Experience From the US Voxelotor Expanded Access Program for Children With Sickle Cell Disease

Clark Brown, MD, PhD[1]; Amma Owusu-Ansah, MBChB[2]; Elizabeth Yang, MD, PhD[3]; Amir Mian, MD[4]; Kenneth Rivlin, MD, PhD[5]; Clarissa Johnson, MD[6]; Sharon A. Singh, MD[7]; Sanjay Shah, MD[8]; Shalu Narang, MD[9]; Robin Miller, MD[10]; Deepika S. Darbari, MD[11]; Alan Anderson, MD[12,13]

[1]Children's Healthcare of Atlanta and the Department of Pediatrics, Emory University, Atlanta, GA, USA; [2]University Hospitals Rainbow Babies & Children's Hospital, Case Western Reserve University School of Medicine, Cleveland, OH, USA; [3]Pediatric Hematology and Oncology Division, Pediatric Specialists of Virginia, Fairfax, VA, USA; [4]Pediatric Hematology Oncology, Children's Hospital of Georgia, Augusta University/Georgia Cancer Center, Augusta, GA, USA; [5]Division of Pediatric Hematology and Oncology, Jacobi Medical Center, NYC Health and Hospitals, Bronx, NY, USA; [6]Division of Hematology and Oncology, Cook Children's Medical Center, Fort Worth, TX, USA; [7]Division of Pediatric Hematology/Oncology and Pediatrics, University of Michigan, Ann Arbor, MI, USA; [8]Division of Hematology/Oncology, Phoenix Children's Hospital, Phoenix, AZ, USA; [9]Division of Hemostasis and Thrombosis, Newark Beth Israel Medical Center & Children's Hospital of New Jersey, Newark, NJ, USA; [10]Nemours Center for Cancer and Blood Disorders, Nemours-Children's Hospital, Wilmington, DE, USA; [11]Center for Cancer and Blood Disorders, Children's National Hospital, Washington, DC, USA; [12]PRISMA Health Comprehensive SCD Program, Greenville, SC, USA; [13]University of South Carolina School of Medicine, Greenville, SC, USA

INTRODUCTION

VOXELOTOR IS AN ORAL, ONCE-DAILY SICKLE HEMOGLOBIN POLYMERIZATION INHIBITOR

Sickle cell disease (SCD) is an inherited systemic disorder in which sickle hemoglobin (HbS) polymerization triggers red blood cell sickling, chronic hemolytic anemia, and recurrent episodes of vaso-occlusion.[1]

SCD-related complications lead to acute and chronic life-threatening events, cumulative organ damage, disability, and early mortality.[2]

Efficacy and safety data from the randomized, placebo-controlled HOPE trial demonstrated the effectiveness of voxelotor in increasing hemoglobin (Hb) levels and reducing markers of hemolysis.[3,4]

Voxelotor, a first-in-class HbS polymerization inhibitor, is approved by the US Food and Drug Administration for the treatment of SCD in adults and pediatric patients aged ≥4 years and received marketing authorization in the European Union and Middle East for the treatment of patients with SCD aged ≥12 years.[5,6]

OBJECTIVE

- To report available results on the safety and efficacy of voxelotor from the expanded access program (EAP) that provided early access to voxelotor for children with SCD who had no alternative options to improve their Hb other than standard treatment

METHODS

Design and Participants

- The EAP is an open-label, real-world program (NCT04724421) that enrolled children aged 4 to 11 years with confirmed SCD of any genotype and a baseline Hb ≤10.5 g/dL to receive once-daily, weight-based oral dosing of voxelotor in addition to standard of care (**Figure 1**); treatment continued until voxelotor became commercially available or the sponsor discontinued the voxelotor EAP.
- Eligible patients were those who were ineligible or unable to participate in actively recruiting clinical studies of voxelotor and had no alternative treatment options, per the treating physician's judgment.
- Patients receiving regular red blood cell transfusions for primary or secondary stroke prevention, as well as those with hepatic dysfunction (characterized by alanine aminotransferase >4 × the upper limit of normal for age) or severe renal dysfunction (estimated glomerular filtration rate <30 mL/min/1.73 m² by Schwartz formula), were excluded.
- Clinical assessments of laboratory parameters (eg, hematological assessments), concomitant medication use, requirement of transfusions, and health status (ie, Clinical Global Impression of Change [CGI-C], Patient Global Impression of Change [PGI-C] questionnaires) were performed every 12 weeks during routine office visits.[7,8]
- Voxelotor safety and overall clinical response were also monitored.
- Data collection and site monitoring were limited.
- The data cutoff for analyses performed in this presentation was February 28, 2022.

