Correlation of Voxelotor Exposure With Hemoglobin Response and Measures of Hemolysis in Patients From the HOPE Study

1020

Jo Howard, MD[1,2]; Elliott Vichinsky, MD[3]; Jennifer Knight-Madden, MBBS, PhD[4]; Margaret Tonda, PharmD[5]; Carla Washington, PhD[5]; Barbara Tong, PhD[5]; Joshua Lehrer-Graiwer, MD, MPhil, FACC[5]; Victor R. Gordeuk, MD[6]

[1]Guy's and St Thomas' NHS Foundation Trust, London, UK; [2]King's College, London, UK; [3]UCSF Benioff Children's Hospital Oakland, Oakland, CA, USA; [4]University of the West Indies, Mona, Jamaica; [5]Global Blood Therapeutics, South San Francisco, CA, USA; [6]University of Illinois at Chicago College of Medicine, Chicago, IL, USA

BACKGROUND

- Sickle cell disease (SCD) is an inherited disorder in which pathology is driven by hemoglobin (Hb) polymerization and red blood cell sickling, leading to chronic hemolysis and anemia as well as episodic vaso-occlusion.[1]
 - Hemolytic anemia has been consistently associated with vasculopathic complications, including stroke and pulmonary hypertension.[2]
 - These manifestations of SCD contribute to cumulative organ damage, which leads to disability and accelerated mortality.[1,3]
- Voxelotor is a first-in-class sickle Hb polymerization inhibitor being developed for the treatment of SCD. In the phase 3 HOPE trial, voxelotor 1500 mg and 900 mg administered orally once daily demonstrated rapid and sustained dose-dependent improvements in Hb levels and reductions in measures of hemolysis compared with placebo.[4]

OBJECTIVE

- To evaluate the association between Hb response and measures of hemolysis in voxelotor-treated patients in the phase 3 HOPE trial in a post hoc analysis.

METHODS

Study Design

- A post hoc analysis of the data from the HOPE trial was performed. The HOPE trial was a phase 3, randomized, placebo-controlled, double-blind, multicenter study comparing the efficacy and safety of 2 different doses of voxelotor with placebo in patients with SCD who were aged 12 to 65 years (**Figure 1**).
 - The primary endpoint was the percentage of patients with a Hb response at week 24, defined as a >1.0 g/dL increase in Hb from baseline.
 - Secondary endpoints included change in markers associated with hemolysis: absolute reticulocyte count, percentage of reticulocytes, indirect bilirubin level, and lactate dehydrogenase (LDH) level.

Figure 1. HOPE Study Design

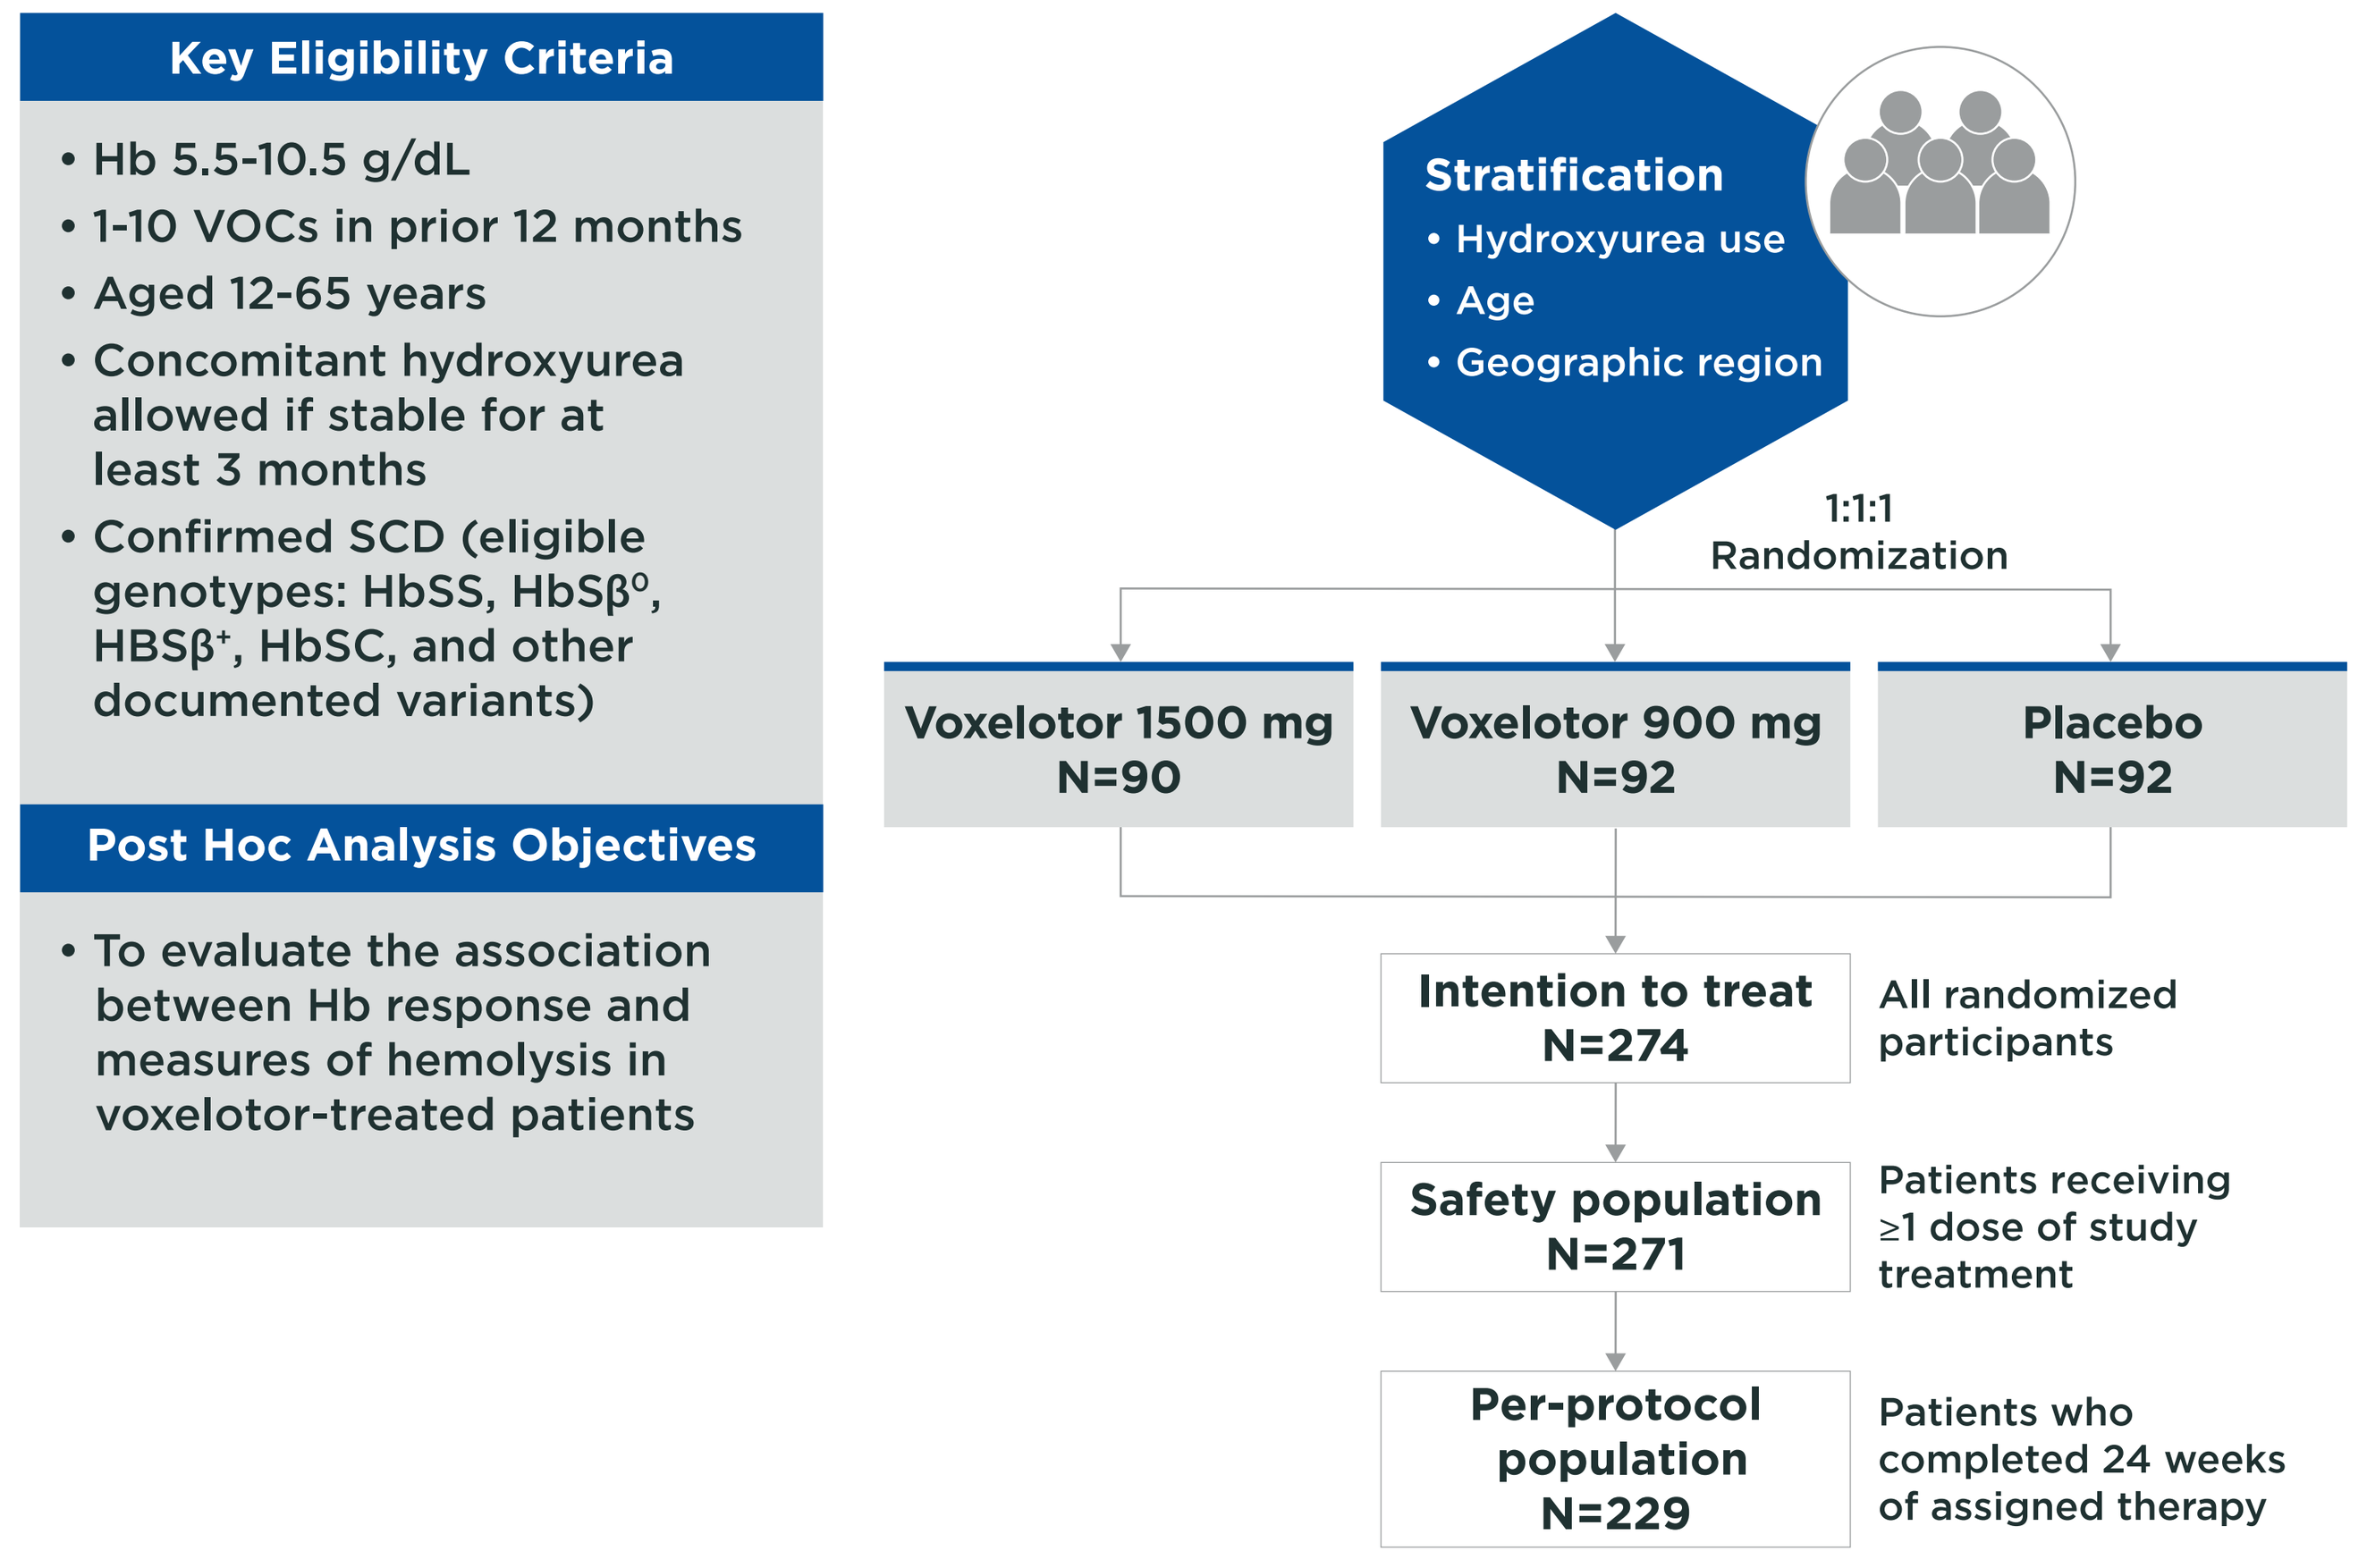
Key Eligibility Criteria
• Hb 5.5-10.5 g/dL
• 1-10 VOCs in prior 12 months
• Aged 12-65 years
• Concomitant hydroxyurea allowed if stable for at least 3 months
• Confirmed SCD (eligible genotypes: HbSS, HbSβ⁰, HBSβ⁺, HbSC, and other documented variants)
Post Hoc Analysis Objectives
• To evaluate the association between Hb response and measures of hemolysis in voxelotor-treated patients
Stratification
• Hydroxyurea use
• Age
• Geographic region
1:1:1 Randomization
Voxelotor 1500 mg N=90
Voxelotor 900 mg N=92
Placebo N=92
Intention to treat N=274
All randomized participants
Safety population N=271
Patients receiving ≥1 dose of study treatment
Per-protocol population N=229
Patients who completed 24 weeks of assigned therapy

