Voxelotor Treatment Is Associated With Decreased Usage of Iron Chelation, Opioids, and Erythropoietin-Stimulating Agents in Patients With Sickle Cell Disease

Nirmish Shah, MD[1]; Thokozeni Lipato, MD[2]; Ofelia Alvarez, MD[3]; Thomas E. Delea, MSIA[4]; Nhat Pham[4]; Alexander Lonshteyn[4]; Derek Weycker, PhD[4]; Andy Nguyen, PhD[5]; Anne Beaubrun, PhD[5]

[1]Duke University School of Medicine, Durham, NC, USA; [2]VCU Health, Richmond, VA, USA; [3]University of Miami Miller School of Medicine, Miami, FL, USA; [4]Policy Analysis Inc. (PAI), Brookline, MA, USA; [5]Global Blood Therapeutics, South San Francisco, CA, USA

GBT

OBJECTIVE

To assess healthcare resource utilization related to sickle cell disease (SCD) management, defined by use of iron chelation therapy, opioid prescription days, and erythropoietin-stimulating agent (ESA) use, in voxelotor-treated patients in the real world.

BACKGROUND

- SCD, an inherited disorder associated with sickle hemoglobin polymerization, results in chronic anemia, hemolysis, and vaso-occlusive crises.[1]
 - Management of SCD complications leads to substantial healthcare cost burden.
- Oxbryta® (voxelotor) tablets were approved by the US Food and Drug Administration in November 2019 for treatment of patients with SCD aged ≥12 years based on results from the pivotal HOPE trial.[2]
 - Voxelotor is a sickle hemoglobin–polymerization inhibitor that significantly increased hemoglobin levels and reduced markers of hemolysis in patients in the HOPE trial.[3]
- A prior analysis of real-world data found reduced rates of transfusions and vaso-occlusive crises in patients after voxelotor treatment.[4]

METHODS

- Medical and pharmacy claims data for patients aged ≥12 years on voxelotor from December 2019 to December 2020 were obtained from the Symphony Health Claims database.
 - Those with ≥1 year's data before the first claim for voxelotor (index date) were included.
- The total day's supplies over a 3-month (90-day) period of iron chelation therapy and of opioid prescription were adjusted for follow-up and compared for the post-index and 3-month pre-index periods.
- The rate of ESA use was assessed in patients with any ESA use 3 months pre-index.

RESULTS

PATIENT DEMOGRAPHICS

A total of 2299 patients receiving voxelotor met the inclusion criteria (Table 1).

Mean follow-up was 109 days.

Table 1
Demographic and Clinical Characteristics

	Patients (N=2299)
Age, mean (range), years	34.9 (12-80)
Female, %	59
Male, %	41
Concomitant therapies, %	
Hydroxyurea	57
L-glutamine	8
Transfusions	14
Opioids	75
Iron chelation	9
ESA	4
Follow-up time, mean, days	109

Concomitant therapies were during 1 year pre-index.
ESA, erythropoietin-stimulating agent.

CHANGE IN TREATMENT DAYS

In the 59 (2.6%) patients with a history of iron chelation therapy in the pre-index period, adjusted mean total treatment days were reduced by 45% post-index (51.5 to 28.7 days; *P*<0.001) (Figure 1).

Twenty-nine (49%) of these patients did not have iron chelation therapy post-index.