Figure 1. EAP for Children With SCD Aged 4 to 11 Years

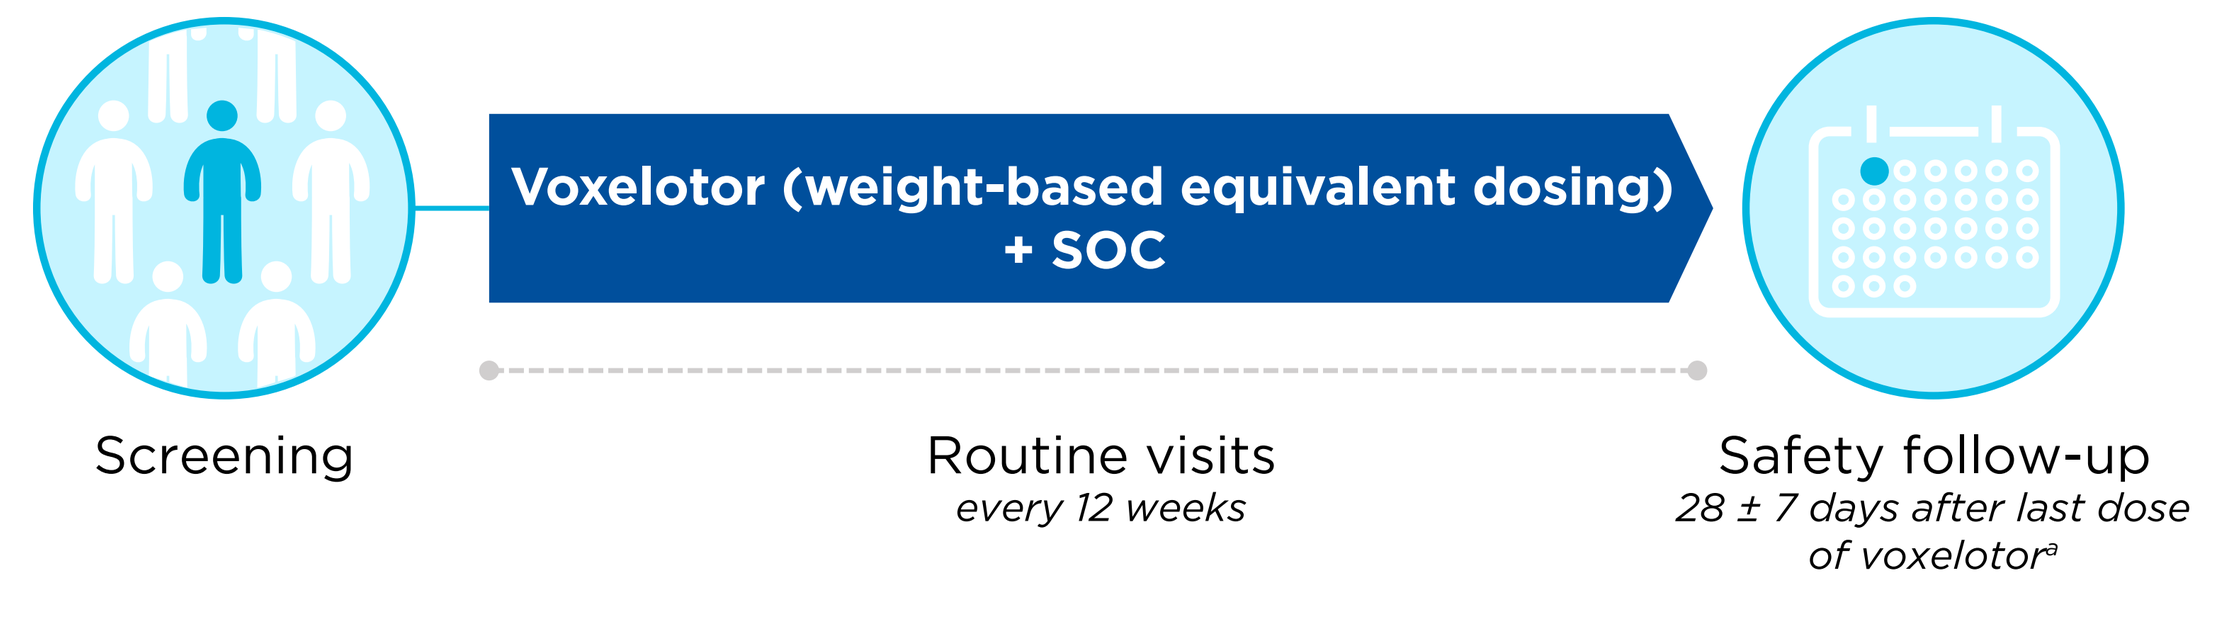
Voxelotor (weight-based equivalent dosing) + SOC
Screening
Routine visits
every 12 weeks
Safety follow-up
28 ± 7 days after last dose of voxelotor[a]

Dosing (dispersible tablets[b])

Age 4-11 years	Dosing
10 to <20 kg	600 mg
20 to <40 kg	900 mg
≥40 kg	1500 mg

[a]Treatment ended with commercial availability of voxelotor.
[b]Dispersible tablets are preservative-free, uncoated tablets that require no special skills or equipment for dispersal and can be dosed across the entire pediatric population, because dispersal is performed before administration.
EAP, expanded access program; SCD, sickle cell disease; SOC, standard of care.

