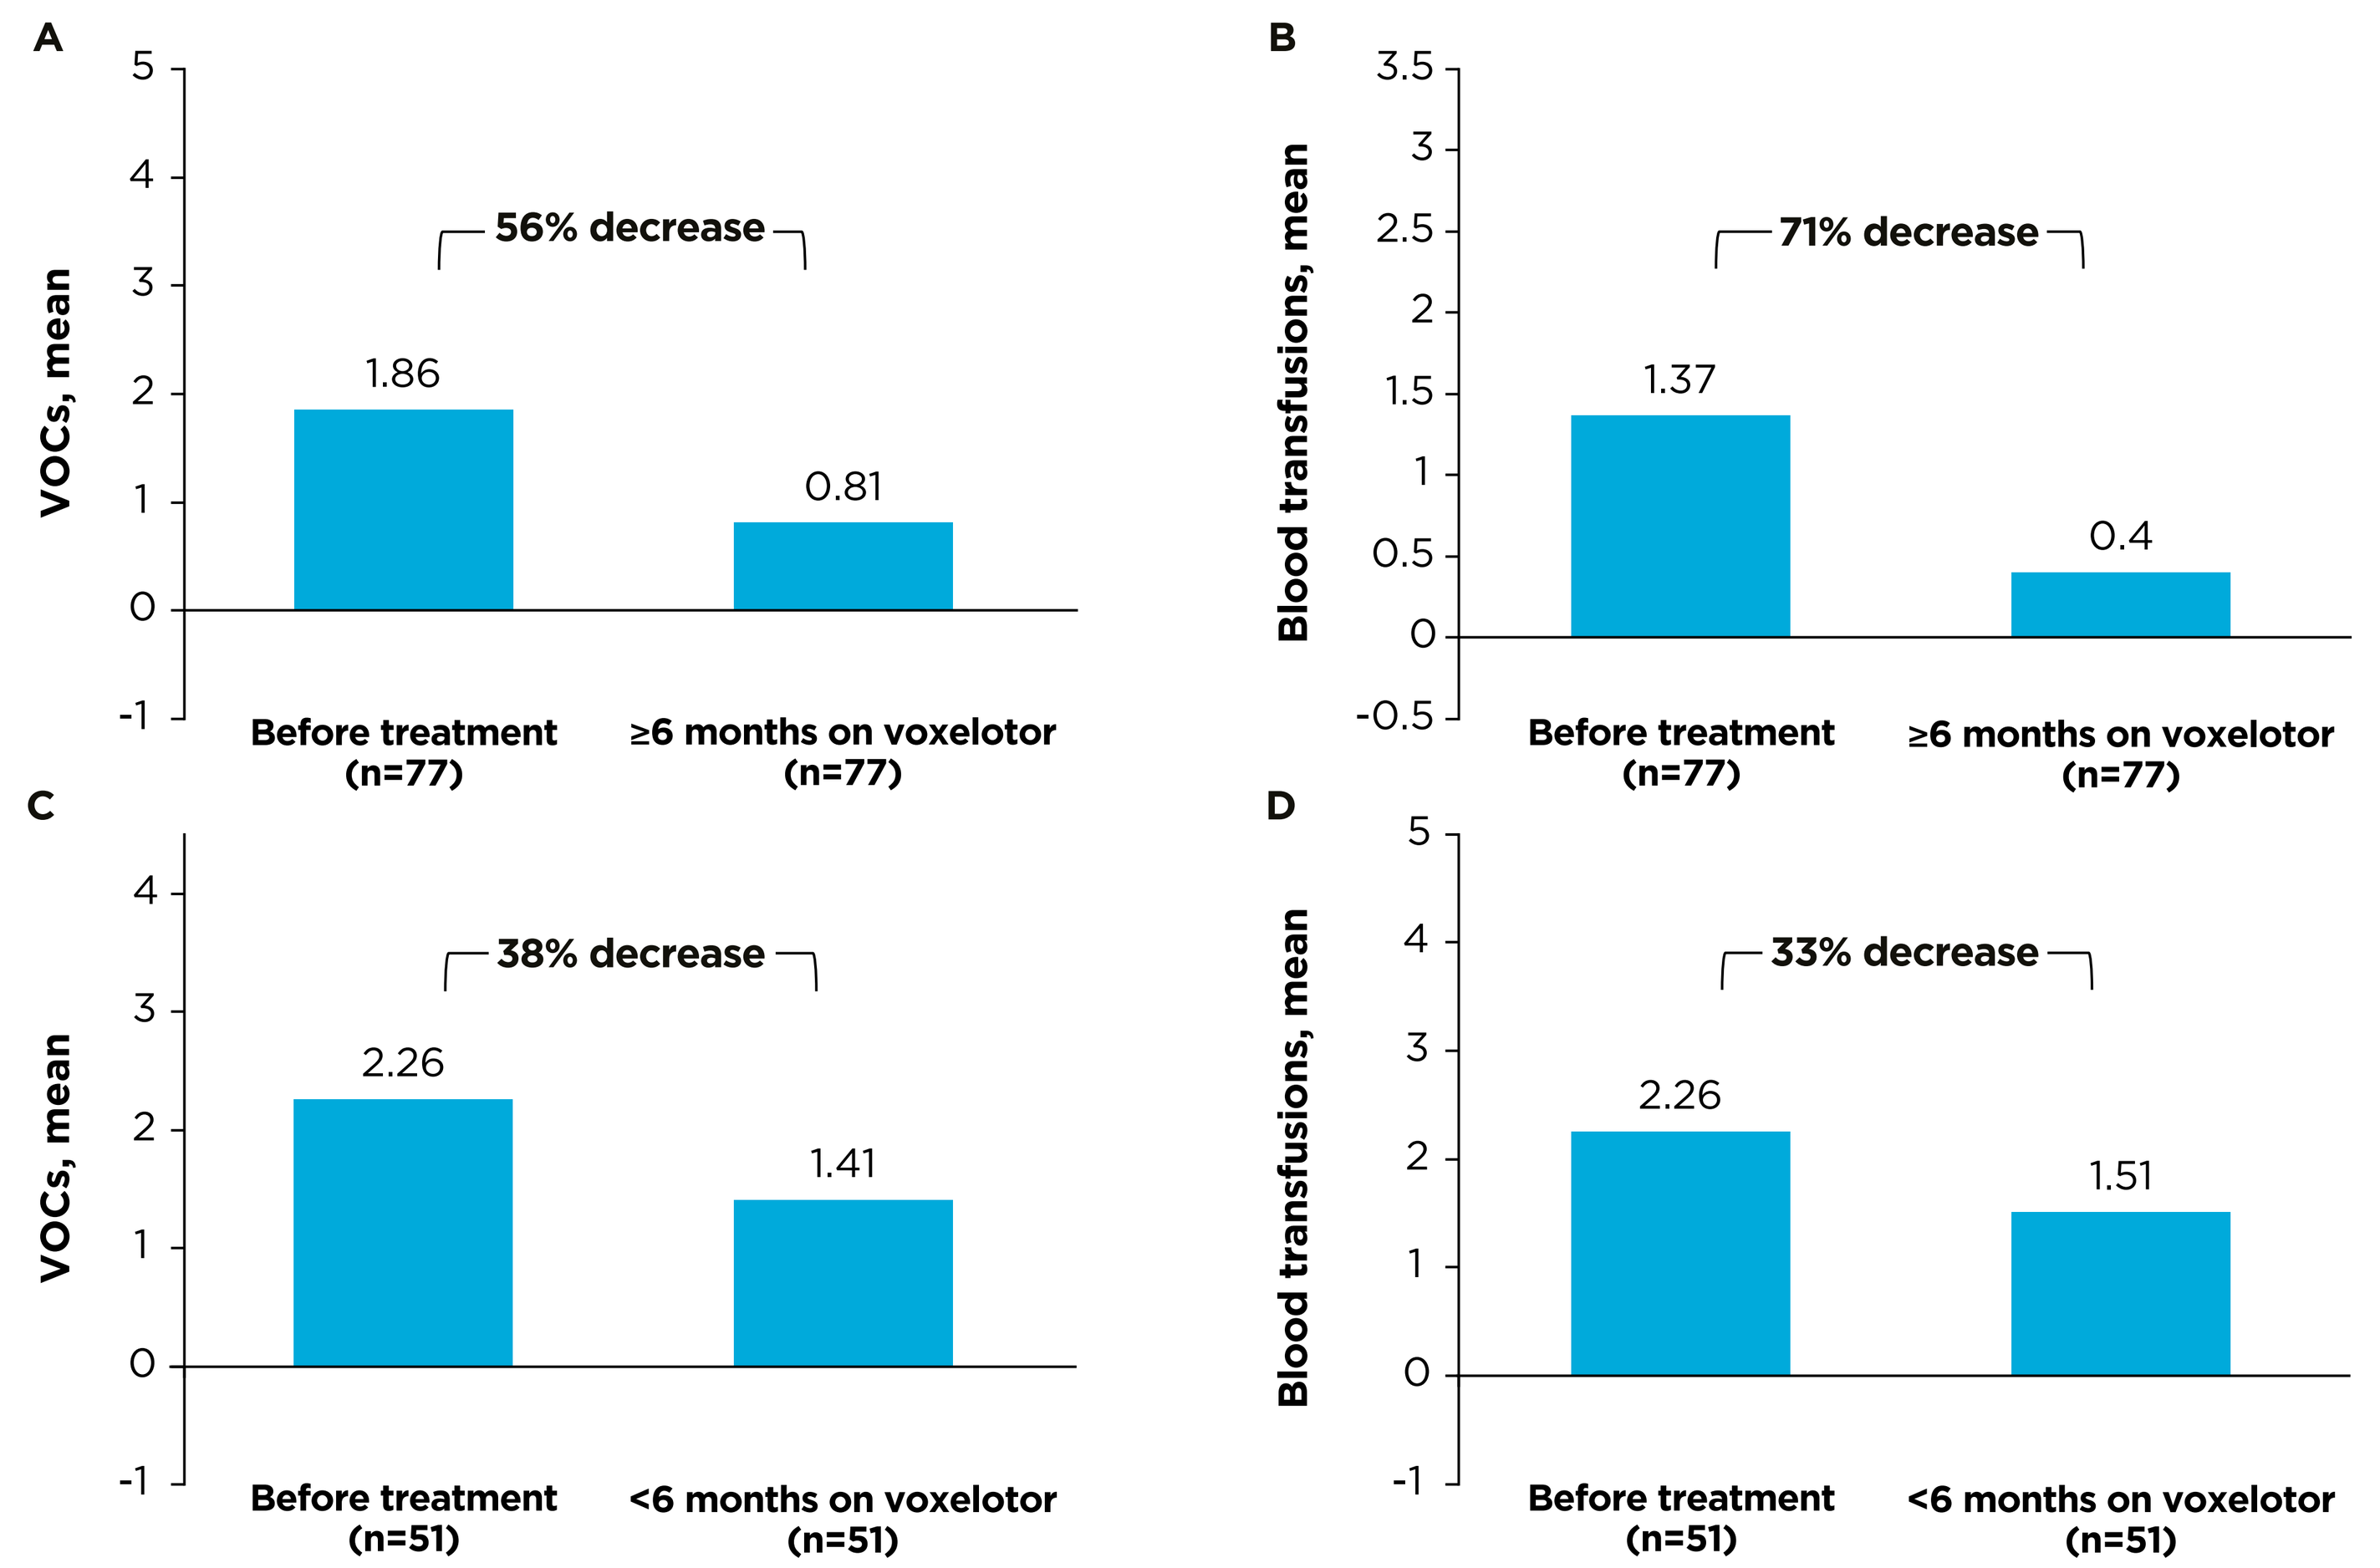
A
VOCs, mean
56% decrease
1.86
0.81
Before treatment (n=77)
≥6 months on voxelotor (n=77)
B
Blood transfusions, mean
71% decrease
1.37
0.4
Before treatment (n=77)
≥6 months on voxelotor (n=77)
C
VOCs, mean
38% decrease
2.26
1.41
Before treatment (n=51)
<6 months on voxelotor (n=51)
D
Blood transfusions, mean
33% decrease
2.26
1.51
Before treatment (n=51)
<6 months on voxelotor (n=51)