Hb, hemoglobin; HbSC, hemoglobin SC disease; HbSS, sickle cell anemia; HbSβ, sickle beta thalassemia; SCD, sickle cell disease; VOC, vaso-occlusive crisis.

- The per-protocol population was defined as participants who completed the week 24 visit of their assigned treatment regimen and who did not initiate hydroxyurea treatment between baseline and week 24.
 - Patients receiving voxelotor 1500 mg and 900 mg were assessed collectively in a voxelotor pooled group.
 - Values of hemolytic markers at baseline were averaged from values at screening and on the day of randomization, and values at 24 weeks were averaged from those at weeks 20 and 24.
- Lab results after initiation of hydroxyurea (for subjects with no baseline hydroxyurea use) or after withdrawal of consent were excluded from the analysis.
- Pharmacokinetic/pharmacodynamic modeling was used to correlate voxelotor exposures with Hb responses and measures of hemolysis.

RESULTS

Baseline Characteristics

- The baseline characteristics were similar across treatment and placebo groups, as previously reported.[4]

Measures of Hemolysis Were Lower in Voxelotor-Treated Patients

- Among patients receiving voxelotor, all measures of hemolysis were consistently lower for those with increases in Hb of >1 g/dL than for those with changes of ≤1 g/dL (**Figure 2**).
 - Hb improvements with voxelotor treatment correlated with lower levels of indirect bilirubin and LDH, and lower absolute reticulocyte count and percentage of reticulocytes, compared with placebo.
- Across all parameters, the degree of reduction in measures of hemolysis was greater in the voxelotor 1500 mg treatment group compared with the 900 mg group (data not shown).[4]

Figure 2. Hb Improvements With Voxelotor Correlated With Decreasing Measures of Hemolysis