Figure 1
Change in Iron Chelation Therapy Treatment Days

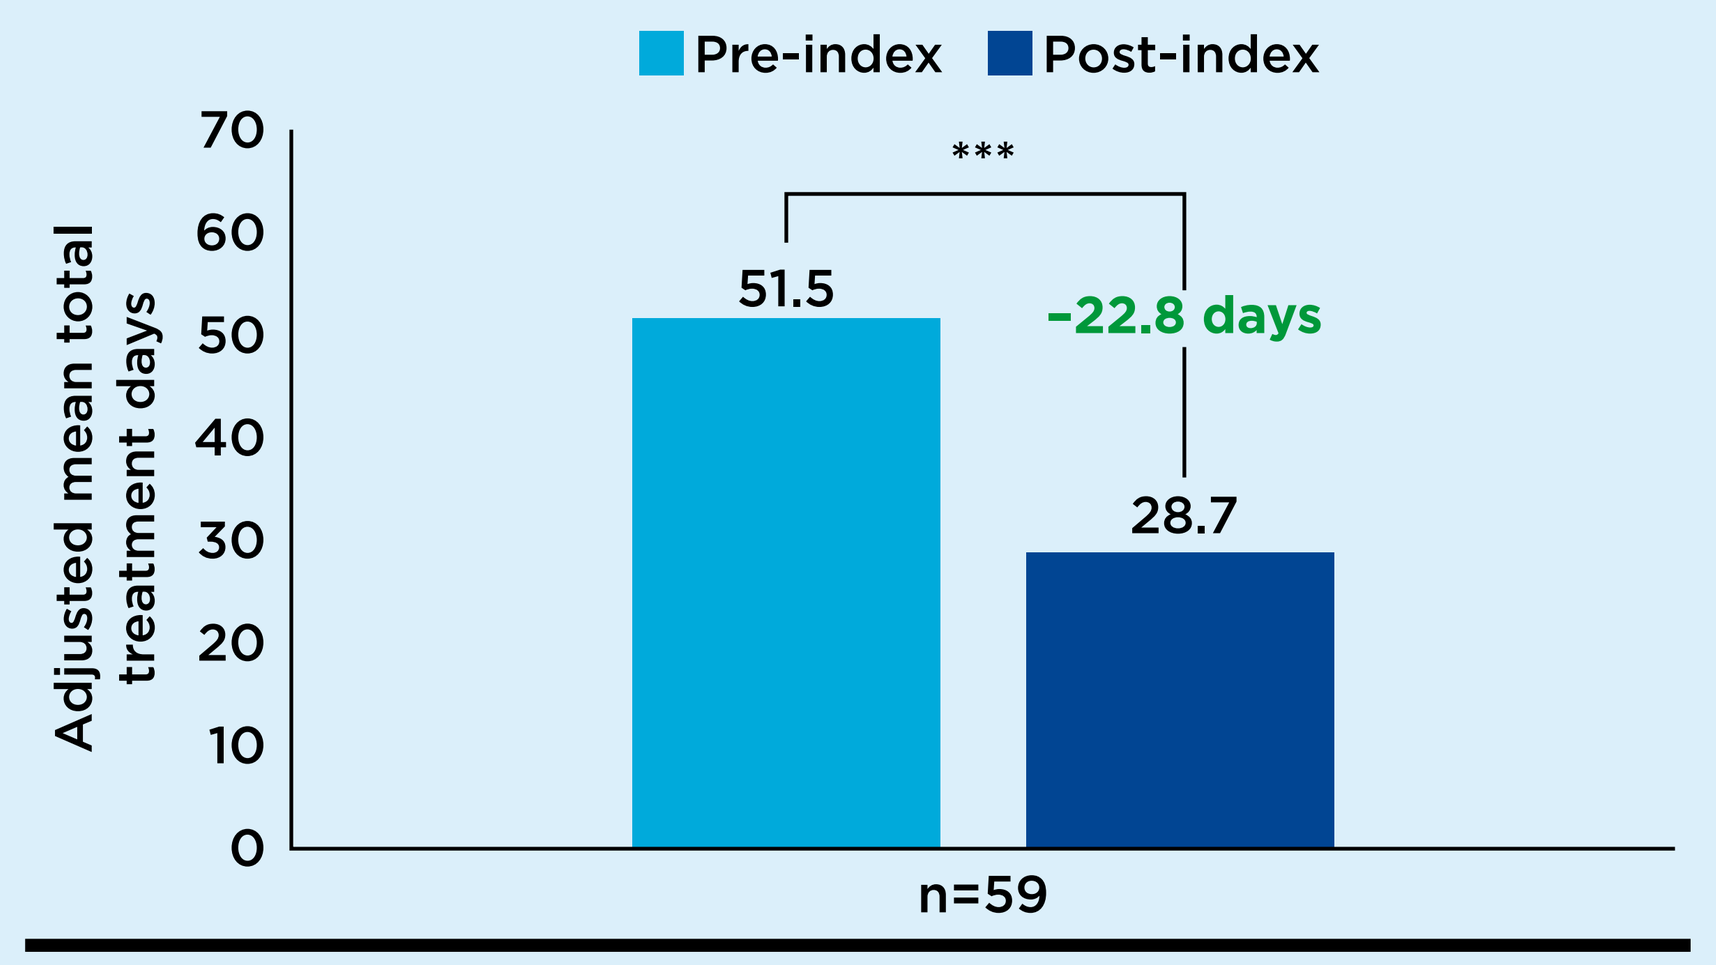
Pre-index
Post-index
Adjusted mean total treatment days
70
60
50
40
30
20
10
0