VOC, vaso-occlusive crisis.

QOL Improvements With Voxelotor

- Among patients who started voxelotor before their most recent visit and had physician-reported QOL assessments (**Figure 3**), 96% (127/132) showed symptomatic improvements across multiple domains:
 - 77% of patients (101/132) had less pain.
 - 77% of patients (101/132) had less fatigue.
 - 26% of patients (34/132) required fewer transfusions or stopped transfusions.
 - 16% of patients (21/132) had missed less work/school.
 - 4% of patients (5/132) had no symptom improvement.

Figure 3. Physician-Reported Improvement in QOL Assessments Among Voxelotor-Treated Patients

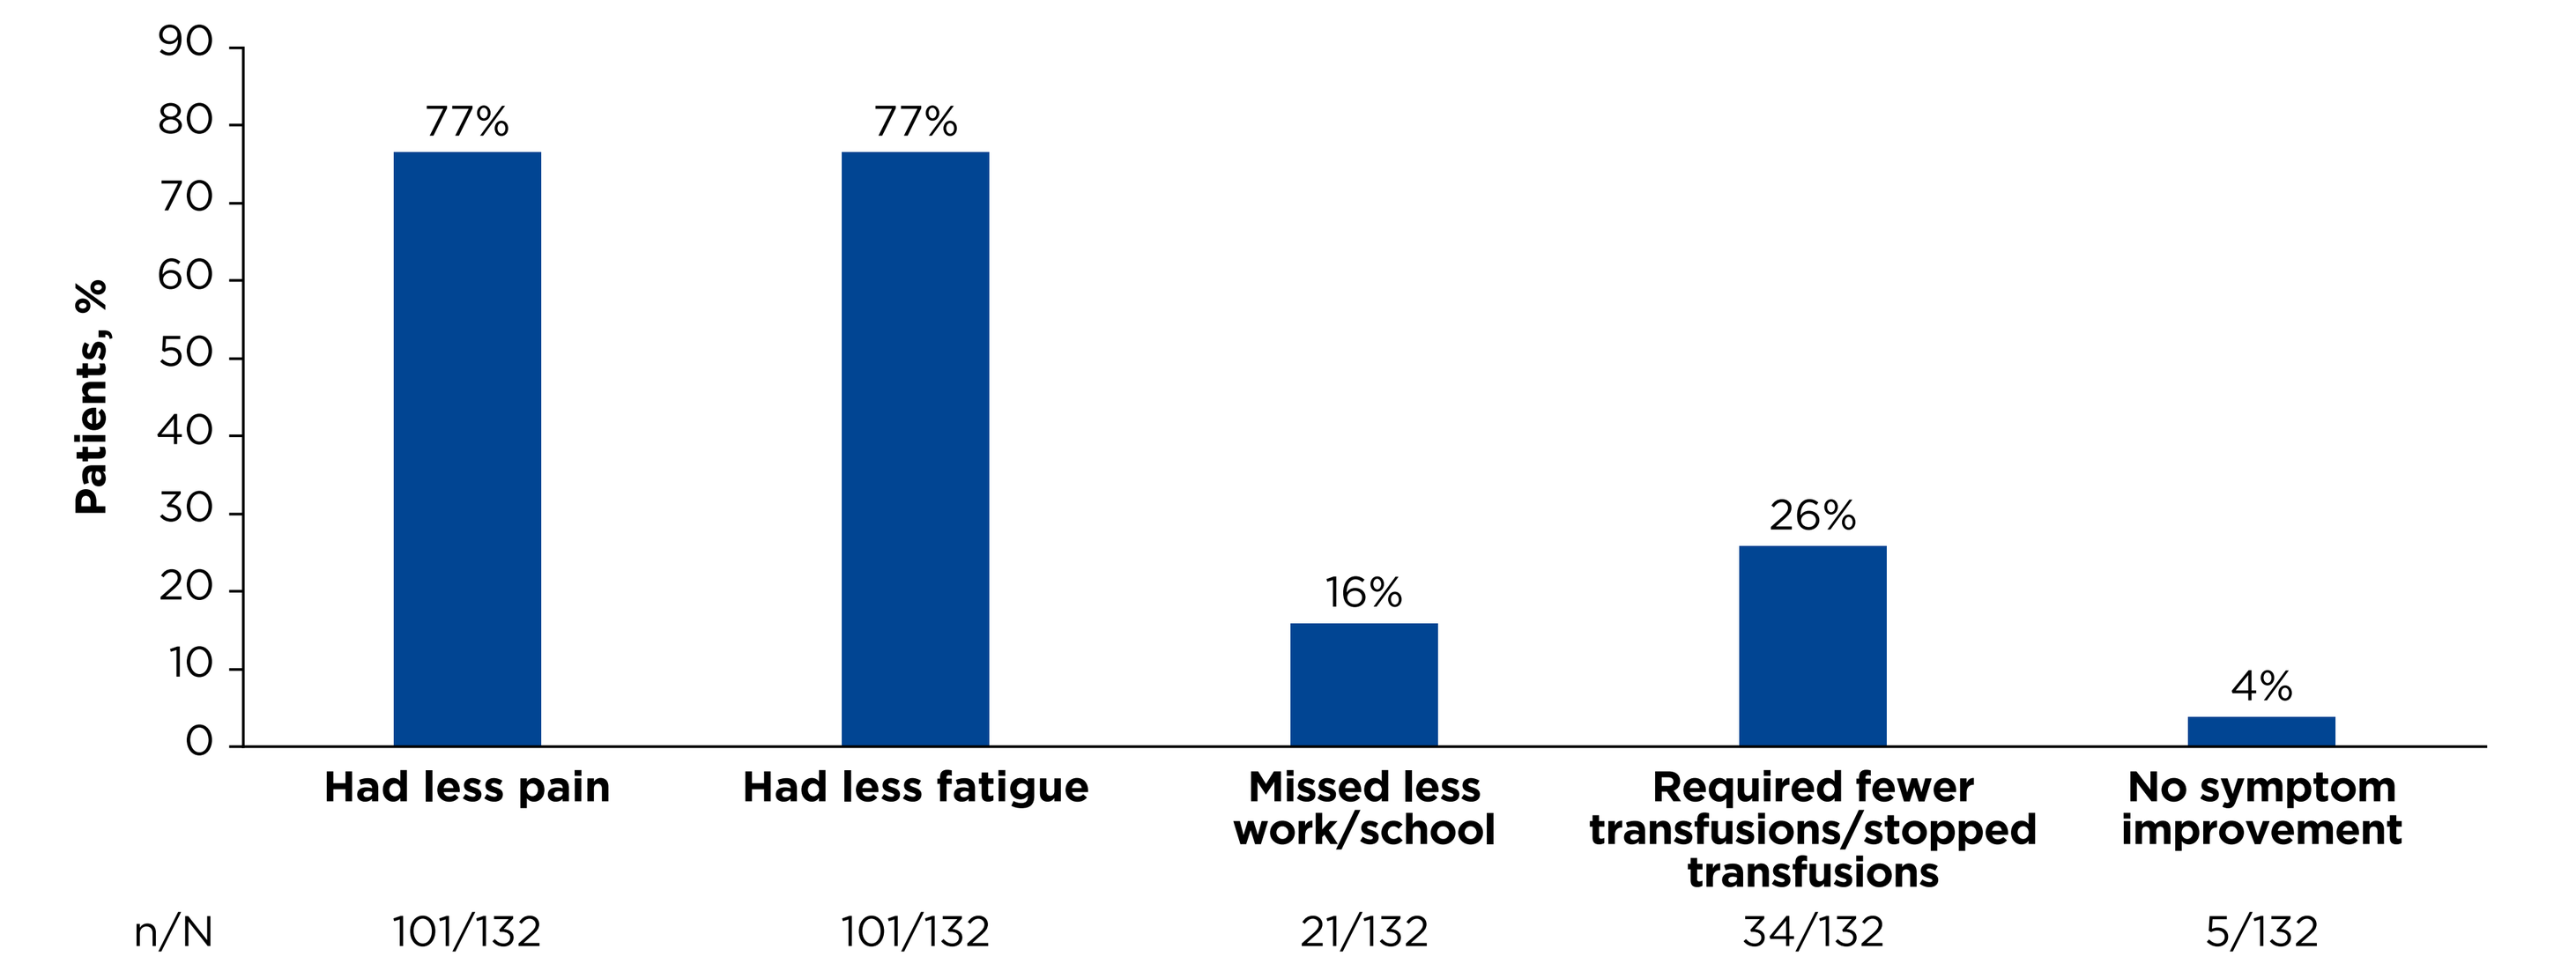
Patients, %
90
80
70
60
50
40
30
20
10
0
77%
77%
16%
26%
4%
Had less pain
Had less fatigue
Missed less work/school
Required fewer transfusions/stopped transfusions
No symptom improvement
n/N
101/132
101/132
21/132
34/132
5/132

