Economic Burden of End-Organ Damage Among Patients With Sickle Cell Disease in the US

3388

Xue Song[1]; Andrew Campbell, MD[2]; Ze Cong, PhD[3]; Irene Agodoa, MD[3]; Diane Martinez, DrPH, MPH[1]; Carolyn R. Lew, PhD[1]; Danae Black, MPH[1]; Helen Varker[1]; Chris Chan, MPH[1]; Sophie Lanzkron, MD, MHS[4]

[1]IBM Watson, Cambridge, MA, USA; [2]Children's National Medical Center, Washington, DC, USA; [3]Global Blood Therapeutics, South San Francisco, CA, USA; [4]Johns Hopkins University, Baltimore, MD, USA

BACKGROUND

- Sickle cell disease (SCD) is an inherited disorder in which pathology is driven by hemoglobin polymerization and red blood cell sickling.[1]
- Chronic anemia and hemolysis in SCD contribute to end-organ damage (EOD) that leads to disability and accelerated mortality (**Figure 1**).[2,3]
- Red blood cell deformation also leads to vaso-occlusion, which manifests as painful crises and cumulative organ damage.[4]
- Owing to these symptoms, the complications associated with SCD, and the resources required to manage them, patients with SCD experience barriers to education and employment.[2]
- While the utilization of medical resources among patients with SCD contributes to high medical costs, the economic burden of EOD among patients with SCD is unknown.

Figure 1. Hemoglobin Polymerization Leads to RBC Sickling and Downstream Clinical Outcomes