51.5
−22.8 days
28.7
n=59

***P<0.001.

CHANGE IN DAYS PRESCRIBED OPIOIDS

In the 1386 (60.3%) patients prescribed opioids in the 3 months pre-index, adjusted opioid prescription days decreased by 4.4 days post-index (*P*=0.004), a 6.4% reduction from a mean of 68.5 days in the 3 months pre-index (Figure 2).

Figure 2
Change in Days Prescribed Opioids

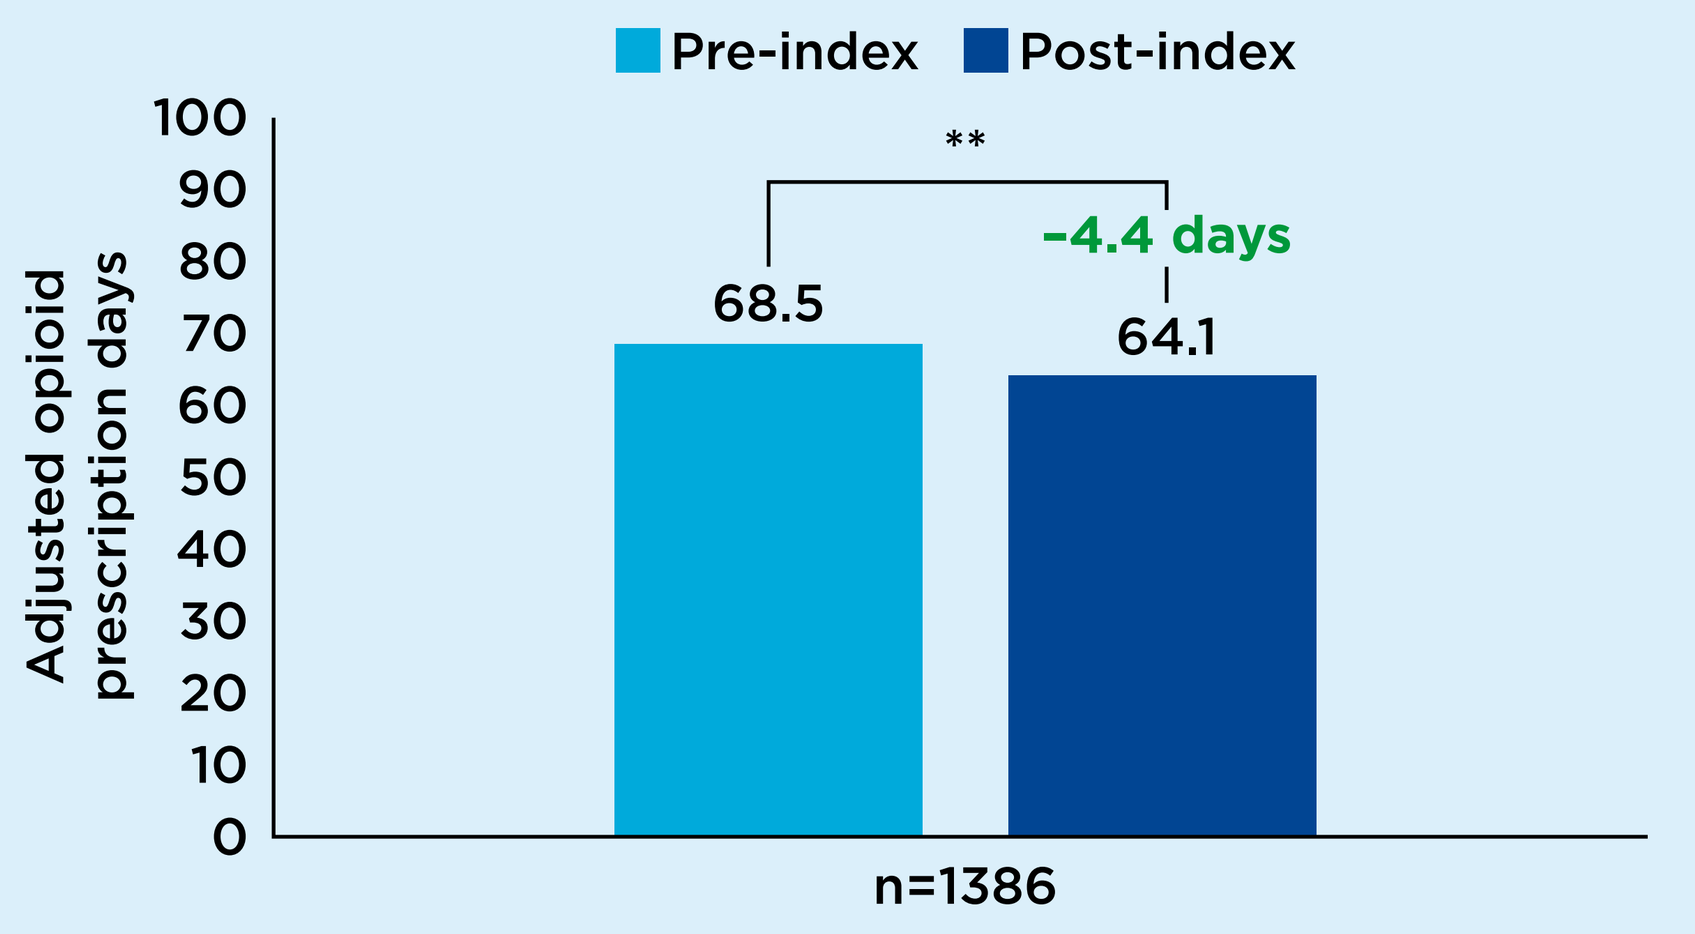
Pre-index
Post-index
Adjusted opioid prescription days
100
90
80
70
60
50
40
30
20
10
0
**
68.5
−4.4 days
64.1
n=1386

**P<0.01.

CHANGE IN ESA

Of the 68 (3%) patients with any ESA use in the 3 months pre-index, 19 (28%) had no ESA use post-index (*P*<0.001) (Figure 3).