A. Indirect Bilirubin

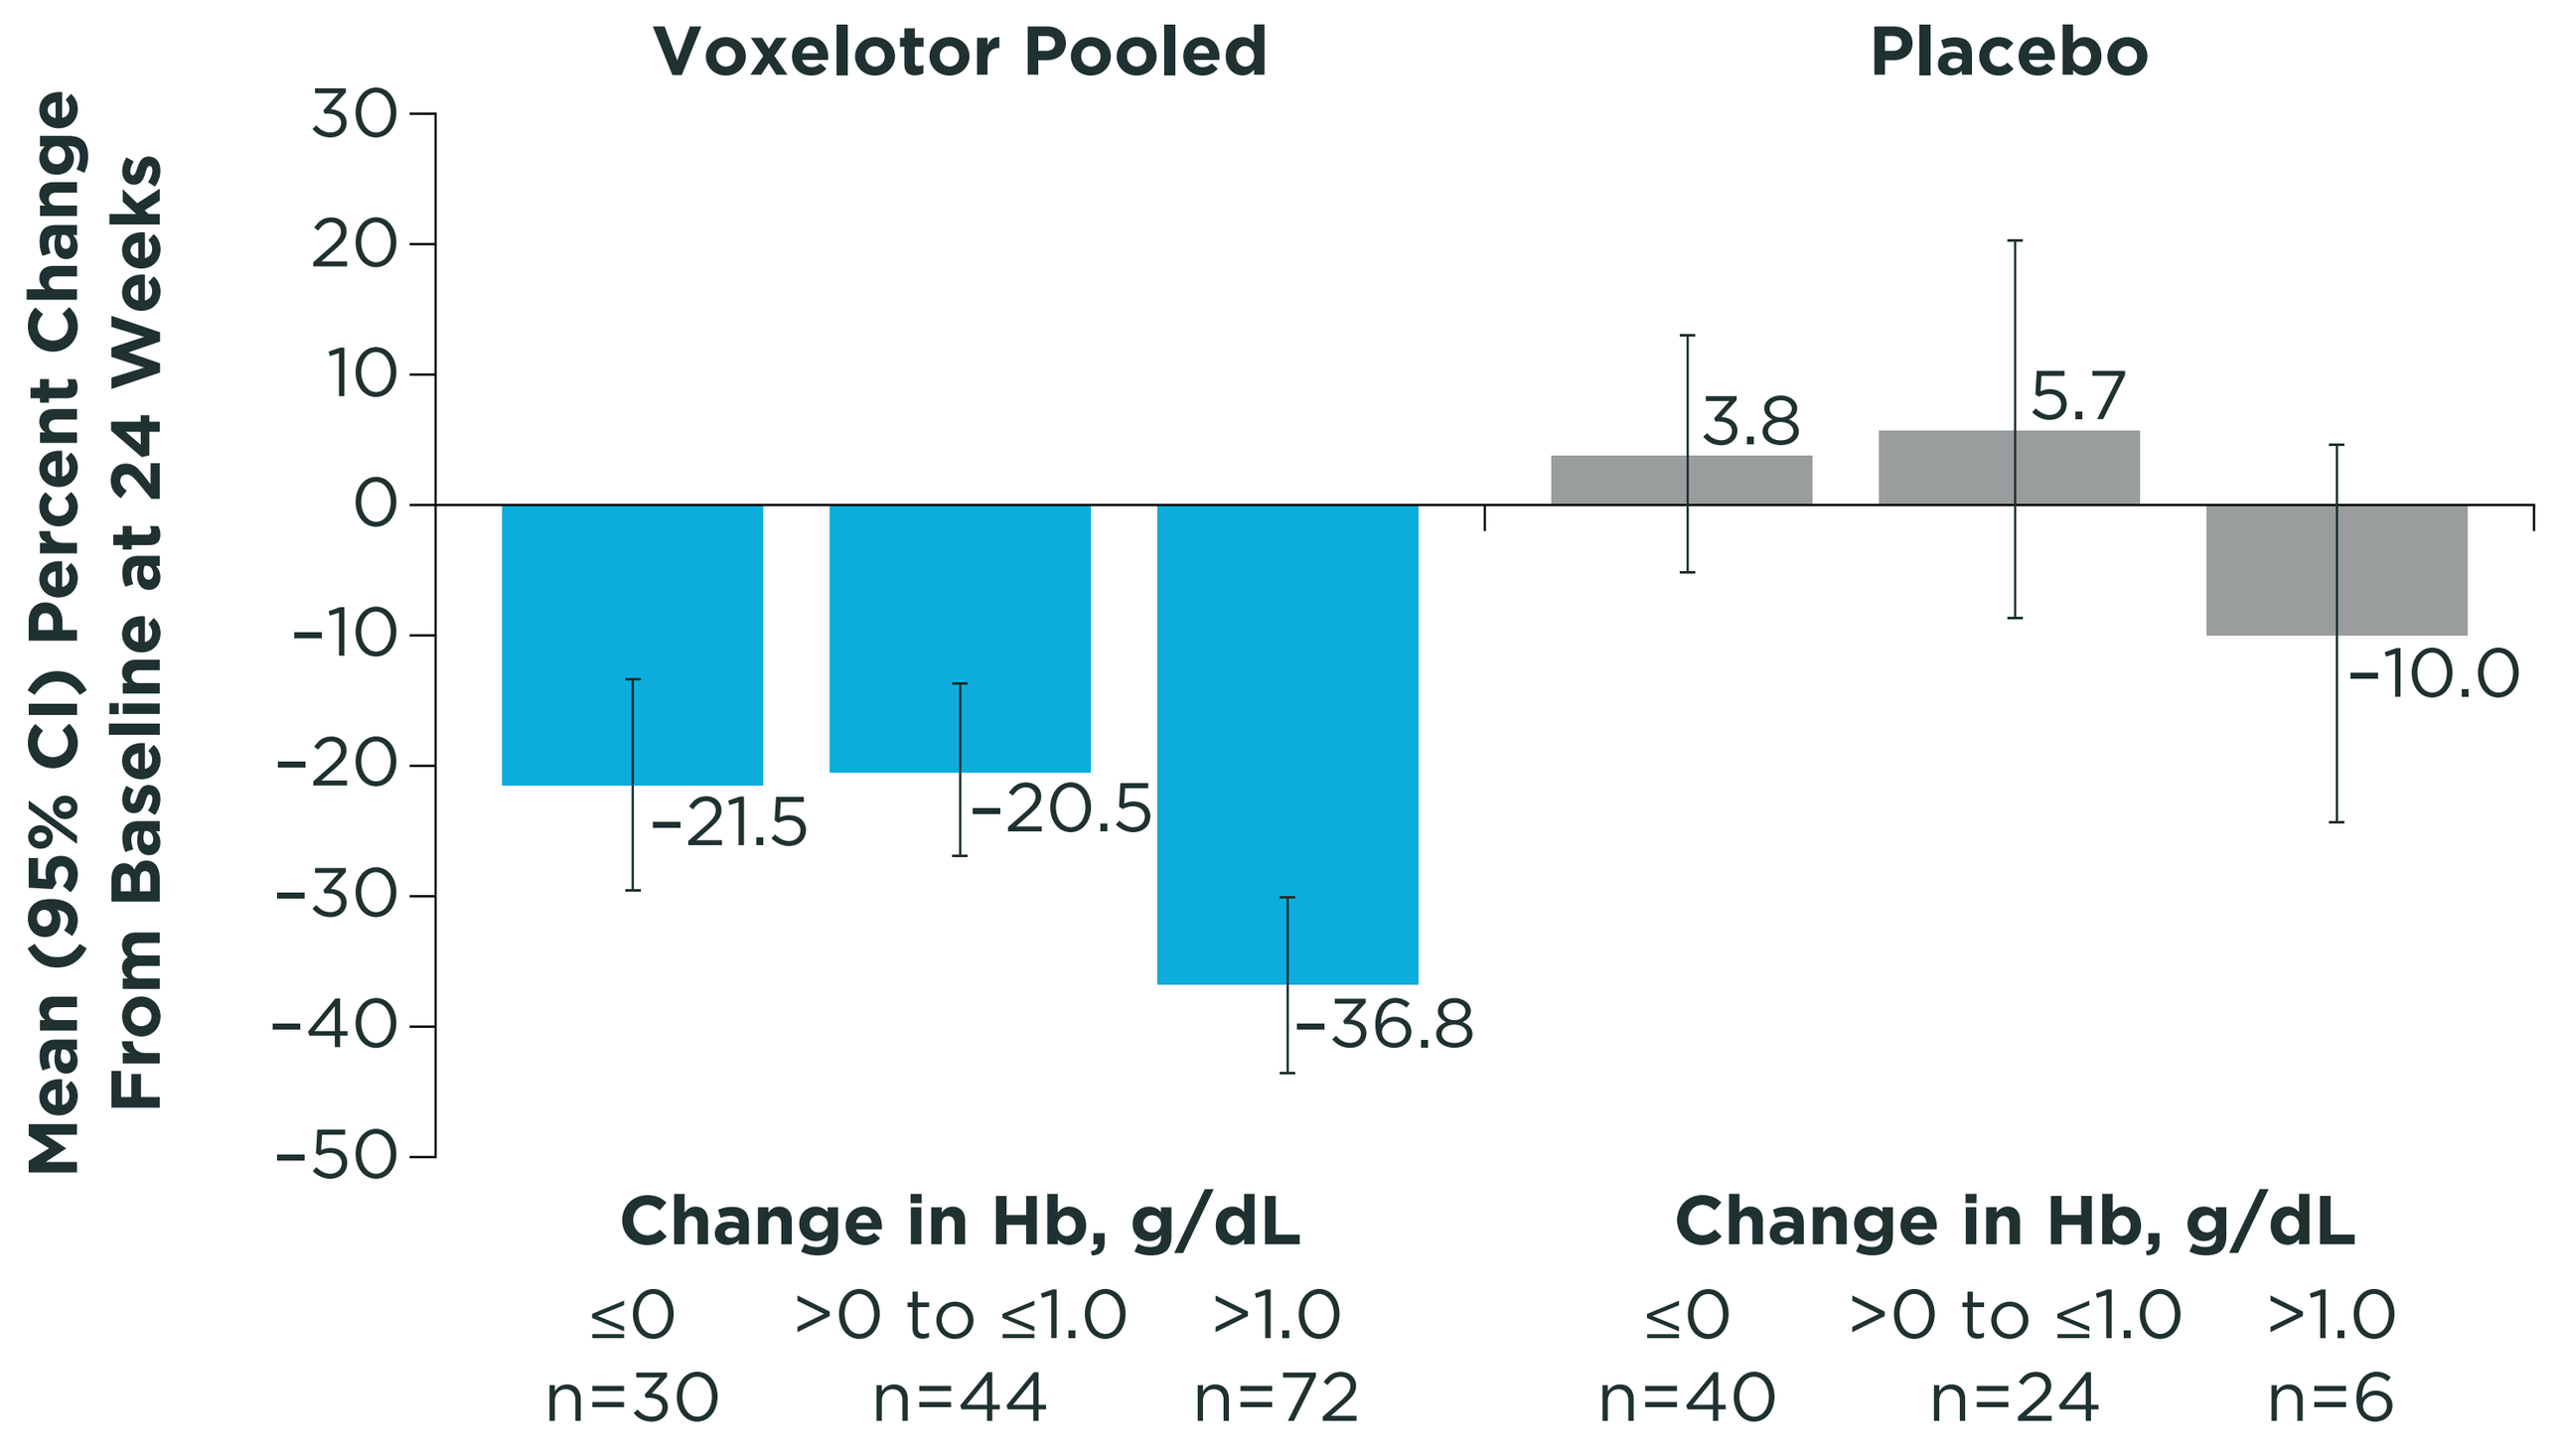
Voxelotor Pooled
Placebo
Mean (95% CI) Percent Change From Baseline at 24 Weeks
30
20
10
0
−10
−20
−30
−40
−50
−21.5
−20.5
−36.8
3.8
5.7
−10.0
Change in Hb, g/dL
≤0 n=30
>0 to ≤1.0 n=44
>1.0 n=72
Change in Hb, g/dL
≤0 n=40
>0 to ≤1.0 n=24
>1.0 n=6