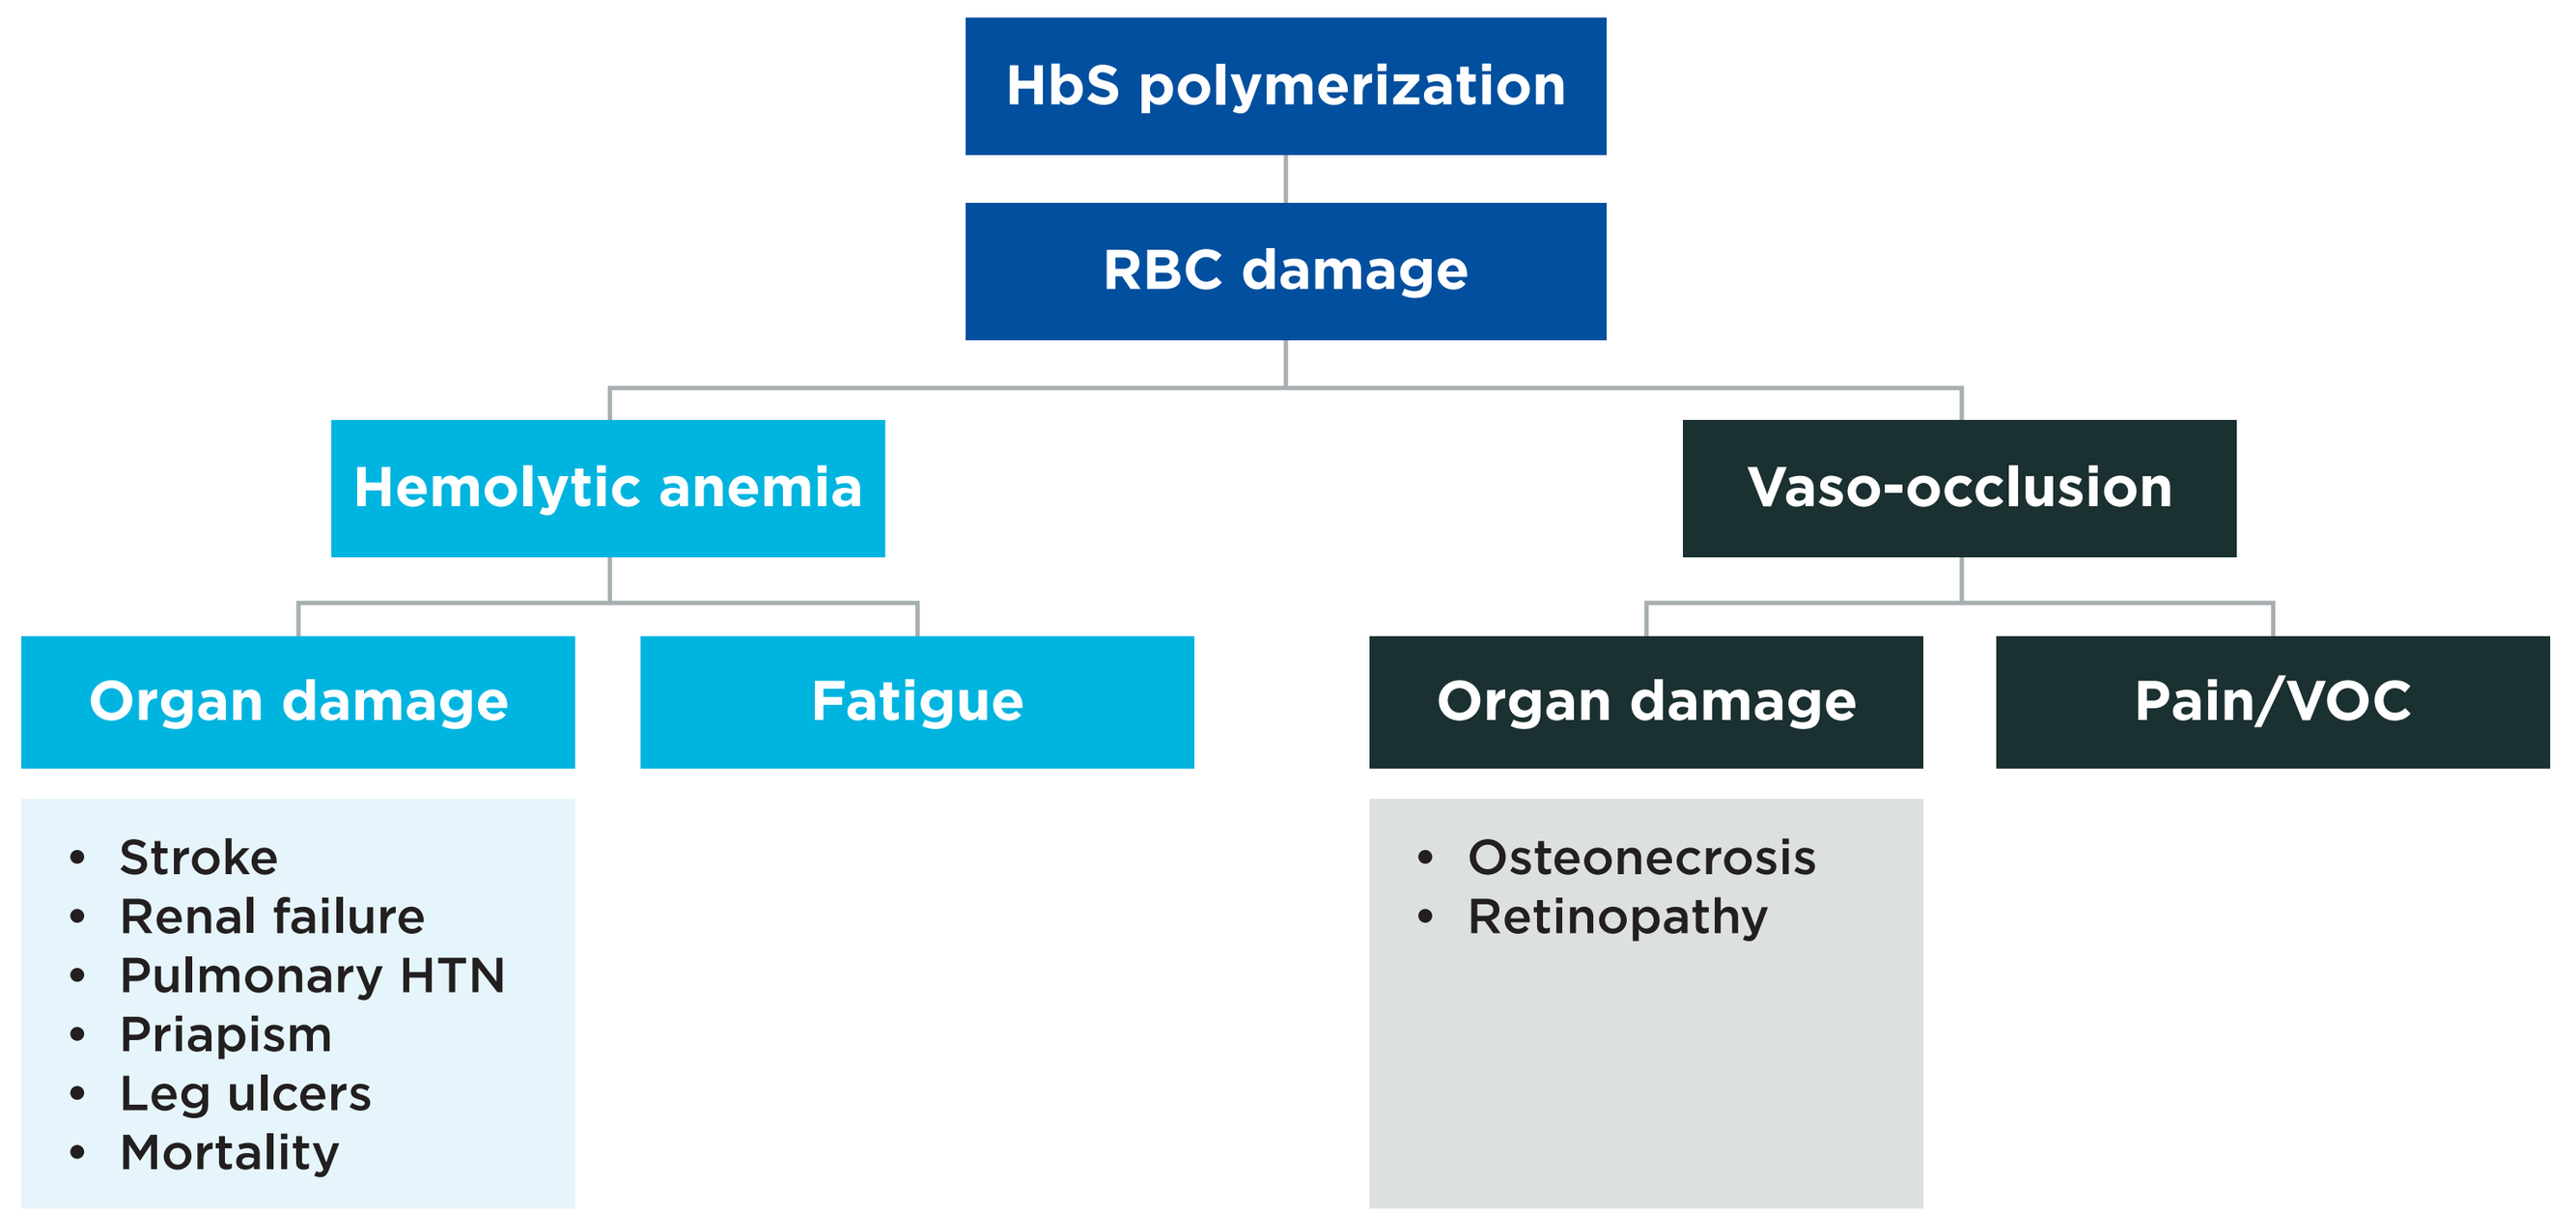
HbS polymerization
RBC damage
Hemolytic anemia
Vaso-occlusion
Organ damage
Fatigue
Organ damage
Pain/VOC
Stroke
Renal failure
Pulmonary HTN
Priapism
Leg ulcers
Mortality
Osteonecrosis
Retinopathy