Figure 3
Change in ESA Use

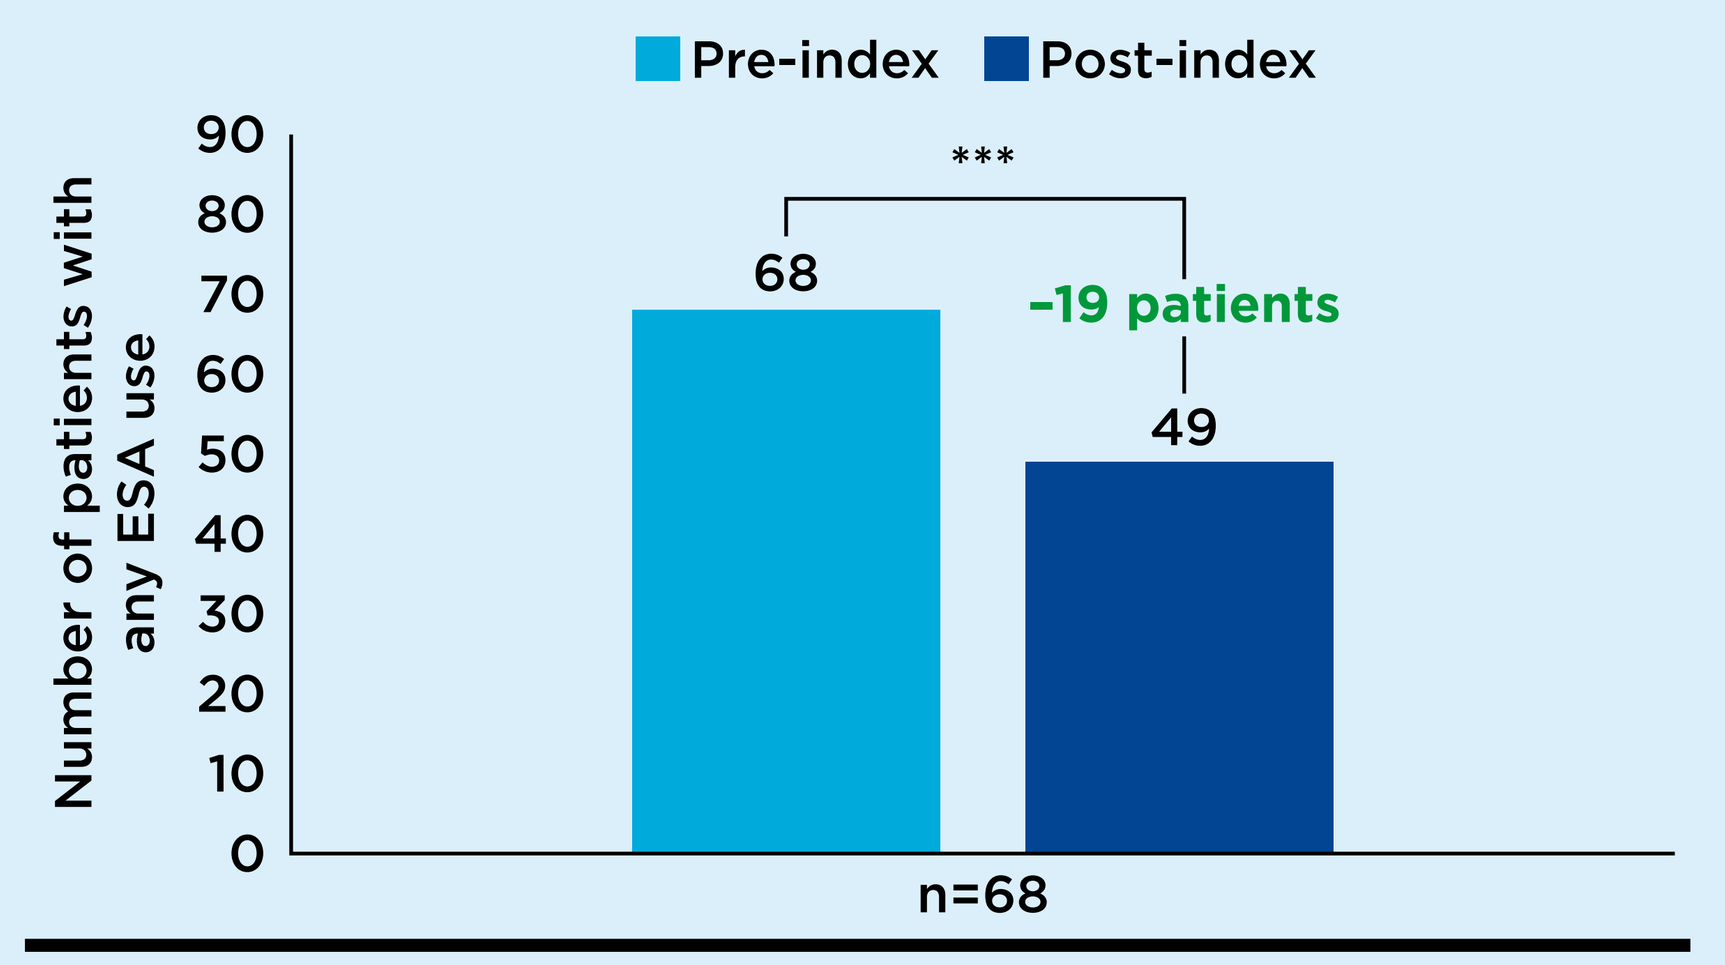
Pre-index
Post-index
Number of patients with any ESA use
90
80
70
60
50
40
30
20
10
0