B. Lactate Dehydrogenase

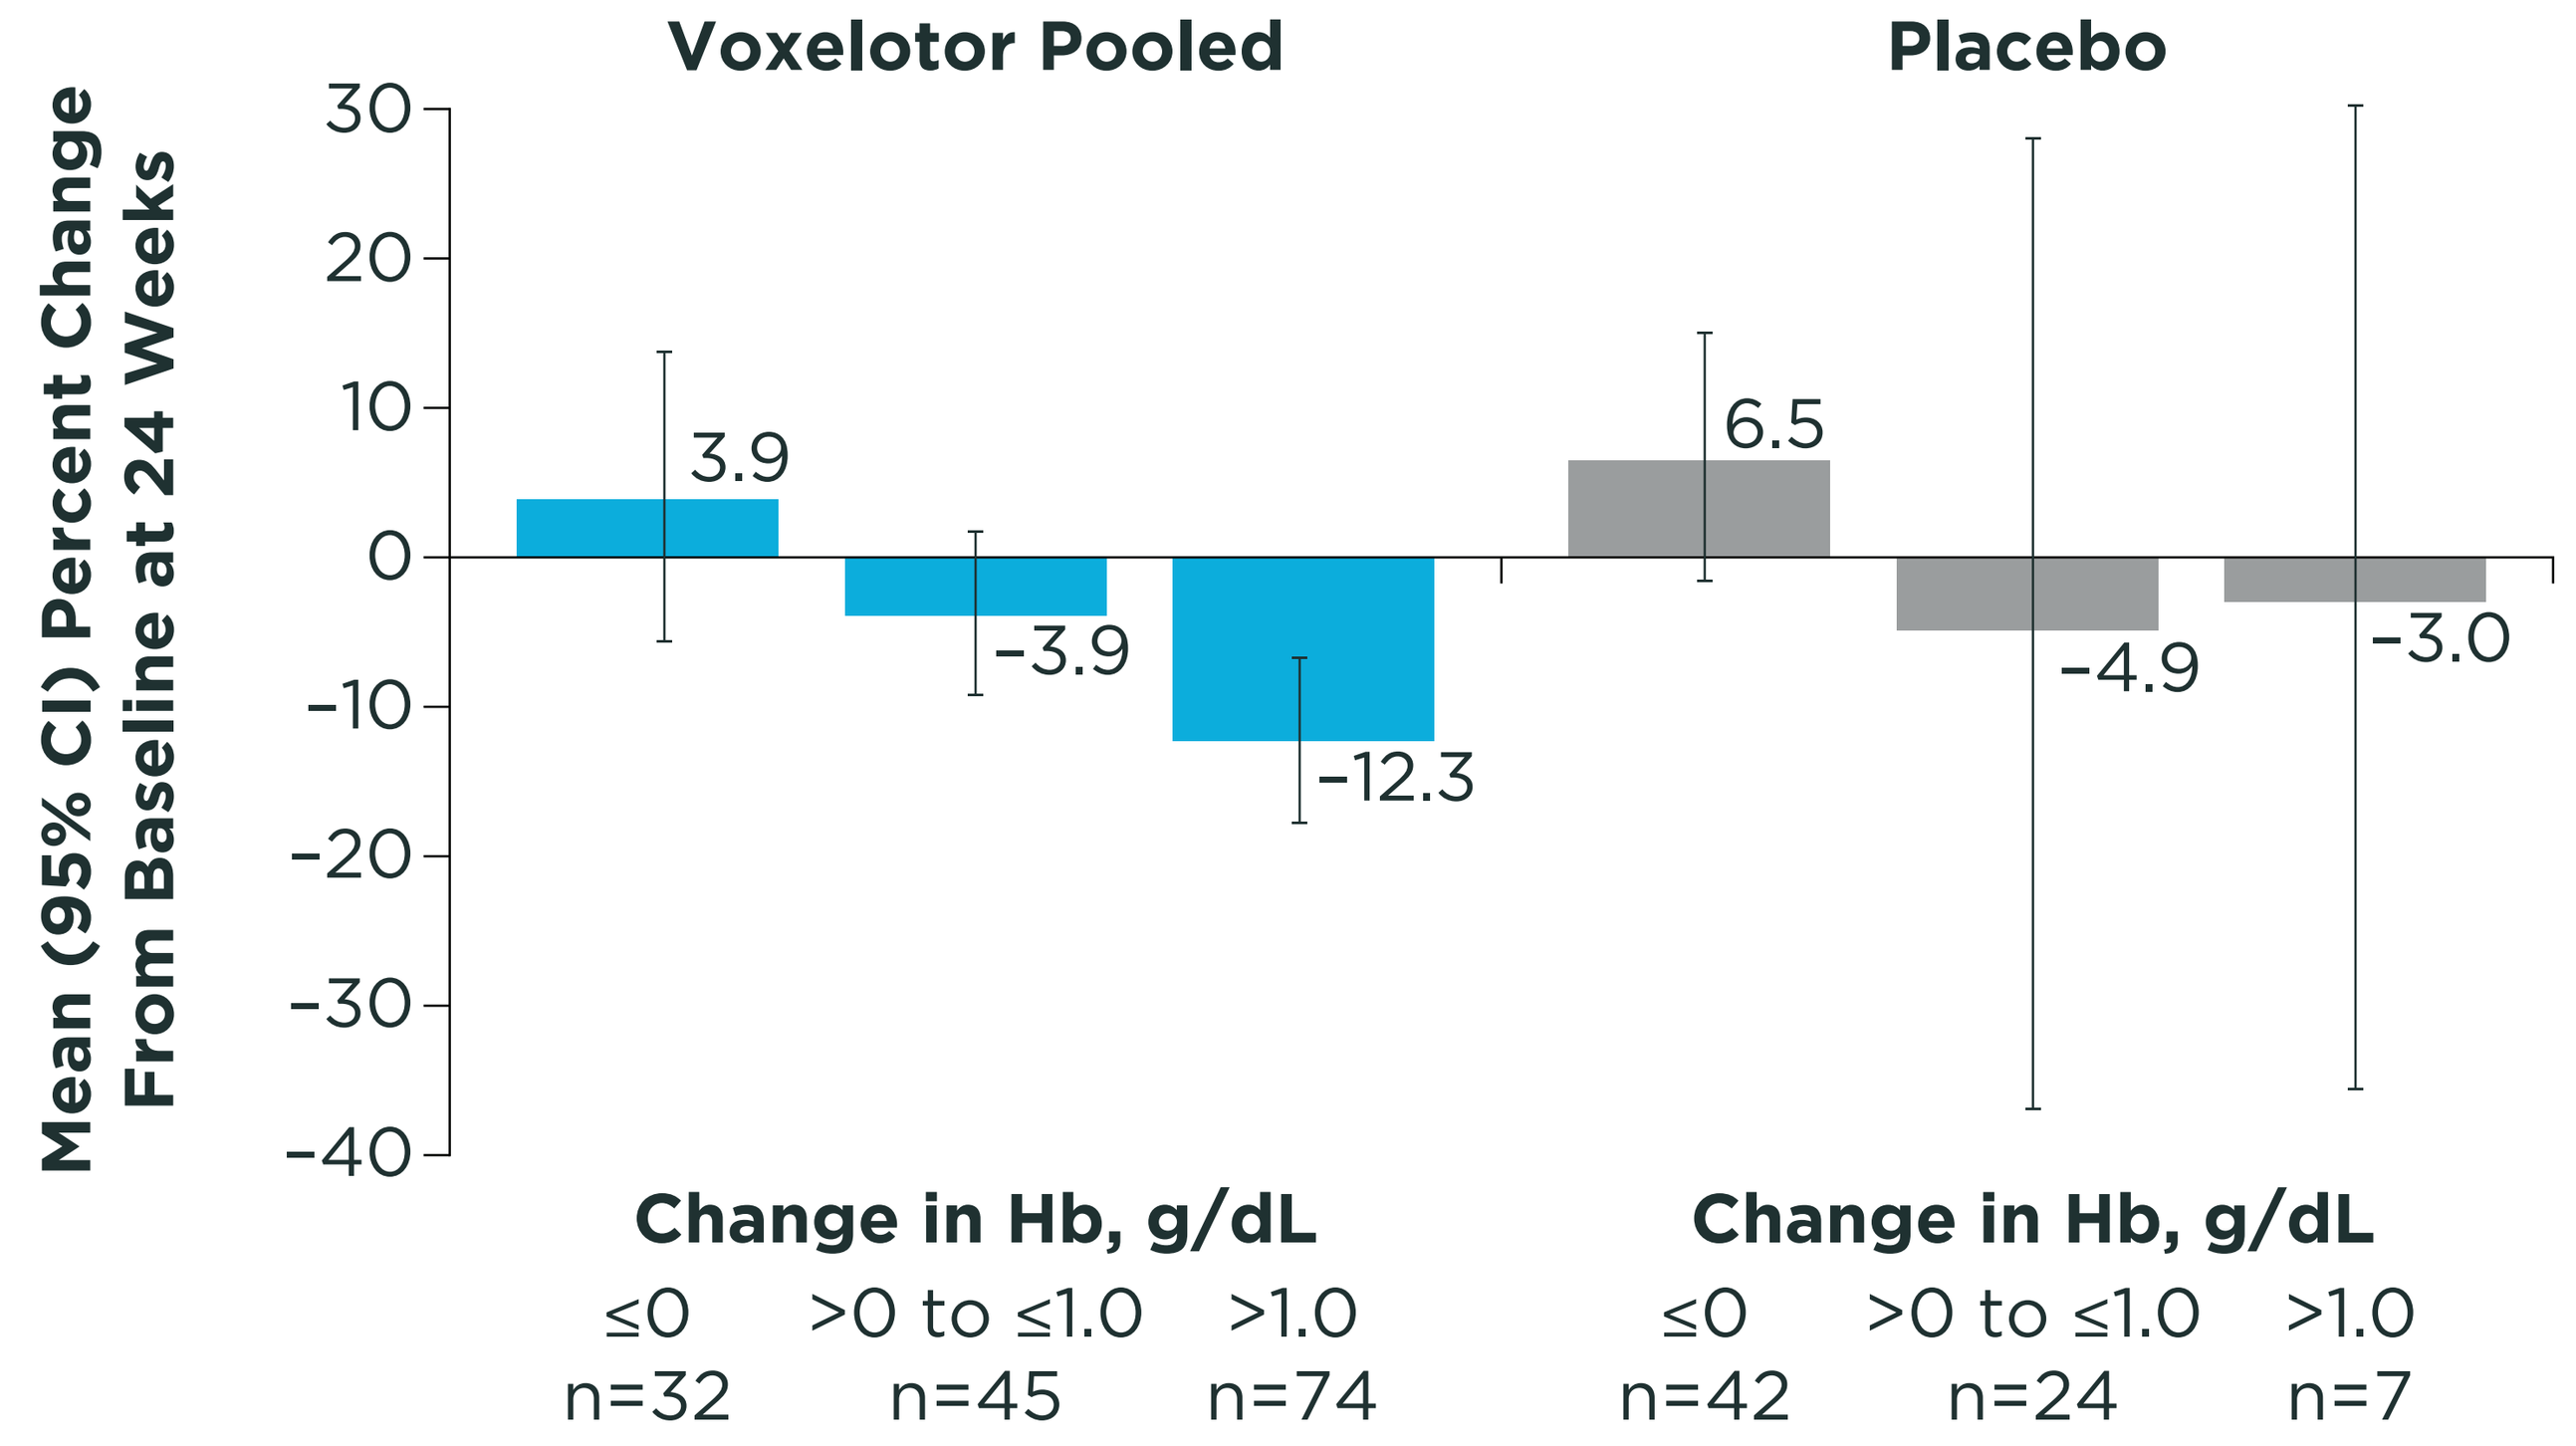
Voxelotor Pooled
Placebo
Mean (95% CI) Percent Change From Baseline at 24 Weeks
30
20
10
0
−10
−20
−30
−40
3.9
−3.9
−12.3
6.5
−4.9
−3.0
Change in Hb, g/dL
≤0 n=32
>0 to ≤1.0 n=45
>1.0 n=74
Change in Hb, g/dL
≤0 n=42
>0 to ≤1.0 n=24
>1.0 n=7