Figure adapted from Eaton WA, Bunn HF. *Blood.* 2017;129(20):2719-2726.
HbS, sickle hemoglobin; HTN, hypertension; RBC, red blood cell; VOC, vaso-occlusive crisis.

OBJECTIVE

- To estimate the impact of EOD on healthcare resource utilization and costs incurred by patients with SCD on Medicaid.

METHODS

- Patients with SCD (defined as having ≥3 non-diagnostic SCD claims on different days within 5 years) enrolled in Medicaid between January 1, 2013 and December 31, 2017 were identified in the MarketScan® Medicaid claims database.
- The first date of SCD diagnosis was the index date. Patients were required to have at least 3 months of continuous enrollment prior to and 1 month following the index date to be included.
- Each patient's post-index period was divided into a series of 3-month intervals. For each 3-month interval, patients' entire available claims histories (as early as January 1, 2008) were checked to identify 4 types of EOD experienced by patients with SCD: stroke (within 1 year and more than 1 year after a stroke-related event), chronic kidney disease (CKD), end-stage renal disease (ESRD), and pulmonary hypertension (PH).
 - Unless otherwise noted, EOD is measured over the patient's entire history from 2008 until 1 day before the current interval start day.
 - Acute stroke is defined as primary diagnosis of stroke in the inpatient hospital setting.
 - CKD is defined as CKD stages 1-4 and no diagnosis of ESRD, no dialysis, and no kidney transplant.
 - ESRD is defined as a diagnosis of ESRD, CKD stage 5, or CKD of any stage with dialysis or kidney transplant.
- Patient characteristics, healthcare resource utilization, and costs were summarized descriptively by type of EOD.
- Three multivariate regression models were employed to estimate the relative cost ratios of patients with versus without EOD, controlling for patients' demographic and clinical characteristics.
- Annualized costs for adult patients with each type of EOD were estimated based on the regression results.

RESULTS

Patient Demographics

- In total, 10,784 patients with SCD on Medicaid were identified.
- Patients were followed for 3.35 years on average, contributing 152,455 intervals (mean age of 18.5 years; female: 54.6%; urban: 84.4%; **Table 1**).
- Other SCD complications included avascular necrosis (3.7%), followed by splenomegaly, splenic sequestration, and hypersplenism (2.3%) and gallstones, cholelithiasis, and cholecystitis (1.3%).

Table 1. Patient Demographics

	All Intervals (n=152,455)
Age, mean (SD), years	18.5 (13.6)
Male, n (%)	69,286 (45.4)
Urban population density, n (%)	128,714 (84.4)
Race, n (%)	
Black	111,839 (73.4)
Unknown	35,890 (23.5)
White	2506 (1.6)
Hispanic	1459 (1.0)
Other	761 (0.5)

SD, standard deviation.

Healthcare Resource Utilization and Costs Among Patients With vs Without EOD

- Patients with EOD had more days in the hospital, physician visits, emergency department visits, outpatient visits, lab tests, and outpatient pharmacy claims per month than patients without EOD (**Figure 2**).