RESULTS

Demographics and Baseline Characteristics

- Patients from 16 sites in the United States were enrolled.
- A total of 87 patients received at least 1 dose of the drug; 60 patients completed the 12-week follow-up visit.
- Most patients were Black or African American with the sickle cell anemia (HbSS) genotype (**Table 1**).
- The main reason physicians chose to administer voxelotor was low Hb counts despite adherence to treatment with hydroxyurea (80%; n=68/85); other reasons (32%, n=27/85) included complications or continued hemolysis with hydroxyurea treatment, the need to improve quality of life, reduction of the need for pain medication, the need to increase Hb or oxygen-carrying capacity/oxygen saturation, and prevention of potential pulmonary hypertension.

Table 1. Patient Demographics and Clinical Characteristics at Program Entry

	Patients (N=87)
Age at screening, years	
Mean (SD; range)	7.9 (2.23; 4-11)
Weight at screening, kg	
Mean (SD; range)	27.2 (7.98; 13-51)
Sex, n (%)	
Female	35 (40.2)
Male	52 (59.8)
Race, n (%)	
Black or African American	83 (95.4)
White	1 (1.1)
Multiracial	1 (1.1)
Not reported	2 (2.3)
Baseline Hb	
Mean (SD; range)	8.3 (1.13; 5.4-10.3)
Baseline HbF (%)[a]	
Mean (SD; range)	17.6 (11.14; 2.6-39.2)
HbS genotypes, n (%)	
HbSS	81 (93.1)
HbSβ⁰	2 (2.3)
HbSC	3 (3.4)
No. of blood transfusions in the previous 12 months, n (%)	
0	54 (62.1)
1	9 (10.3)
2	10 (11.5)
3	4 (4.6)
4	2 (2.3)
5	2 (2.3)
6	1 (1.1)
≥7	4 (4.6)

	Patients (N=87)
Baseline HU/hydroxycarbamide use, n (%)	
Yes	74 (85.1)
No	13 (14.9)
No. of VOCs in the previous 12 months requiring hospitalization, n (%)	
0	58 (66.7)
1	9 (10.3)
2	5 (5.7)
3	5 (5.7)
4	2 (2.3)
5	2 (2.3)
6	2 (2.3)
≥7	3 (3.4)
Historical SCD complications, n (%)[b]	
Pain crisis	30 (34.5)
Acute chest syndrome	24 (27.6)
Scleral icterus	22 (25.3)
Splenic sequestration	19 (21.8)
Dactylitis	12 (13.8)
Cholecystectomy	3 (3.4)
Pulmonary hypertension	2 (2.3)
Hepatic sequestration	1 (1.1)
Stroke	1 (1.1)
Other	45 (51.7)
Concurrent SCD complications, n (%)	
Scleral icterus	16 (18.4)
Pulmonary hypertension	2 (2.3)
Cholecystectomy	1 (1.1)
Splenic sequestration	1 (1.1)
Other	39 (44.8)

[a]N=50.
[b]Patients may have reported multiple complications.
Hb, hemoglobin; HbF, fetal hemoglobin; HbS, sickle hemoglobin; HbSβ⁰, sickle beta zero thalassemia; HbSC, hemoglobin SC disease; HbSS, homozygous for SCD; HU, hydroxyurea; SCD, sickle cell disease; VOC, vaso-occlusive crisis.