C. Absolute Reticulocyte Count

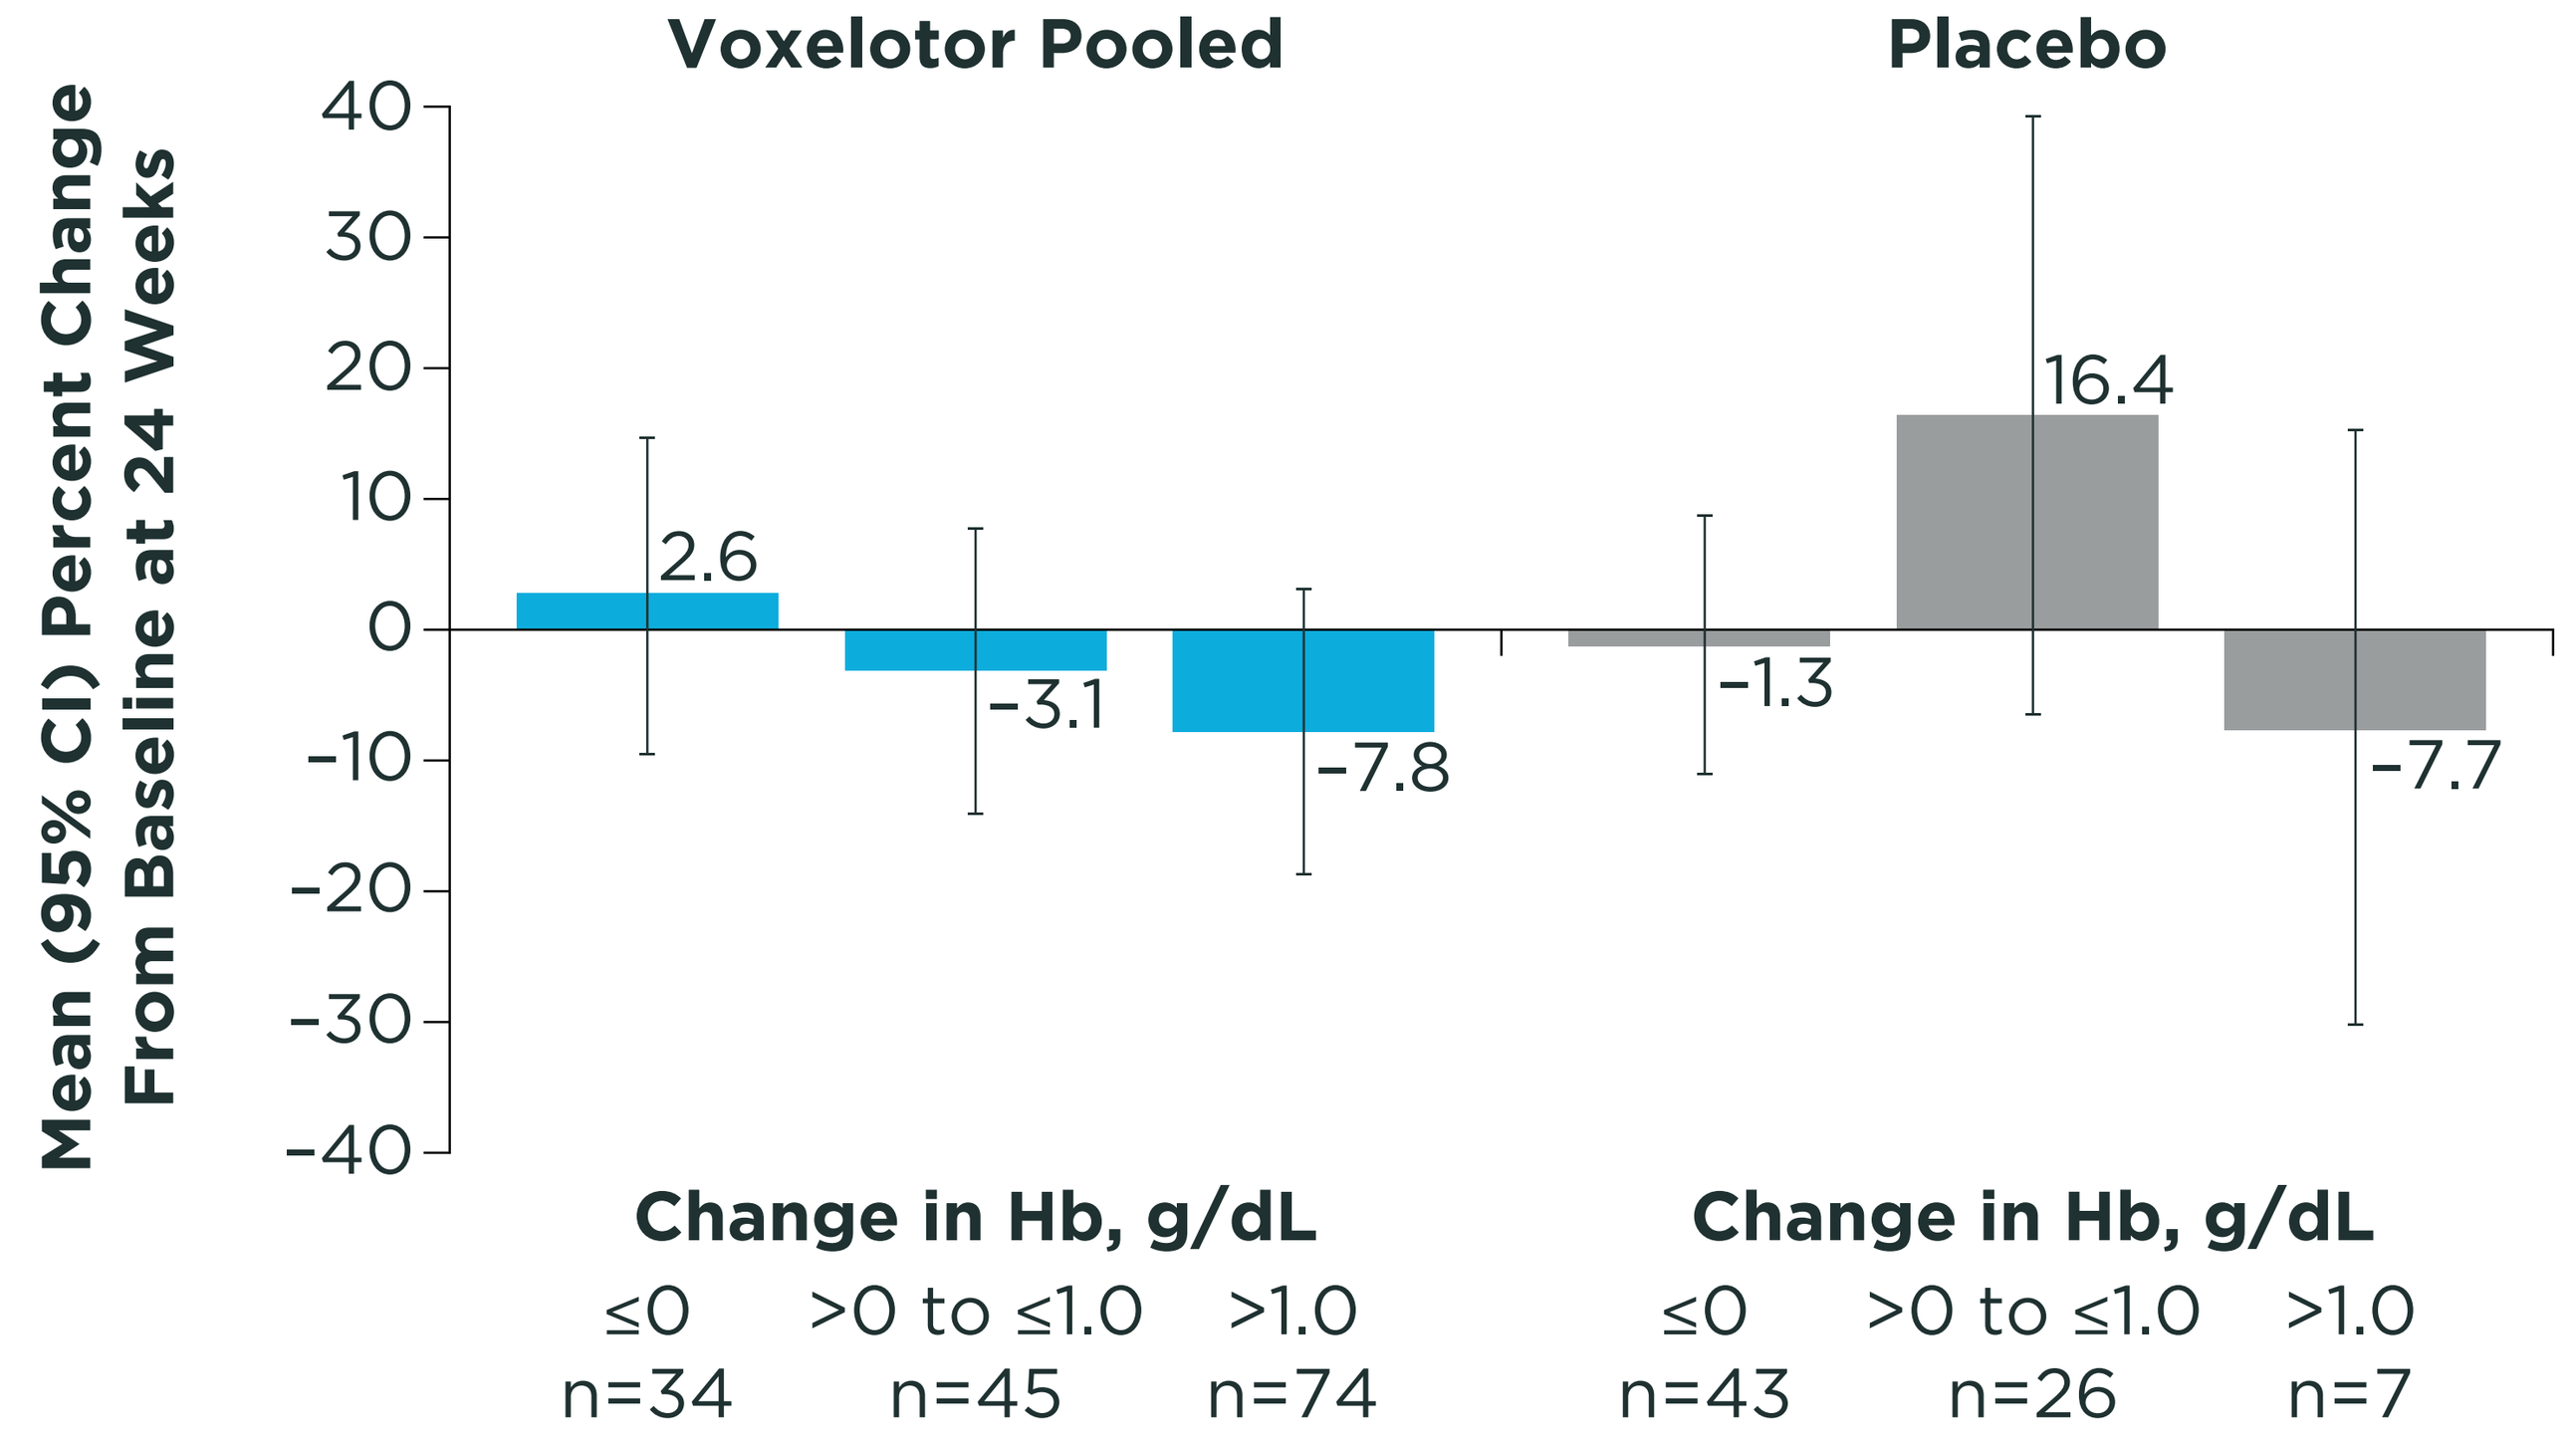
Voxelotor Pooled
Placebo
Mean (95% CI) Percent Change From Baseline at 24 Weeks
40
30
20
10
0
−10
−20
−30
−40
2.6
−3.1
−7.8
−1.3
16.4
−7.7
Change in Hb, g/dL
≤0 n=34
>0 to ≤1.0 n=45
>1.0 n=74
Change in Hb, g/dL
≤0 n=43
>0 to ≤1.0 n=26
>1.0 n=7