Patients with SCD aged ≥12 years, receiving voxelotor as part of their current regimen and starting current treatment prior to most recent visit.
QOL, quality of life; SCD, sickle cell disease.

- Since initiating treatment with voxelotor:
 - Patients on average missed 5 fewer days of school or work.
 - Patients reported experiencing less pain (**Figure 4**).

Figure 4. Physician-Reported Improvement in Pain Scores Among Voxelotor-Treated Patients

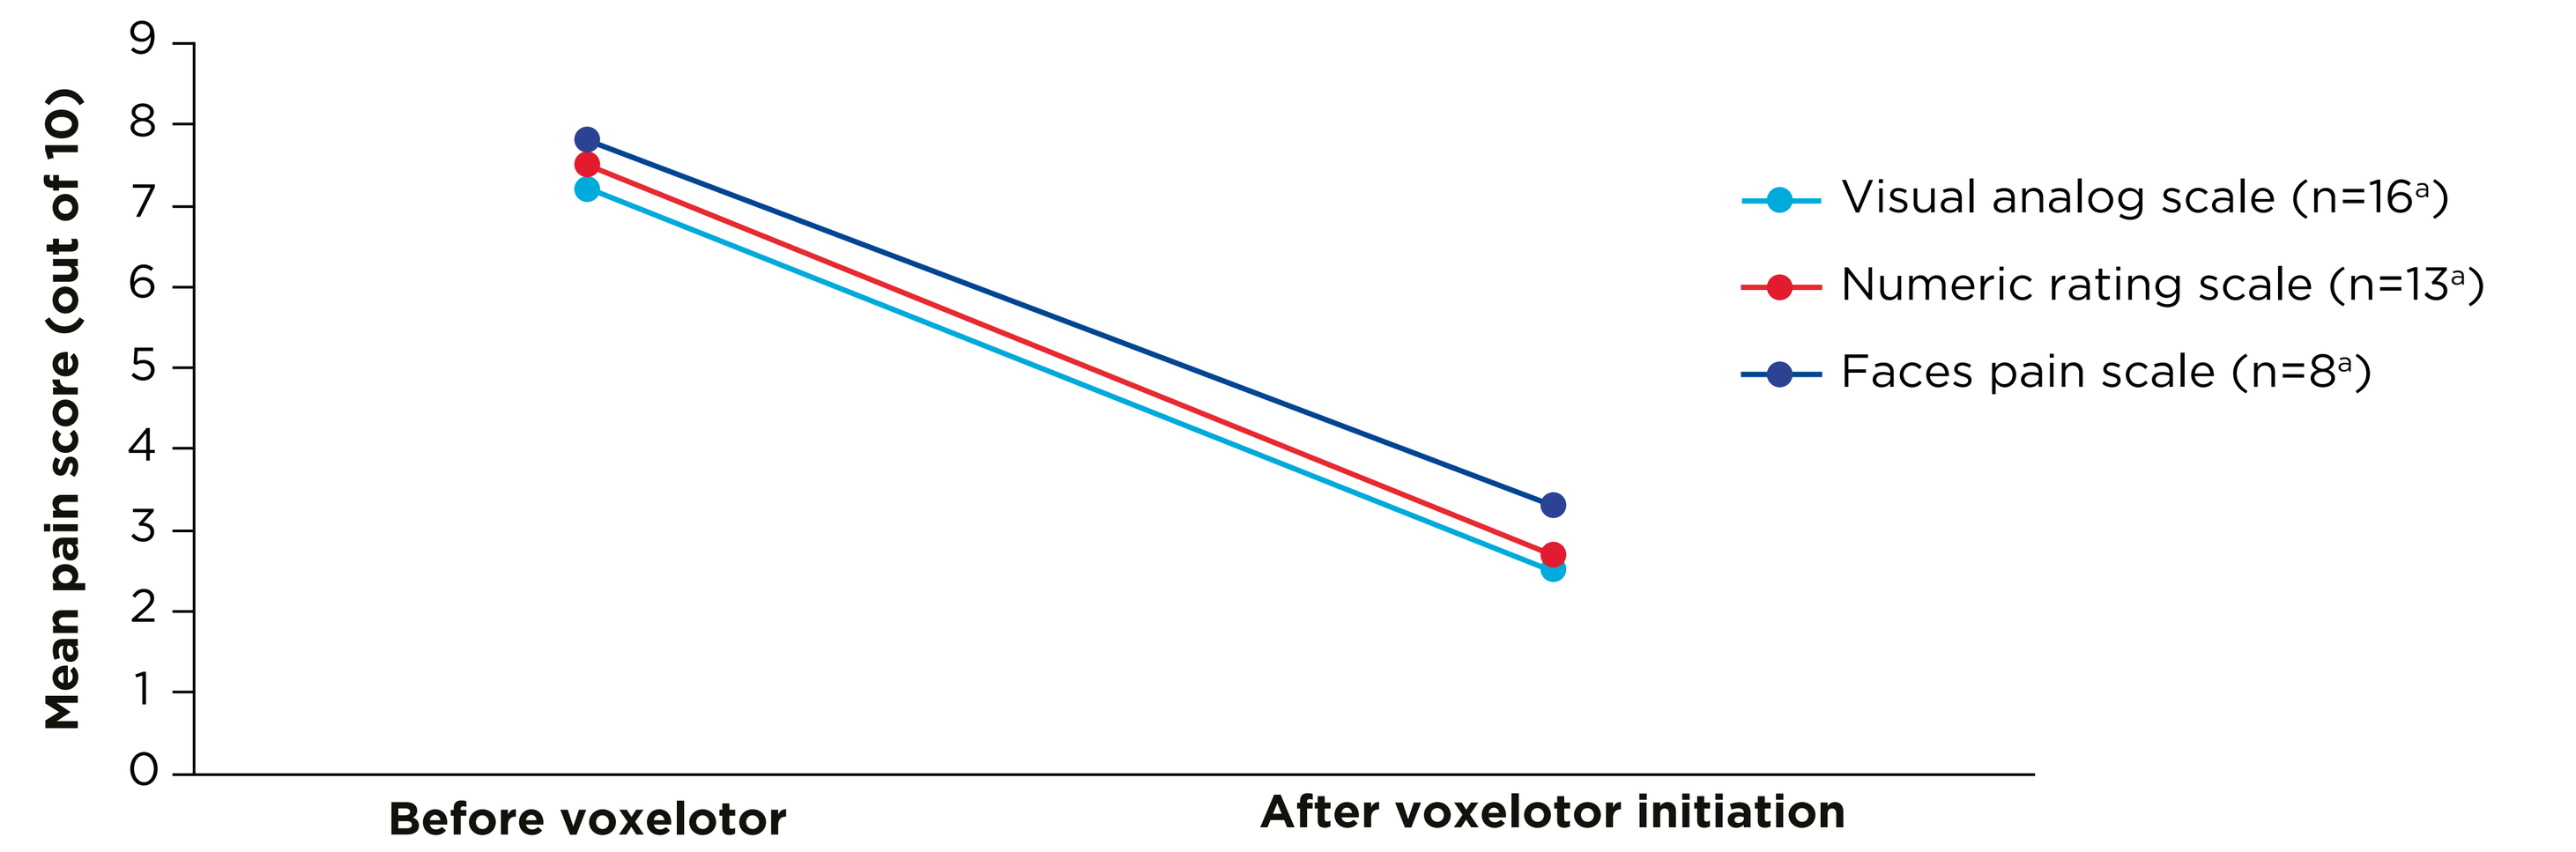
Mean pain score (out of 10)
9
8
7
6
5
4
3
2
1
0
Visual analog scale (n=16a)
Numeric rating scale (n=13a)
Faces pain scale (n=8a)
Before voxelotor
After voxelotor initiation

[a]Low base. Lower scores indicate less pain.