Figure 2. Hb Levels in Most Patients Improved With Voxelotor at Week 12[a]

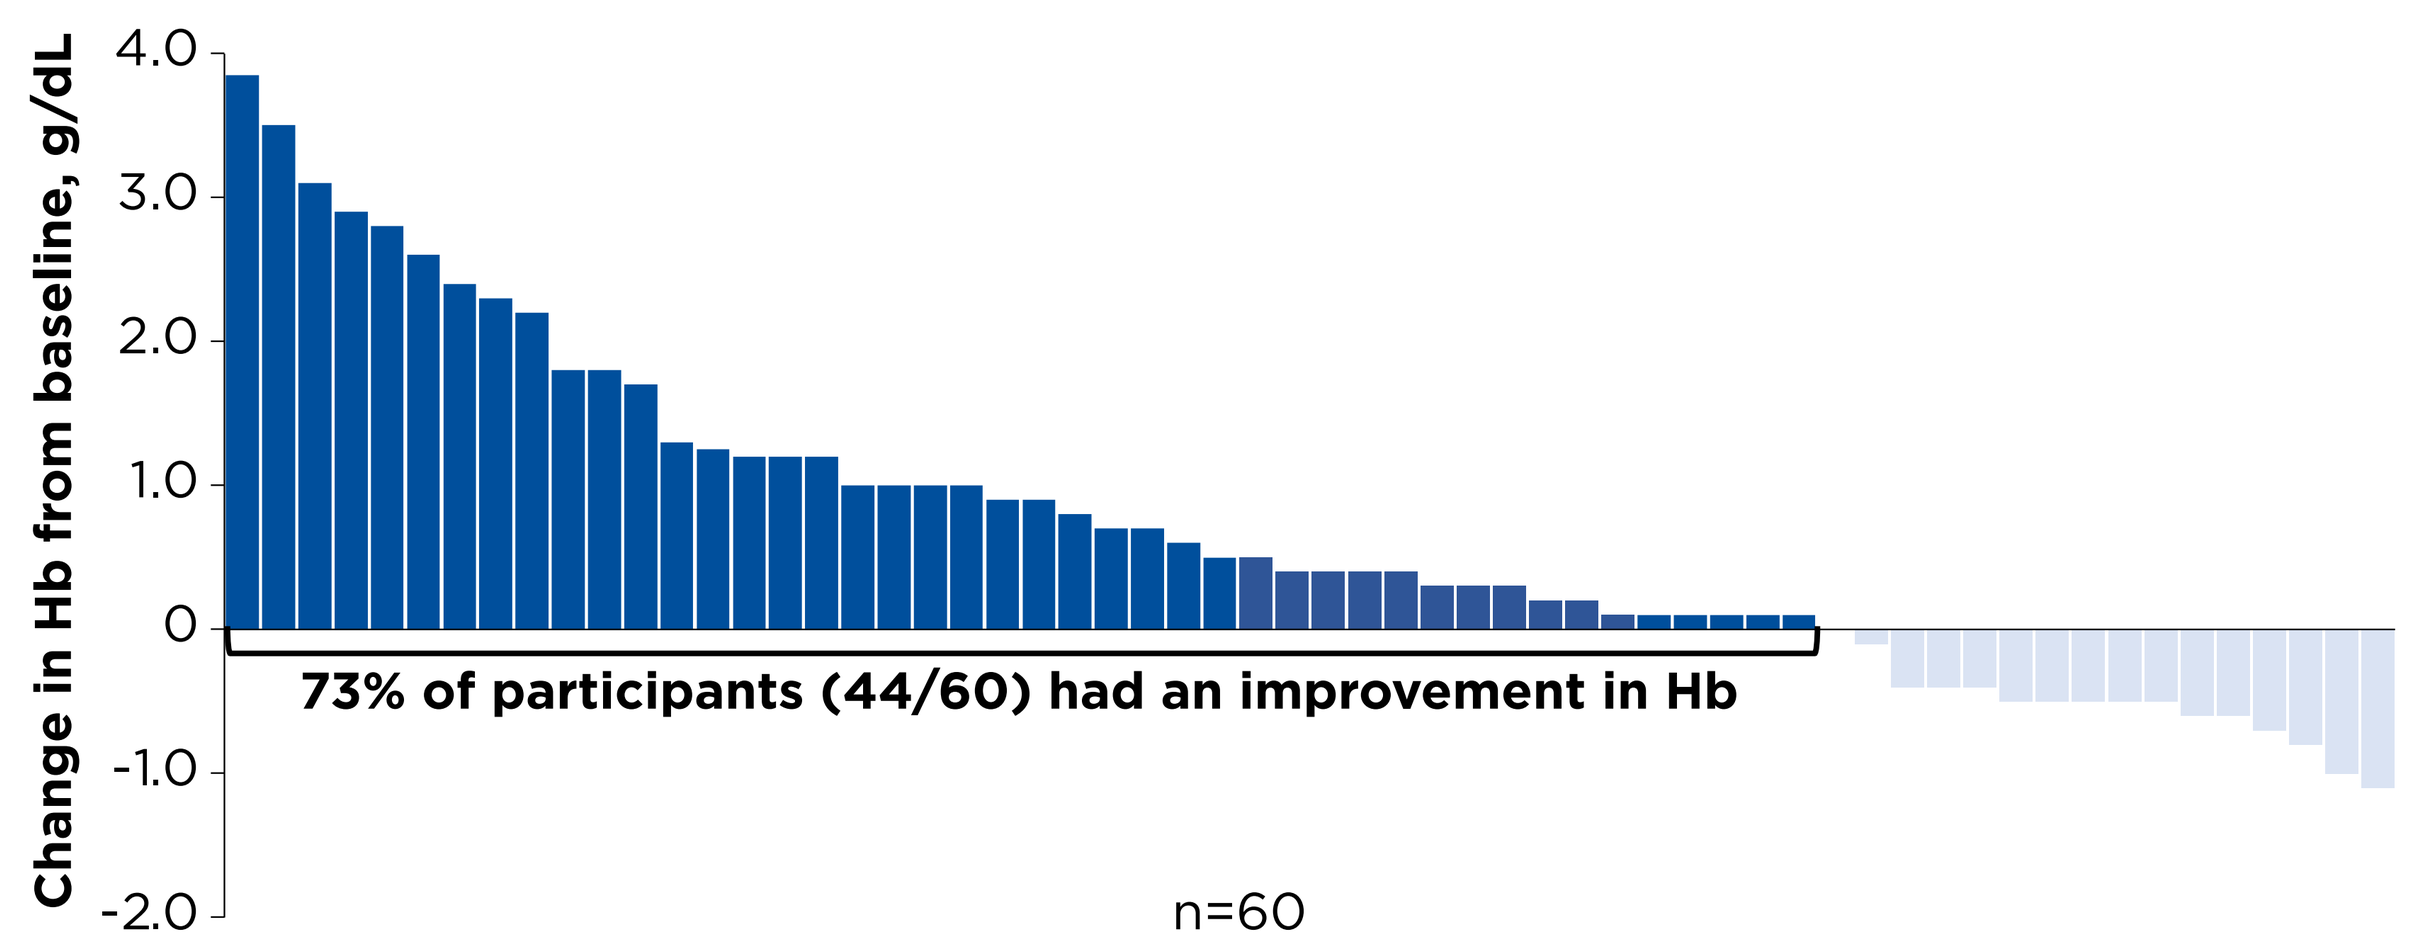
Change in Hb from baseline, g/dL
4.0
3.0
2.0
1.0
0
-1.0
-2.0
73% of participants (44/60) had an improvement in Hb
n=60