68
−19 patients
49
n=68

***P<0.001.
ESA, erythropoiesis-stimulating agent.

CONCLUSIONS

Based on the first 13 months of real-world practice data post-approval of voxelotor, the use of iron chelation therapy, opioid prescription days, and ESA use all decreased after voxelotor initiation in a large population of patients receiving the therapy.

Analysis limitations include the possibility of secular trend biases or general regression to the mean.

This study suggests that voxelotor treatment may reduce healthcare resource utilization and the economic burden associated with management of SCD.

REFERENCES

1. Kato GJ, Piel FB, Reid CD, et al. Sickle cell disease. *Nat Rev Dis Primers*. 2018;4:18010.
2. Oxbryta. Prescribing information. Global Blood Therapeutics, Inc; 2019.
3. Vichinsky E, Hoppe CC, Ataga KI, et al. A phase 3 randomized trial of voxelotor in sickle cell disease. *N Engl J Med*. 2019;381:509-519.
4. Zaidi AU, Lipato T, Alvarez OA, et al. Real-world effectiveness of voxelotor for treating sickle cell disease in the US. Presented at: 62nd American Society of Hematology Virtual Annual Meeting and Exposition; December 5-8, 2020. Abstract 2627.

ACKNOWLEDGMENTS

We thank all the patients with sickle cell disease, families, and caregivers who contributed to this study.

DISCLOSURES

Nirmish Shah: Speaker: Global Blood Therapeutics, Novartis, Alexion; research funding: Global Blood Therapeutics, Novartis; consultant: Global Blood Therapeutics, bluebird bio, CSL Behring. **Thokozeni Lipato**: Speaker: Global Blood Therapeutics. **Ofelia Alvarez**: Nothing to disclose. **Thomas E. Delea**: Employee, equity ownership: Policy Analysis Inc.; research funding: Global Blood Therapeutics, Novartis. **Nhat Pham**: Employee: Policy Analysis Inc.; research funding: Global Blood Therapeutics, Novartis. **Alexander Lonshteyn**: Employee: Policy Analysis Inc.; research funding: Global Blood Therapeutics, Novartis. **Derek Weycker**: Employee, equity ownership: Policy Analysis Inc.; research funding: Global Blood Therapeutics, Novartis. **Andy Nguyen**: Employee, equity ownership: Global Blood Therapeutics. **Anne Beaubrun**: Employee, equity ownership: Global Blood Therapeutics.

Dylan Mori, PhD (Healthcare Consultancy Group, with funding from Global Blood Therapeutics) provided editorial assistance in the preparation of this report.

This study was funded by Global Blood Therapeutics.

Presented at: AMCP Nexus 2021; October 18-21, 2021
© 2021 Global Blood Therapeutics