D. Percentage of Reticulocytes

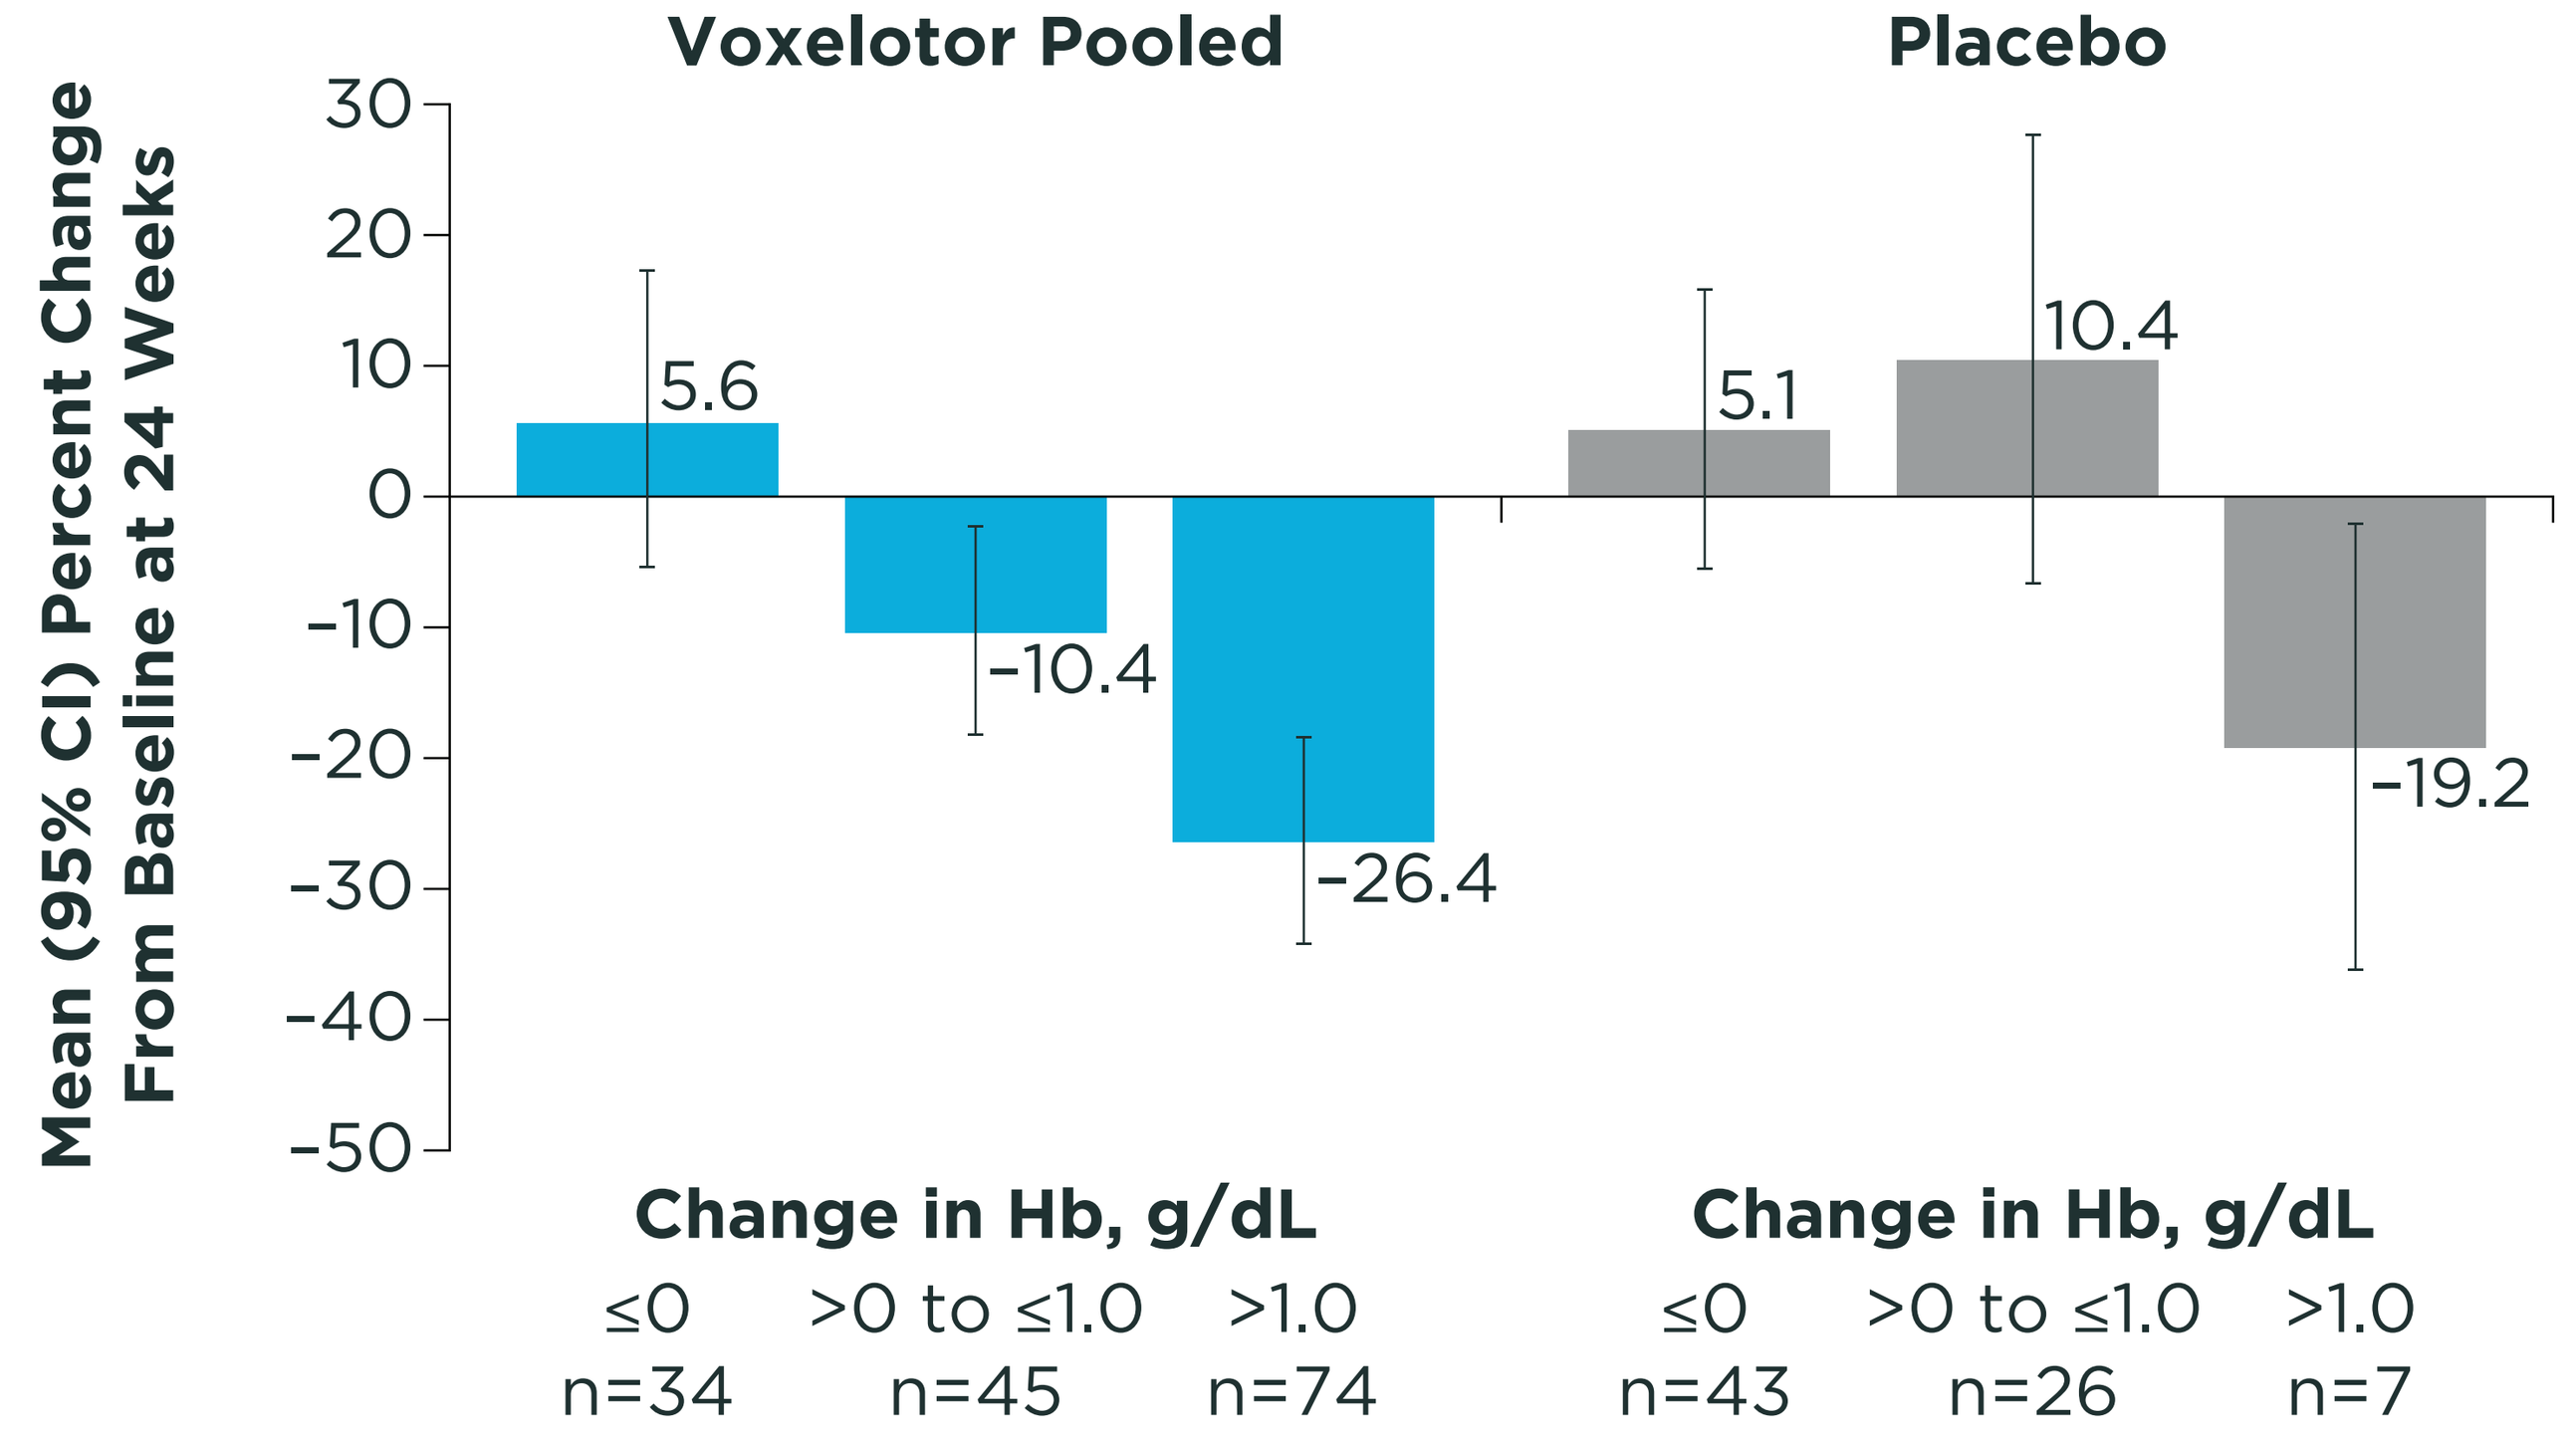
Voxelotor Pooled
Placebo
Mean (95% CI) Percent Change From Baseline at 24 Weeks
30
20
10
0
−10
−20
−30
−40
−50
5.6
−10.4
−26.4
5.1
10.4
−19.2
Change in Hb, g/dL
≤0 n=34
>0 to ≤1.0 n=45
>1.0 n=74
Change in Hb, g/dL
≤0 n=43
>0 to ≤1.0 n=26
>1.0 n=7