[a]As of February 28, 2022, 60 patients completed the 12-week follow-up.
Hb, hemoglobin.

- Among the 60 patients who completed a 12-week follow-up visit, 44 (73%) had improvements in Hb levels (**Figure 2**).
- Markers of hemolysis also improved in many patients after 12 weeks of treatment with voxelotor (**Figure 3**).

Figure 3. Improvement in Hemolysis Markers at Week 12[a]

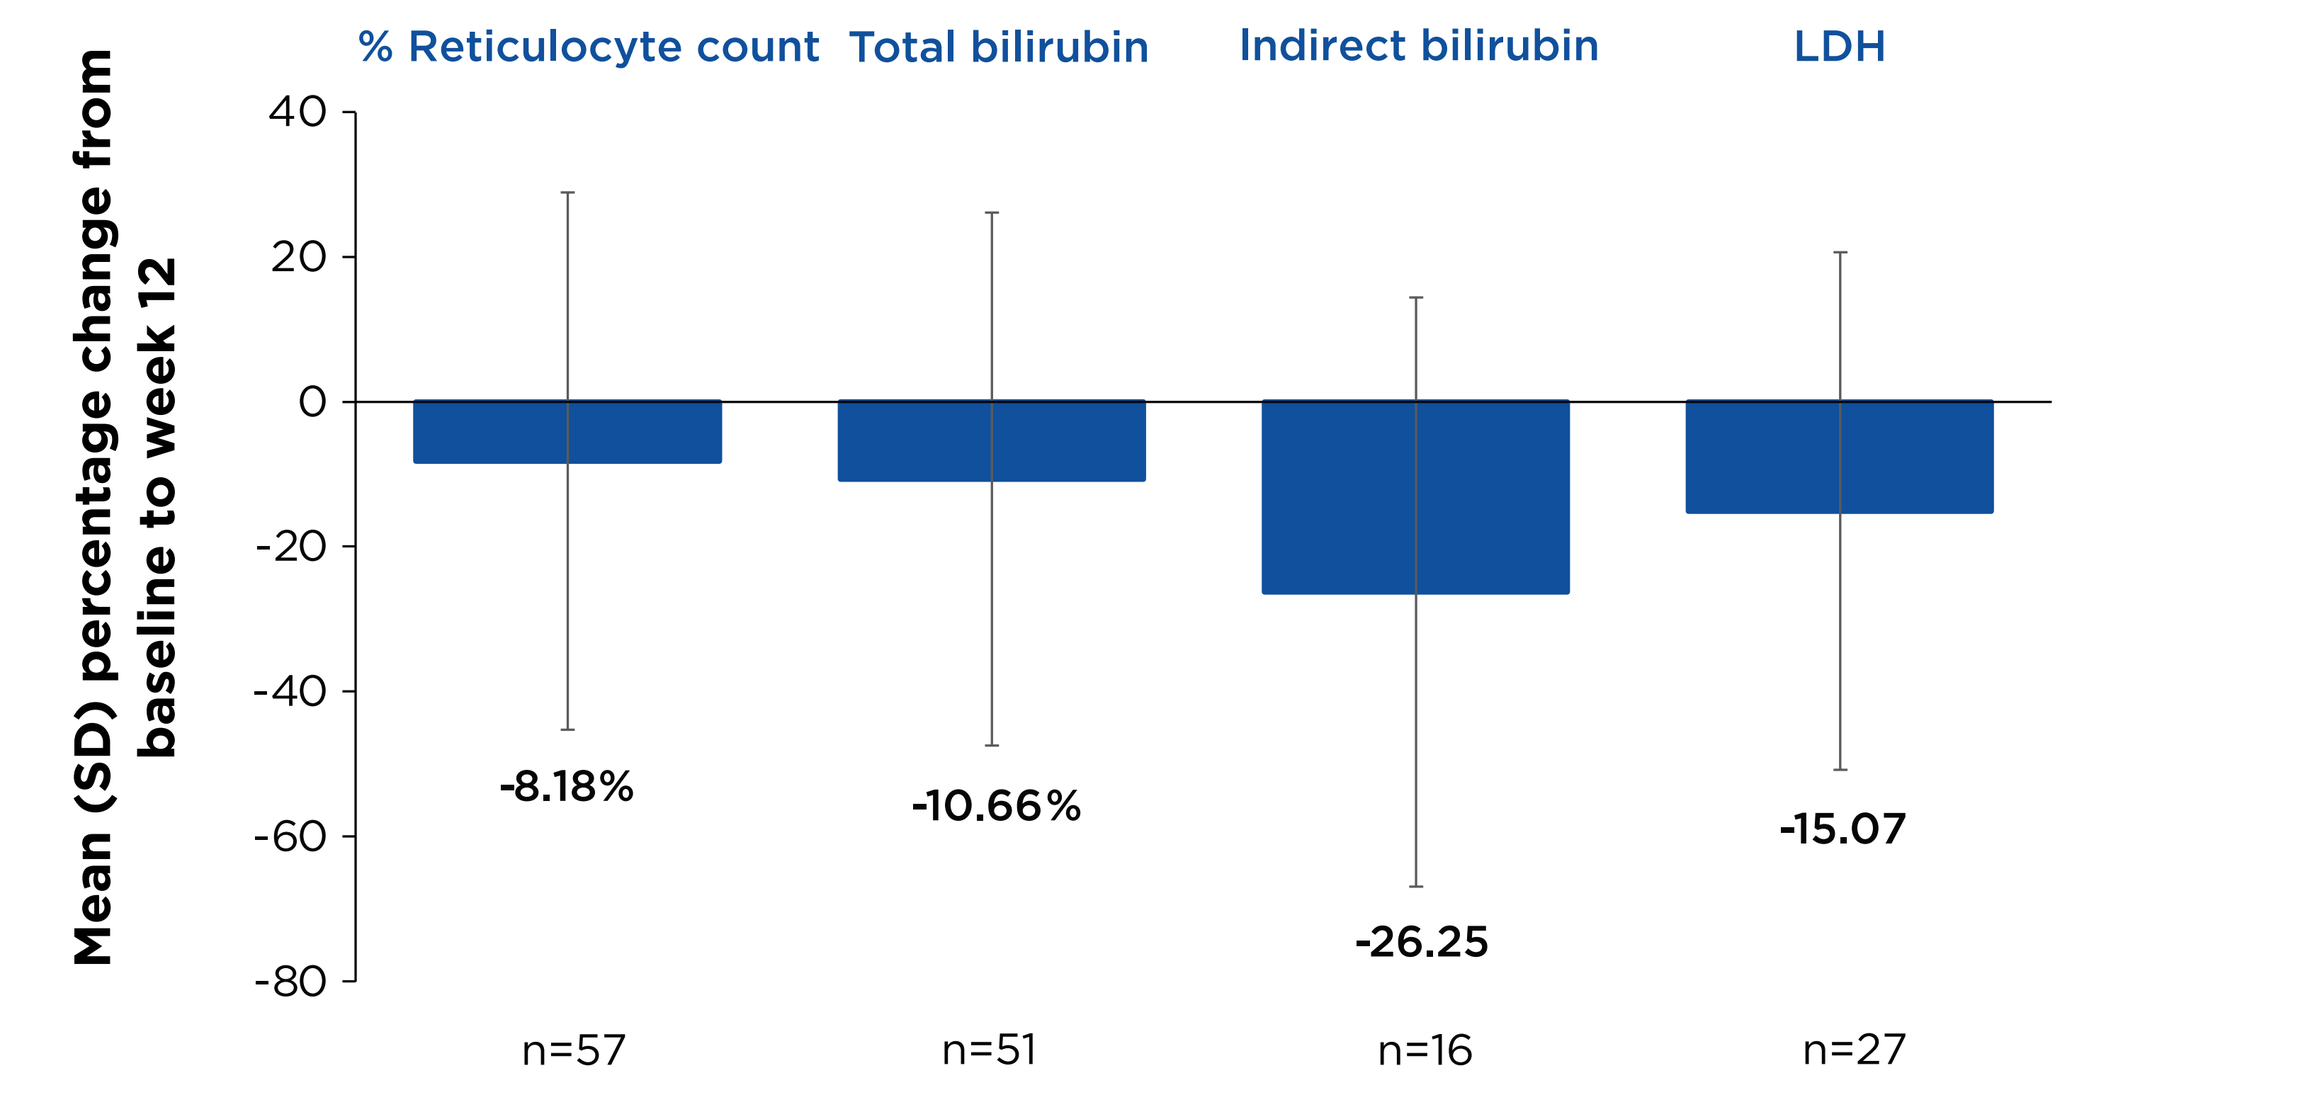
Mean (SD) percentage change from baseline to week 12
% Reticulocyte count
Total bilirubin
Indirect bilirubin
LDH
40
20
0
-20
-40
-60
-80
-8.18%
-10.66%
-26.25
-15.07
n=57
n=51
n=16
n=27