CONCLUSIONS

- In this multicenter retrospective chart review, patients with SCD experienced fewer VOCs and required fewer transfusions following voxelotor treatment initiation.
- According to their physicians, voxelotor-treated patients experienced improvements in SCD symptoms and other QOL indicators, including missing fewer days of work or school, since starting treatment.
- The data presented here complement findings from the pivotal Hemoglobin Oxygen Affinity Modulation to Inhibit HbS Polymerization (HOPE) trial[3] and previous real-world analyses that demonstrated improvements in patient health status and reduction in pain with voxelotor.[8-10]

ACKNOWLEDGMENTS

- We thank all the patients with sickle cell disease, families, caregivers, and investigators contributing to this study.
- Zeynep Turan, PhD (Healthcare Consultancy Group, funded by Global Blood Therapeutics), provided editorial and writing assistance.

DISCLOSURES

- **Ken Bridges:** Employee, equity ownership: Global Blood Therapeutics. **Nicole Allie:** Employee: Ipsos UK Ltd. **Kelly Kueh:** Employee: Ipsos Malaysia. **Dale Chandler:** Employee: Ipsos UK Ltd. **Clare Tong:** Employee, equity ownership: Global Blood Therapeutics. **Andy Nguyen:** Employee, equity ownership: Global Blood Therapeutics. **Anne Beaubrun:** Employee, equity ownership: Global Blood Therapeutics.
- This study was supported by Global Blood Therapeutics.

REFERENCES

1. Kato GJ, Piel FB, Reid CD, et al. Sickle cell disease. *Nat Rev Dis Primers*. 2018;4:18010.
2. Powars DR, Chan LS, Hiti A, Ramicone E, Johnson C. Outcome of sickle cell anemia: a 4-decade observational study of 1056 patients. *Medicine (Baltimore)*. 2005;84(6):363-376.
3. Vichinsky E, Telfer P, Inati A, et al. Incidence of vaso-occlusive crisis does not increase with achieving higher hemoglobin levels on voxelotor treatment or after discontinuation: analyses of the HOPE study. *Blood*. 2019;134(Supplement_1):2313.
4. Oxbryta. Prescribing information. Global Blood Therapeutics; December 2021.
5. Global Blood Therapeutics. MHRA grants marketing authorization for GBT's Oxbryta® (voxelotor) for use in Great Britain for the treatment of hemolytic anemia in patients with sickle cell disease age 12 years and older. Updated July 26, 2022. Accessed August 26, 2022. https://ir.gbt.com/news-releases/news-release-details/mhra-grants-marketing-authorization-gbts-oxbrytar-voxelotor-use
6. Data on File. Global Blood Therapeutics, South San Francisco, CA.
7. Sherman RE, Anderson SA, Dal Pan GJ, et al. Real-world evidence—what is it and what can it tell us? *N Engl J Med*. 2016;375(23):2293-2297.
8. Muschick K, Fuqua T, Stoker-Postier C, Anderson AR. Real-world data on voxelotor to treat patients with sickle cell disease. *Eur J Haematol*. 2022;109(2):154-161.
9. Andemariam B, Achebe M, Clay ELJ, et al. Real-world experience of patients with sickle cell disease treated with voxelotor: a multicenter, retrospective study. *Blood*. 2021;138:3100.
10. Shah N, Lipato T, Alvarez O, et al. Real-world effectiveness of voxelotor for treating sickle cell disease in the US: a large claims data analysis. *Expert Rev Hematol*. 2022;15(2):167-173.

Presented at AMCP Nexus; October 11-14, 2022; National Harbor, MD, USA.

© 2022 Global Blood Therapeutics.