Voxelotor pooled includes both voxelotor 1500 mg and 900 mg groups.
CI, confidence interval; Hb, hemoglobin; LDH, lactate dehydrogenase.

- Decreases in indirect bilirubin and LDH levels, absolute reticulocyte count, and percentage of reticulocytes correlated with increases in voxelotor whole blood concentrations (*P*<0.001; **Figure 3**).

Figure 3. Decreases in Measures of Hemolysis Correlated With Increases in Voxelotor Whole Blood Concentrations

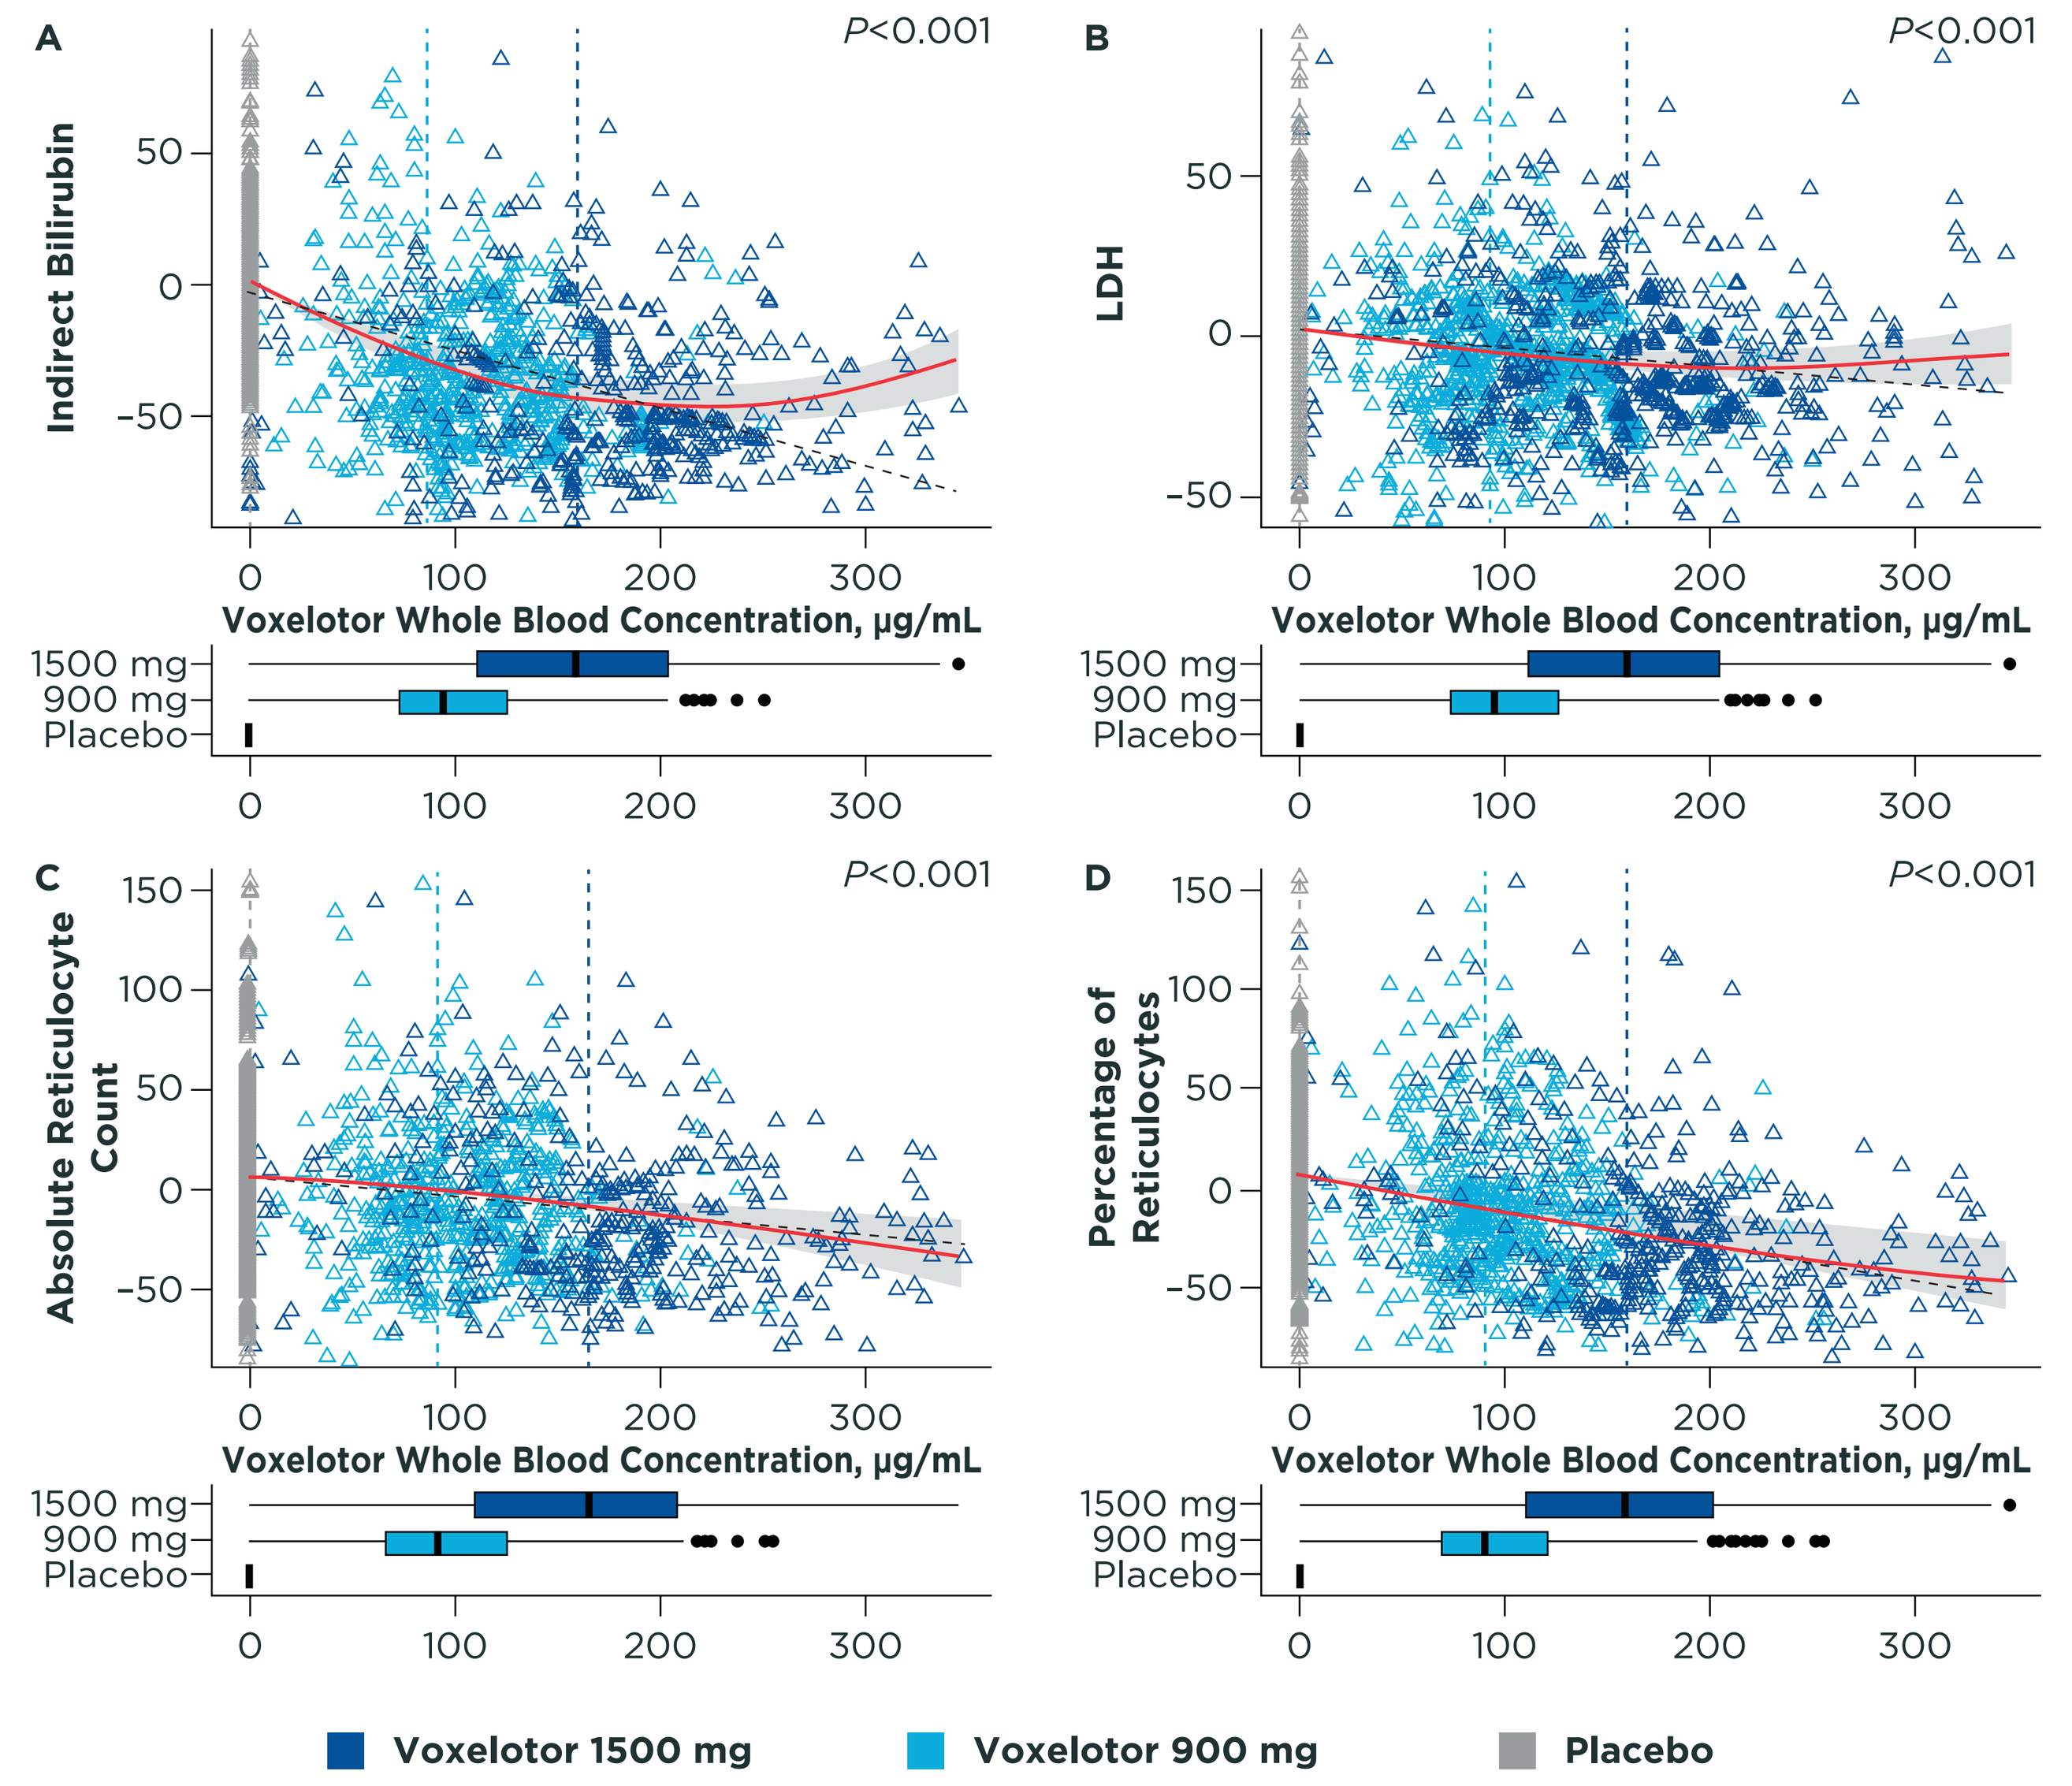
A
P<0.001
Indirect Bilirubin
B
P<0.001
LDH
C
P<0.001
Absolute Reticulocyte Count
D
P<0.001
Percentage of Reticulocytes
Voxelotor Whole Blood Concentration, μg/mL
1500 mg
900 mg
Placebo
0
100
200
300
Voxelotor 1500 mg
Voxelotor 900 mg
Placebo

Changes in measures of hemolysis are expressed as percentage change from baseline. Dashed vertical lines indicate median voxelotor blood concentrations for each treatment group. Box plots show the distribution of voxelotor concentrations in whole blood for each dosage group (the vertical black line within each box indicates the median, and the left and right edges correspond to the 25th and 75th percentiles, respectively). The solid red line and gray shaded area represent a second-degree polynomial regression and 95% confidence interval through the data, respectively. The black dashed line is a linear regression line through the data.
LDH, lactate dehydrogenase.

CONCLUSIONS

- Treatment with voxelotor was associated with reductions in indirect bilirubin level, LDH level, absolute reticulocyte count, and percentage of reticulocytes.
- Among patients treated with voxelotor, those with Hb increases of >1.0 g/dL had the greatest reduction in these markers; those with Hb increases of ≤1.0 g/dL experienced a greater reduction in most markers than patients who did not have an increase in Hb.
- Decreases in measures of hemolysis correlated with increases in voxelotor whole blood concentrations.
- These results suggest that the mechanism by which voxelotor raises Hb is related to a reduction in hemolysis, which may modify the morbidity of SCD by improving hemolytic anemia.