[a]As of February 28, 2022, 60 patients completed the 12-week follow-up.
LDH, lactate dehydrogenase; SD, standard deviation.

- Compared with baseline, most patients and clinicians considered patients' symptoms improved at treatment week 12; CGI-C scores improved in 68% (40/59) of patients and PGI-C scores improved in 63% (37/59) of patients (**Table 2**).

Table 2. Voxelotor Improved Patient Health Status at Week 12

	CGI-C	PGI-C
	n=59	n=59
(1) Very much improved	8 (13.6%)	12 (20.3%)
(2) Much improved	15 (25.4%)	10 (16.9%)
(3) Minimally improved	17 (28.8%)	15 (25.4%)
(4) No change	19 (32.2%)	22 (37.3%)
(5) Minimally worse	0	0
(6) Much worse	0	0
(7) Very much worse	0	0

CGI-C, Clinical Global Impression of Change; PGI-C, Patient Global Impression of Change.

- Voxelotor was generally well tolerated, with only 7% (n=6) of patients discontinuing due to a treatment-emergent adverse event (**Table 3**); treatment-emergent adverse events that led to discontinuation were rash/hives, arthralgia, face edema, abdominal pain, loss of appetite/anorexia, and diarrhea.
- The most common drug-related treatment-emergent adverse events were nausea, abdominal pain, vomiting, headache, and diarrhea (**Table 3**).

Table 3. Safety Summary

TEAEs assessed[a] as related to voxelotor, unrelated to SCD (reported in ≥5% of patients)	Patients (N=87)
Patients with ≥1 TEAE, n (%)	**33 (37.9)**
Nausea	10 (11.5)
Abdominal pain	7 (8.0)
Vomiting	7 (8.0)
Headache	6 (6.9)
Diarrhea	5 (5.7)
Voxelotor dose discontinuations and modifications due to adverse events, n (%)	
Dose reduction	2 (2.3)
Dose interruption	8 (9.2)
Discontinuation	6 (6.9)

[a]As assessed by the treating physician.
SCD, sickle cell disease; TEAE, treatment-emergent adverse event.

- Overall, a positive association between reported medication adherence and clinical improvement was observed.
- Although several challenges may have prevented some program participants from taking voxelotor consistently as prescribed, certain best practices were identified to improve treatment compliance and adherence (**Table 4**).

Table 4. Best Practices Derived From the EAP

Participant expectations

- Participants/caregivers should be informed that to receive potential clinical benefit, they are expected to adhere to the prescribed dosing regimen: **oral, once-daily administration of voxelotor** in the form of a dispersible tablet for oral suspension.
- **In the case of a missed dose, normal dosing must be resumed the next day.**
- As appropriate, this message needs to be reinforced.