Figure 2. Mean Number of Healthcare Resource Utilizations, PPPM

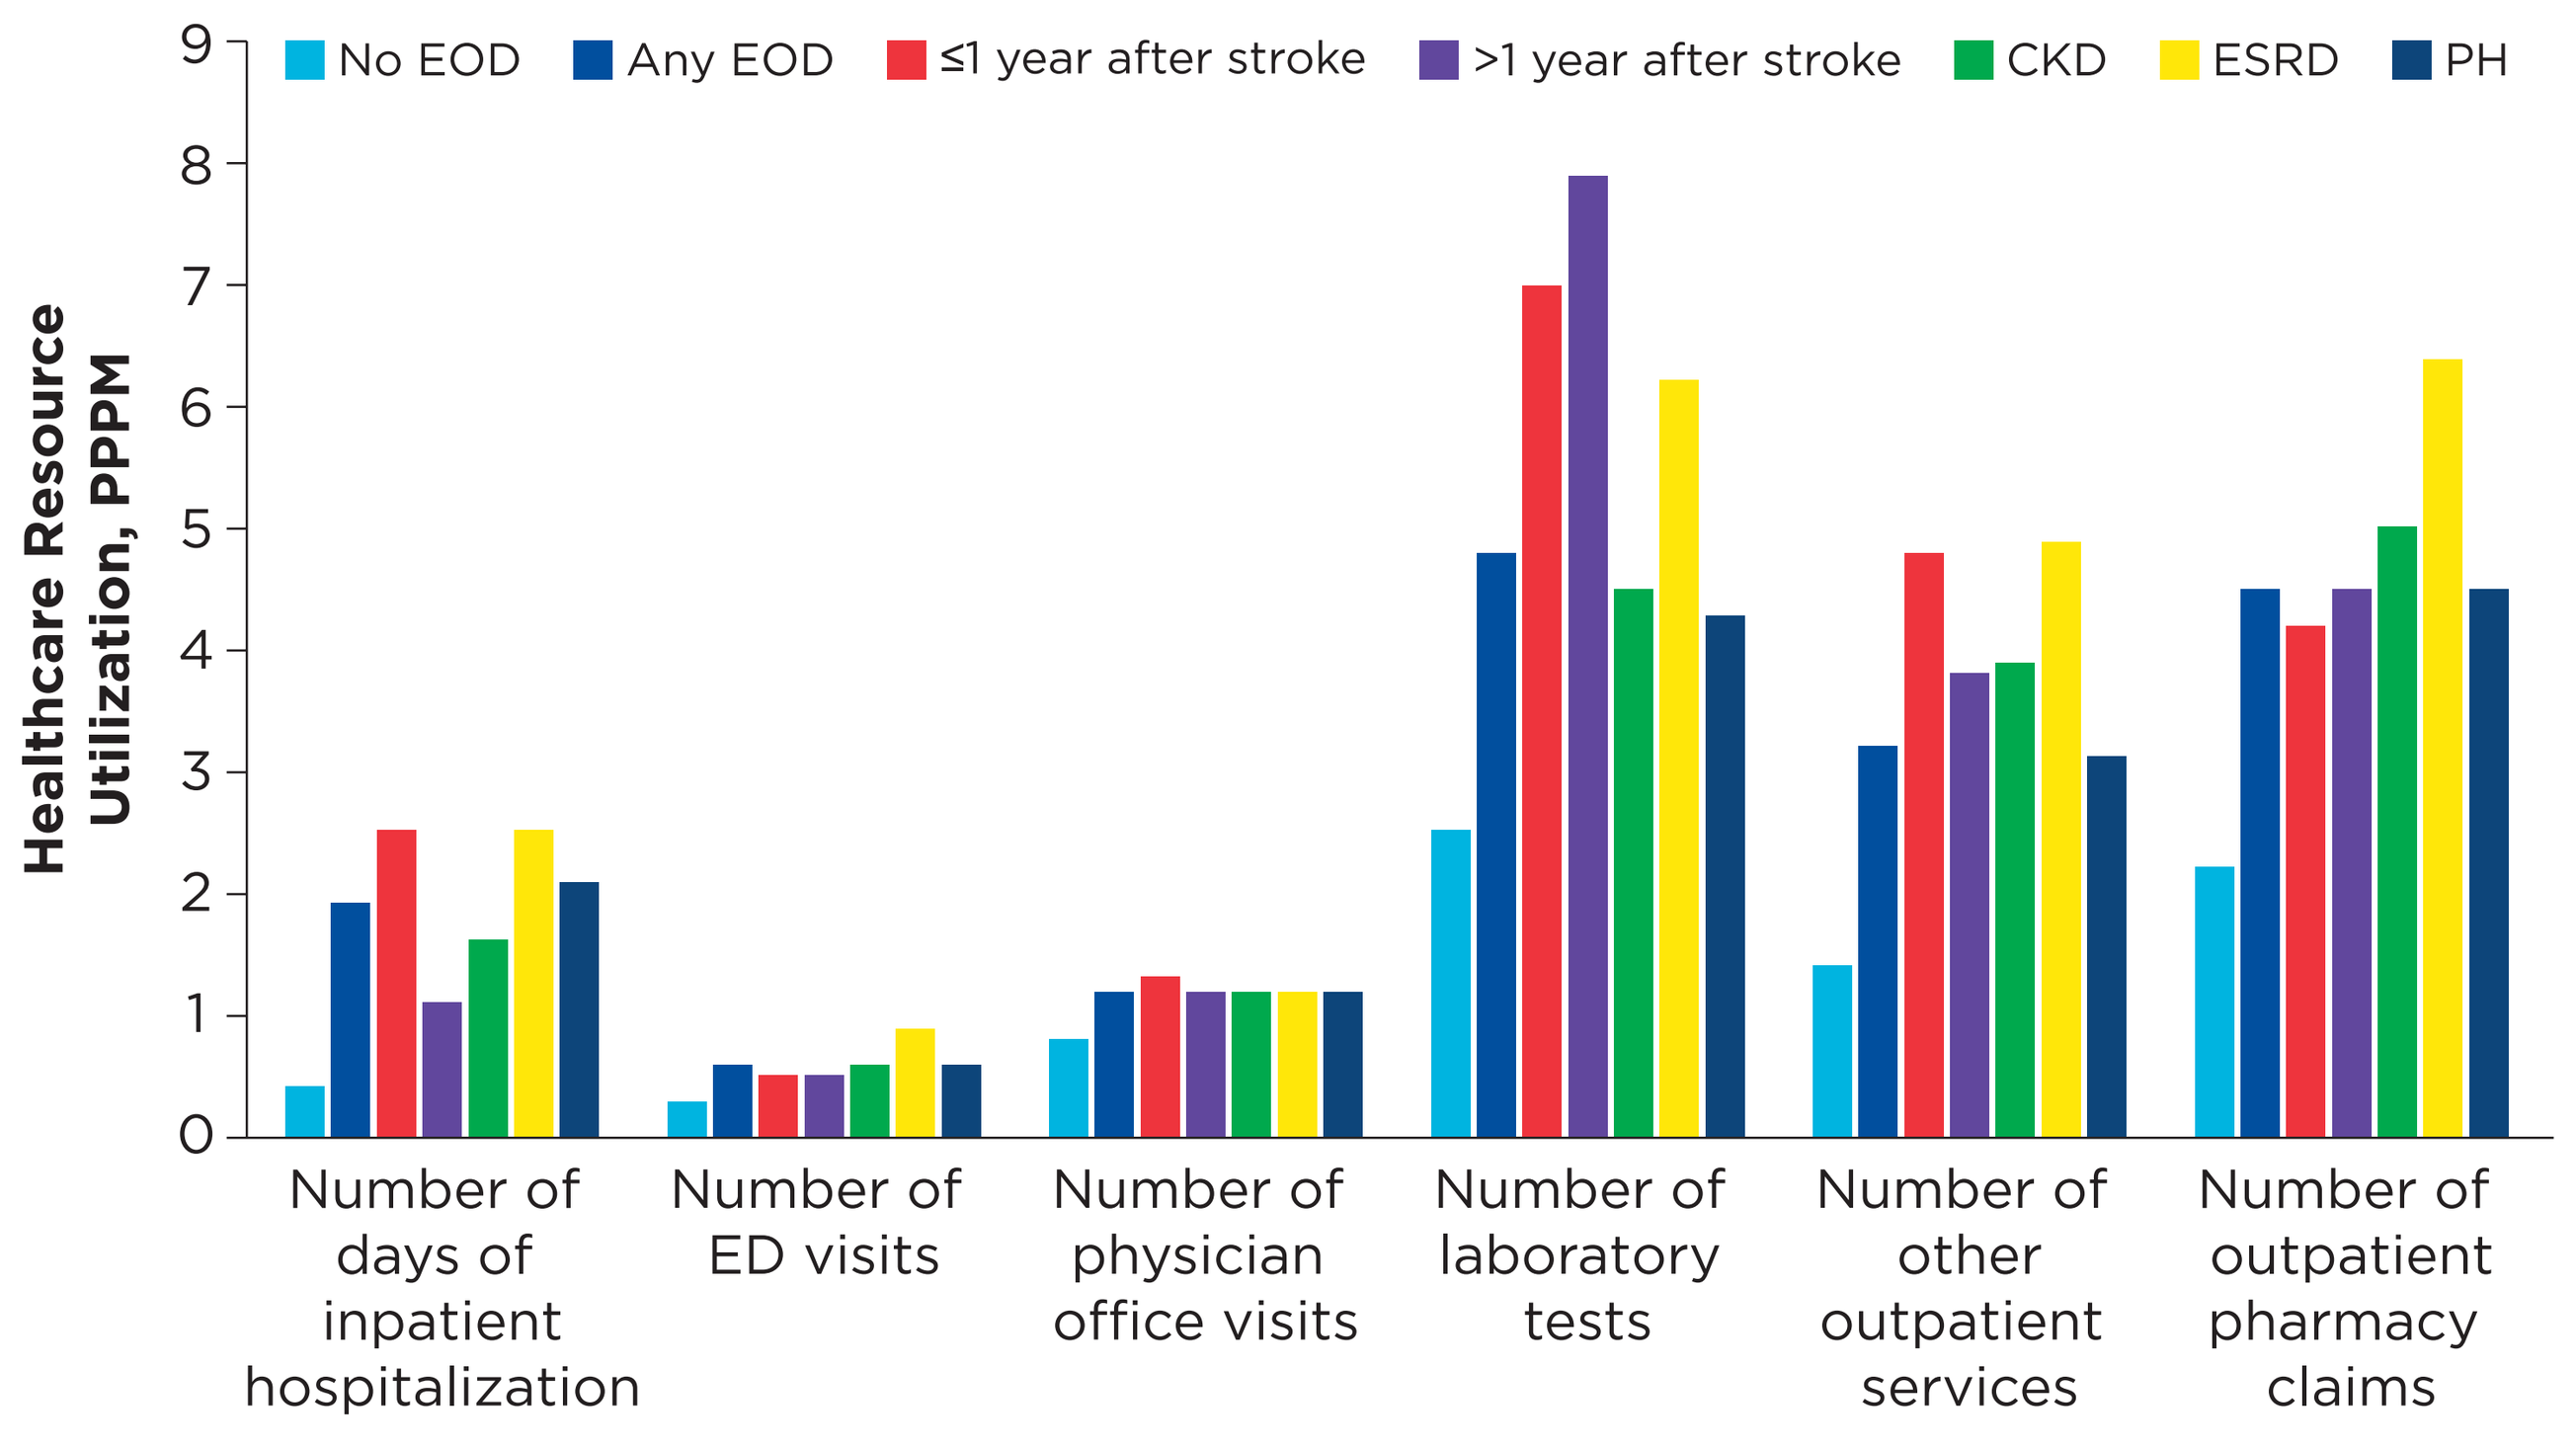
No EOD
Any EOD
≤1 year after stroke
>1 year after stroke
CKD
ESRD
PH
Healthcare Resource Utilization, PPPM
0
1
2
3
4
5
6
7
8
9
Number of days of inpatient hospitalization
Number of ED visits
Number of physician office visits
Number of laboratory tests
Number of other outpatient services
Number of outpatient pharmacy claims