References

1. Kato GJ, Piel FB, Reid CD, et al. Sickle cell disease. *Nat Rev Dis Primers*. 2018;4:18010.
2. Kato GJ, Steinberg MH, Gladwin MT. Intravascular hemolysis and the pathophysiology of sickle cell disease. *J Clin Invest*. 2017;127(3):750-760.
3. Ware RE, de Montalembert M, Tshilolo L, Abboud MR. Sickle cell disease. *Lancet*. 2017;390(10091):311-323.
4. Vichinsky E, Hoppe CC, Ataga KI, et al. A phase 3 randomized trial of voxelotor in sickle cell disease. *N Engl J Med*. 2019;381(6):509-519.

Acknowledgments

- We thank all the patients with sickle cell disease, families, caregivers, HOPE investigators, research nurses, study coordinators, and support staff who contributed to this study.

Disclosures

- **Jo Howard:** Novartis, Terumo, Addmedica, speakers bureau; Global Blood Therapeutics, Imara, consultant. **Elliott Vichinsky:** Global Blood Therapeutics, consultant; Agios, Pfizer, research funding. **Jennifer Knight-Madden:** Global Blood Therapeutics, research funding; Global Blood Therapeutics, Addmedica, Bluebird Bio, Nova Laboratories, Abbott International, Abbott Nutrition, institution conference sponsorship. **Margaret Tonda, Carla Washington, Barbara Tong, Joshua Lehrer-Graiwer**: Global Blood Therapeutics, employees and stockholders. **Victor R. Gordeuk:** Global Blood Therapeutics, Emmaus, Incyte, Novartis, research funding; Modus Therapeutics, consultant.
- Nelson Jen, PhD (Healthcare Consultancy Group, with funding from Global Blood Therapeutics) provided editorial assistance in the preparation of this report.
- This study was sponsored by Global Blood Therapeutics.

For a copy of this poster, scan the QR code with your Android™ phone, Blackberry®, or iPhone®. No personal information will be collected. This is not associated with any marketing or promotional activity.

https://bit.ly/34j5MCv

Presented at: 61st ASH Annual Meeting and Exposition; December 7-10, 2019; Orlando, FL, USA

© 2019 Global Blood Therapeutics