Motivation/participant engagement

- Participants/caregivers need to be followed up between visits, especially after the initial visit.
- Sharing lab results may encourage participants/caregivers to continue treatment.

Taste/texture

- For younger patients, using a syringe to administer oral suspension of voxelotor may be helpful.
- Oral suspension aftertaste may be reduced by drinking more of the liquid in which voxelotor was suspended or eating pleasant-tasting food.
- Suspend voxelotor in clear liquid that is pleasant for the patient. Options that have worked include **Sprite, lemonade, apple juice, and white grape juice.**

Adverse events

- Patients should be informed that drug-related adverse events, including diarrhea, nausea, and abdominal pain, are likely but manageable.
- Adverse events can be mitigated (eg, taking voxelotor at night with food for nausea or taking loperamide for diarrhea).
- Dose can be temporarily modified as needed if the initial mitigation fails.

EAP, expanded access program.

CONCLUSIONS

Preliminary findings of the expanded access program indicate that voxelotor treatment was associated with increased Hb levels, reductions in markers of hemolysis, and improvements in both CGI-C and PGI-C scores.

Certain best practices were identified to improve treatment compliance and adherence among expanded access program participants, including taking voxelotor with food and dispersing tablets in clear beverages.

The safety profile of voxelotor among expanded access program participants was consistent with previous reports in adult and adolescent patients.

Further evaluation of data from the expanded access program, with additional data from all study sites and longer follow-up, is planned.

REFERENCES

1. Kato GJ, Piel FB, Reid CD, et al. Sickle cell disease. *Nat Rev Dis Primers*. 2018;4:18010.
2. Ware RE, de Montalembert M, Tshilolo L, Abboud MR. Sickle cell disease. *Lancet*. 2017;390(10091):311-323.
3. Vichinsky E, Hoppe CC, Ataga KI, et al. A phase 3 randomized trial of voxelotor in sickle cell disease. *N Engl J Med*. 2019;381(6):509-519.
4. Howard J, Ataga KI, Brown RC, et al. Voxelotor in adolescents and adults with sickle cell disease (HOPE): long-term follow-up results of an international, randomised, double-blind, placebo-controlled, phase 3 trial. *Lancet Haematol.* 2021;8(5):e323-e333.
5. Oxbryta. Prescribing information. Global Blood Therapeutics; December 2021.
6. European Commission approves Oxbryta® (voxelotor) for the treatment of hemolytic anemia in patients with sickle cell disease age 12 years and older. News release. Global Blood Therapeutics. February 16, 2022. Accessed February 16, 2022. https://ir.gbt.com/news-releases/news-release-details/european-commission-approves-oxbrytar-voxelotor-treatment
7. Busner J, Targum SD. The clinical global impressions scale: applying a research tool in clinical practice. *Psychiatry (Edgmont).* 2007;4(7):28-37.
8. Idowu M, Haque A, Williams EM, et al. Patient perception of voxelotor treatment benefit in sickle cell disease. *J Invest Med.* 2022. In Press.

ACKNOWLEDGMENTS

- **We thank all the patients with sickle cell disease, families, caregivers, research nurses, study coordinators, and support staff who contributed to this study.**

DISCLOSURES

- **Clark Brown:** Consultant: Global Blood Therapeutics, Imara, Novartis, Novo Nordisk; research support: Global Blood Therapeutics, Forma Therapeutics, Imara, Novartis, Pfizer. **Amma Owusu-Ansah:** Nothing to disclose. **Elizabeth Yang:** Nothing to disclose. **Amir Mian:** Nothing to disclose. **Kenneth Rivlin:** Consultant: Global Blood Therapeutics, Pfizer; research support: Global Blood Therapeutics, Pfizer, Novartis. **Clarissa Johnson:** Nothing to disclose. **Sharon A. Singh:** Consultant: Global Blood Therapeutics, Emmaus Medical. **Sanjay Shah:** Nothing to disclose. **Shalu Narang:** Advisory board member: Genentech. **Robin Miller:** Nothing to disclose. **Deepika S. Darbari:** Consultant: Global Blood Therapeutics, Novartis, Hilton Publishing. **Alan Anderson:** Consultant: Global Blood Therapeutics; research support: Global Blood Therapeutics.

- Healthcare Consultancy Group provided medical writing, editorial assistance, and creative design, funded by Global Blood Therapeutics.
- This study was supported by Global Blood Therapeutics.

Presented at the 2022 American Society of Pediatric Hematology/Oncology (ASPHO) Conference; May 4-7, 2022; Pittsburgh, PA, USA

© 2022 Global Blood Therapeutics