CKD, chronic kidney disease; ED, emergency department; EOD, end-organ damage; ESRD, end-stage renal disease; PH, pulmonary hypertension; PPPM, per patient per month.

Multivariate Regression Results Among Patients With SCD and Total Healthcare Costs

- The mean (SD) cost per hospitalization for acute stroke was $55,314 ($76,847).
- In multivariate regression model 1 (accounting for EOD only), patients with any EOD had significantly higher costs than those without these conditions (cost ratios: 2.10-4.66).
- After controlling for patient demographic characteristics (model 2) and additional clinical characteristics (model 3), the results were similar.
- The costs of patients with SCD in the first year after stroke are 4.68 times as high as the costs of patients without any organ damage (2.08 times if more than 1 year after stroke; 2.32 times for PH; 2.19 times for CKD; and 3.40 times for ESRD; **Table 2**).

Table 2. Multivariate Regression Results for Total Healthcare Costs (Cost Ratios)

	Model 1 Cost Ratio	95% CI	Model 2 Cost Ratio	95% CI	Model 3 Cost Ratio	95% CI
EOD						
No EOD	1.00	-	1.00	-	1.00	-
≤1 year after acute stroke	**4.66**[a]	(3.74-5.81)	**4.63**[a]	(3.58-5.99)	**4.68**[a]	(3.70-5.92)
>1 year after acute stroke	**2.10**[a]	(1.75-2.52)	**2.05**[a]	(1.73-2.42)	**2.08**[a]	(1.77-2.44)
CKD	**2.75**[a]	(2.24-3.39)	**2.21**[a]	(1.84-2.65)	**2.19**[a]	(1.82-2.64)
ESRD	**4.34**[a]	(3.21-5.86)	**3.38**[a]	(2.59-4.41)	**3.40**[a]	(2.58-4.48)
PH	**3.30**[a]	(2.94-3.70)	**2.49**[a]	(2.22-2.79)	**2.32**[a]	(2.08-2.60)
Age groups, years						
<6			0.36[a]	(0.33-0.40)	0.40[a]	(0.35-0.44)
6-11			0.38[a]	(0.34-0.41)	0.40[a]	(0.36-0.44)
12-17			0.53[a]	(0.48-0.59)	0.55[a]	(0.50-0.60)
18-30			1.00		1.00	
31-44			0.88	(0.78-1.00)	0.87[c]	(0.77-0.99)
45-54			0.58[a]	(0.49-0.68)	0.60[a]	(0.51-0.70)
55-64			0.45[a]	(0.36-0.57)	0.46[a]	(0.36-0.58)
65+			0.30[a]	(0.20-0.46)	0.32[a]	(0.21-0.49)
Gender						
Female (vs male)			1.04	(0.96-1.11)	1.06	(0.99-1.14)
Population density						
Rural (vs urban)			1.00	(0.91-1.10)	1.00	(0.92-1.10)
Other SCD complications						
Avascular necrosis					2.10[a]	(1.89-2.32)
Gallstones, cholelithiasis, cholecystitis					2.06[a]	(1.81-2.35)
Priapism					1.65[a]	(1.38-1.97)
Leg ulcers					1.60[b]	(1.21-2.10)
Osteomyelitis					1.84[a]	(1.47-2.30)

The transitional age group (**18-30 years**) had significantly **higher costs** than other age groups.

Having **other SCD complications** also significantly **increased the total costs**.

[a]P<0.0001; [b]P<0.01; [c]P<0.05.
CI, confidence interval; CKD, chronic kidney disease; EOD, end-organ damage; ESRD, end-stage renal disease; PH, pulmonary hypertension; SCD, sickle cell disease.

- Based on model 3, after controlling for patient demographics and clinical characteristics, the predicted mean annual costs for adult patients with SCD in the first year after a stroke are $285,816; $127,393 if more than 1 year after a stroke; and $135,493, $209,172, or $148,174 if the patient had CKD, ESRD, or PH, respectively (**Figure 3**).
- Patients with multiple SCD complications had even higher costs, with the predicted mean annual cost for adult patients with CKD and avascular necrosis being $270,513.

Figure 3. Estimated Annual Cost per Patient per Year for Adult Patients With SCD on Medicaid

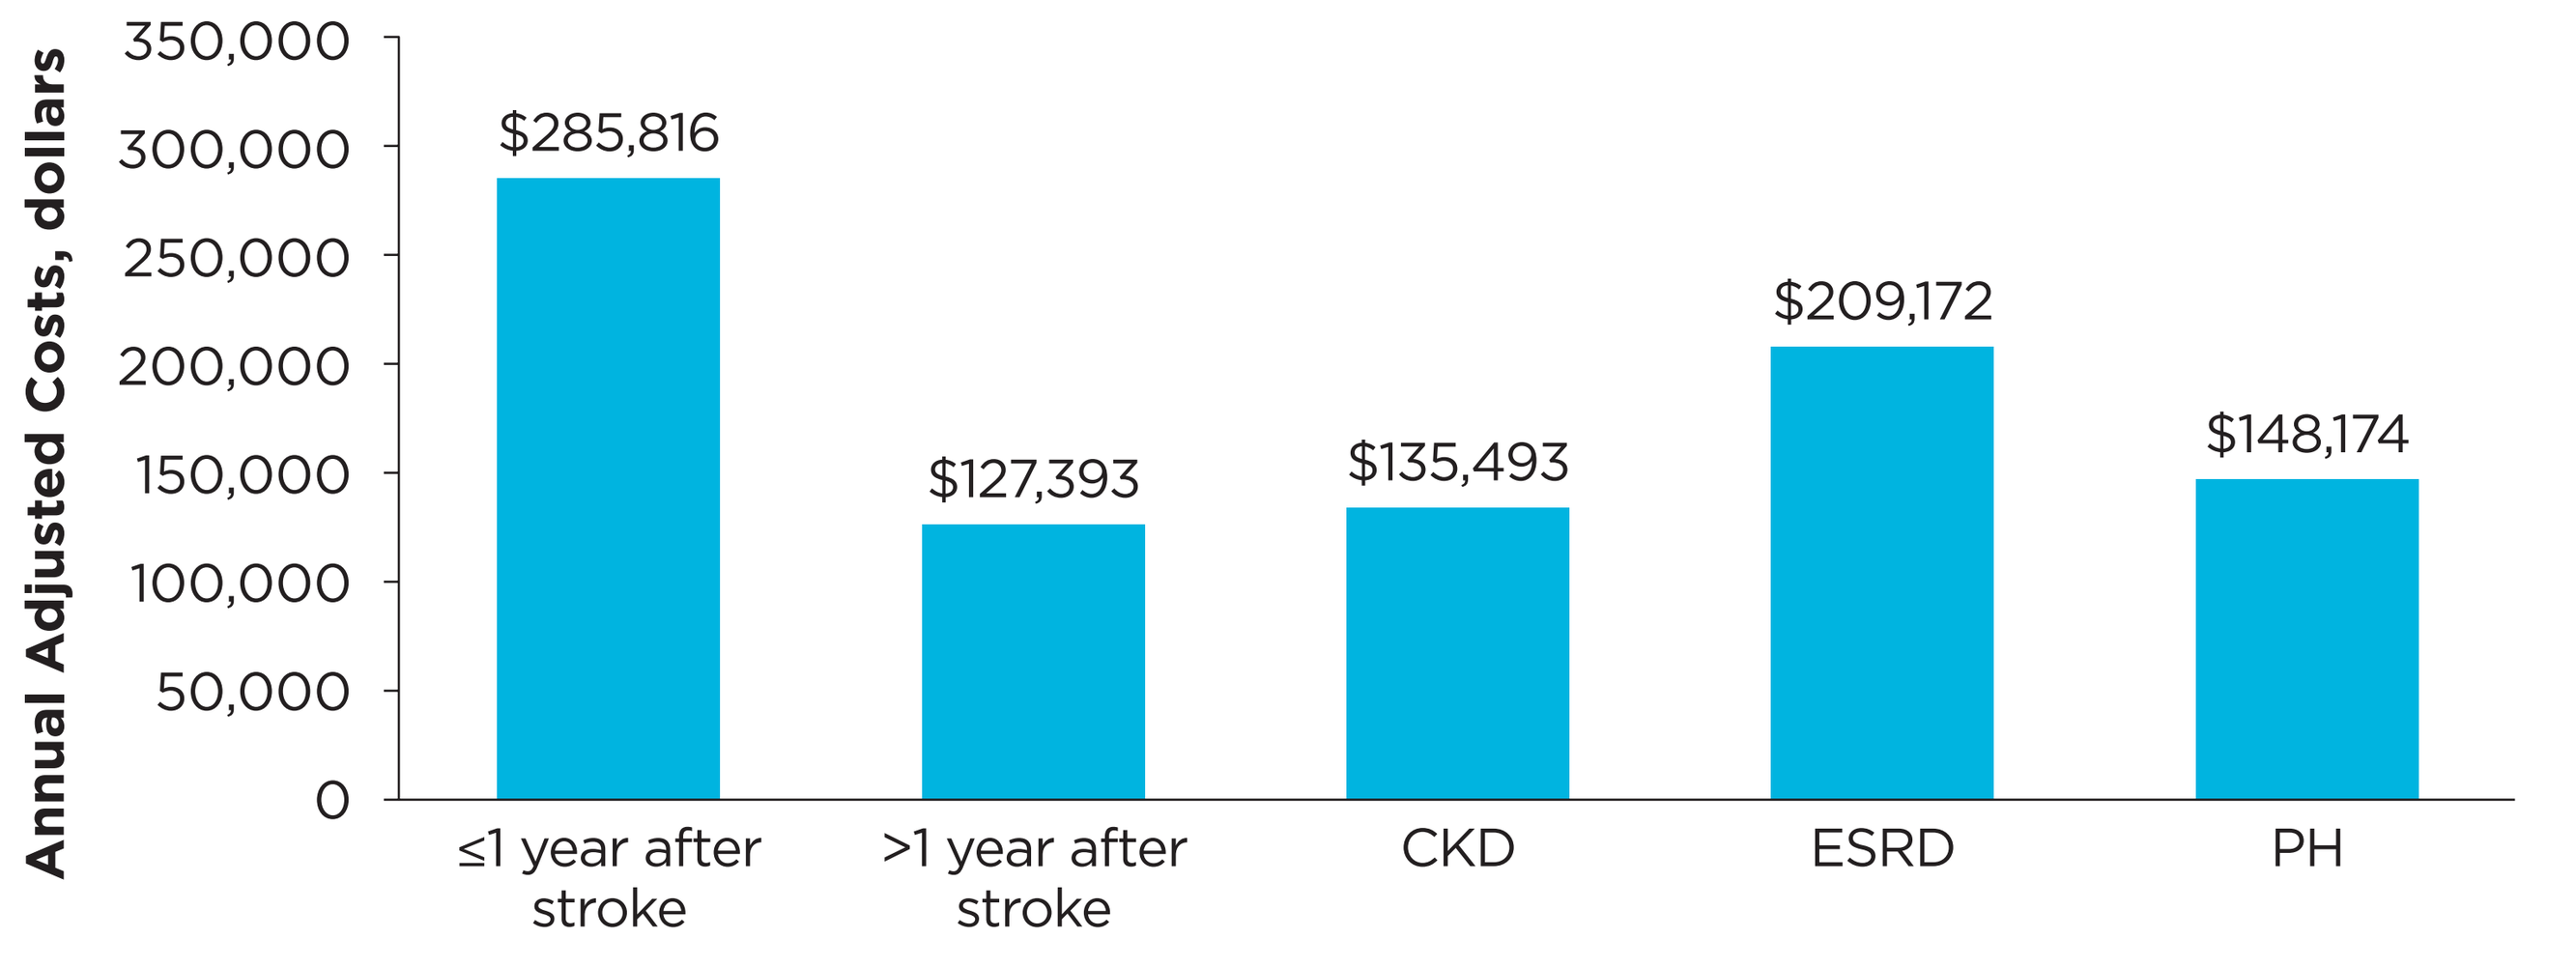
Annual Adjusted Costs, dollars
0
50,000
100,000
150,000
200,000
250,000
300,000
350,000
$285,816
$127,393
$135,493
$209,172
$148,174
≤1 year after stroke
>1 year after stroke
CKD
ESRD
PH

CKD, chronic kidney disease; ESRD, end-stage renal disease; PH, pulmonary hypertension; SCD, sickle cell disease.

LIMITATIONS

- Results of this analysis may not be generalizable to patients with other insurance (eg, commercial, Medicare) or patients without health insurance coverage.
- The potential for misclassification of SCD, covariates, or study outcomes was present, as patients were identified through administrative claims data as opposed to medical records. As with any claims database, the MarketScan Research Database relies on administrative claims data for clinical detail. These data are subject to data coding limitations and data entry error.
- Diagnoses of EOD prior to January 1, 2008 were not available.
- While statistical analysis was conducted to adjust for patients' characteristics, systemic differences may still exist between patients with and without EOD.

CONCLUSIONS

- SCD is associated with substantial economic burden. When patients experience EOD such as stroke, renal dysfunction, or cardiopulmonary conditions, this economic burden is significantly elevated.
- SCD management strategies that can potentially reduce the risks of EOD offer both clinical and economic value to patients and society.
- Medicaid policy decisions regarding SCD management should consider the chronic and progressive nature of the disease and its associated economic burden and improve access to disease-modifying interventions.

References

1. Rees DC, Williams TN, Gladwin MT. Sickle-cell disease. *Lancet*. 2010;376(9757):2018-2031.
2. Swanson ME, Grosse SD, Kulkarni R. Disability among individuals with sickle cell disease: literature review from a public health perspective. *Am J Prev Med*. 2011;41(6 Suppl 4):S390-S397.
3. Kato GJ, Piel FB, Reid CD, et al. Sickle cell disease. *Nat Rev Dis Primers*. 2018;4:18010.
4. Piel FB, Steinberg MH, Rees DC. Sickle cell disease. *N Engl J Med*. 2017;376(16):1561-1573.

Acknowledgments

- We thank all the patients with sickle cell disease, families, caregivers, research nurses, study coordinators, and support staff who contributed to this study.

Disclosures

- **Andrew Campbell, Sophie Lanzkron:** Global Blood Therapeutics, consultants. **Xue Song, Diane Martinez, Carolyn R. Lew, Danae Black, Helen Varker, Chris Chan:** Global Blood Therapeutics, research support. **Irene Agodoa, Ze Cong:** Global Blood Therapeutics, employees and stockholders.
- Nelson Jen, PhD (Healthcare Consultancy Group, with funding from Global Blood Therapeutics) provided editorial assistance in the preparation of this report.
- This study was sponsored by Global Blood Therapeutics.

For a copy of this poster, scan the QR code with your Android™ phone, Blackberry®, or iPhone®. No personal information will be collected. This is not associated with any marketing or promotional activity.

https://bit.ly/2JRPcBW

Presented at: 61st ASH Annual Meeting and Exposition; December 7-10, 2019; Orlando, FL, USA

© 2019 Global Blood Therapeutics